A Phase 2, Multicenter Study of the EZH2 Inhibitor Tazemetostat in Adults (Epithelioid Sarcoma Cohort) (NCT02601950)

1615PD

Mrinal Gounder[1], Patrick Schöffski[2], Victor Villalobos[3], Gregory Cote[4], Rashmi Chugh[5], Tom Wei-Wu Chen[6], Thierry Jahan[7], Elizabeth Loggers[8], Antoine Italiano[9], Abha Gupta[10], Mark Agulnik[11], Steven Attia[12], Robin L. Jones[13], Brian A. Van Tine[14], George D. Demetri[15], Maria Roche[16], Inbal Sapir[16], Scott Daigle[16], Alicia Clawson[16], Silvia Stacchiotti[17]

[1]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [2]University Hospitals Leuven, Leuven Cancer Institute, Leuven, Belgium; [3]University of Colorado, Denver, CO, USA; [4]Massachusetts General Hospital, Boston, MA, USA; [5]Michigan Medicine Comprehensive Cancer Center, Ann Arbor, MI, USA; [6]National Taiwan University Hospital, Taipei, Taiwan; [7]University of California, San Francisco, CA, USA; [8]Fred Hutchinson Cancer Research Center, Seattle, WA, USA; [9]Institut Bergonie, Bordeaux, France; [10]The Hospital for Sick Children and Princess Margaret Cancer Center, Toronto, Ontario, Canada; [11]Northwestern Memorial Hospital, Chicago, IL, USA; [12]Mayo Clinic in Florida, Jacksonville, FL, USA; [13]Royal Marsden Hospital and Institute of Cancer Research, London, UK; [14]Washington University in St. Louis School of Medicine, St. Louis, MO, USA; [15]Dana Farber Cancer Institute and Ludwig Center at Harvard Medical School, Boston, MA, USA; [16]Epizyme, Cambridge, MA, USA; [17]Fondazione IRCCS Instituto Nazionale Tumori, Milan, Italy

BACKGROUND

- Epithelioid sarcoma (ES) is a rare soft tissue sarcoma (STS); currently available treatments for metastatic STS have been shown to have limited activity in terms of durable clinical benefit and significant toxicities[1-3]
- Loss of integrase interactor 1 (INI1), a subunit of the SWItch/Sucrose Non-Fermentable (SWI/SWF) complex and a negative regulator of enhancer of zeste homolog 2 (EZH2), occurs in over 90% of ES tumors and is a characteristic diagnostic feature of ES[4-7]
 - EZH2, a histone methyltransferase in polycomb repressive complex 2 (PRC2), is a known oncogenic driver[5-7]
- In INI1-deficient preclinical models, EZH2 inhibition reprograms pathways related to abnormal cell growth, leading to cell death or differentiation and subsequent tumor control and regression[8]
- Tazemetostat, a potent, selective, orally available inhibitor of EZH2, has shown clinical activity in malignant rhabdoid tumors and in patients with ES[9,10] (Figure 1)
- Here we report interim phase 2 safety and activity of tazemetostat in patients with ES

Figure 1. Tazemetostat inhibits EZH2-mediated repression of transcription

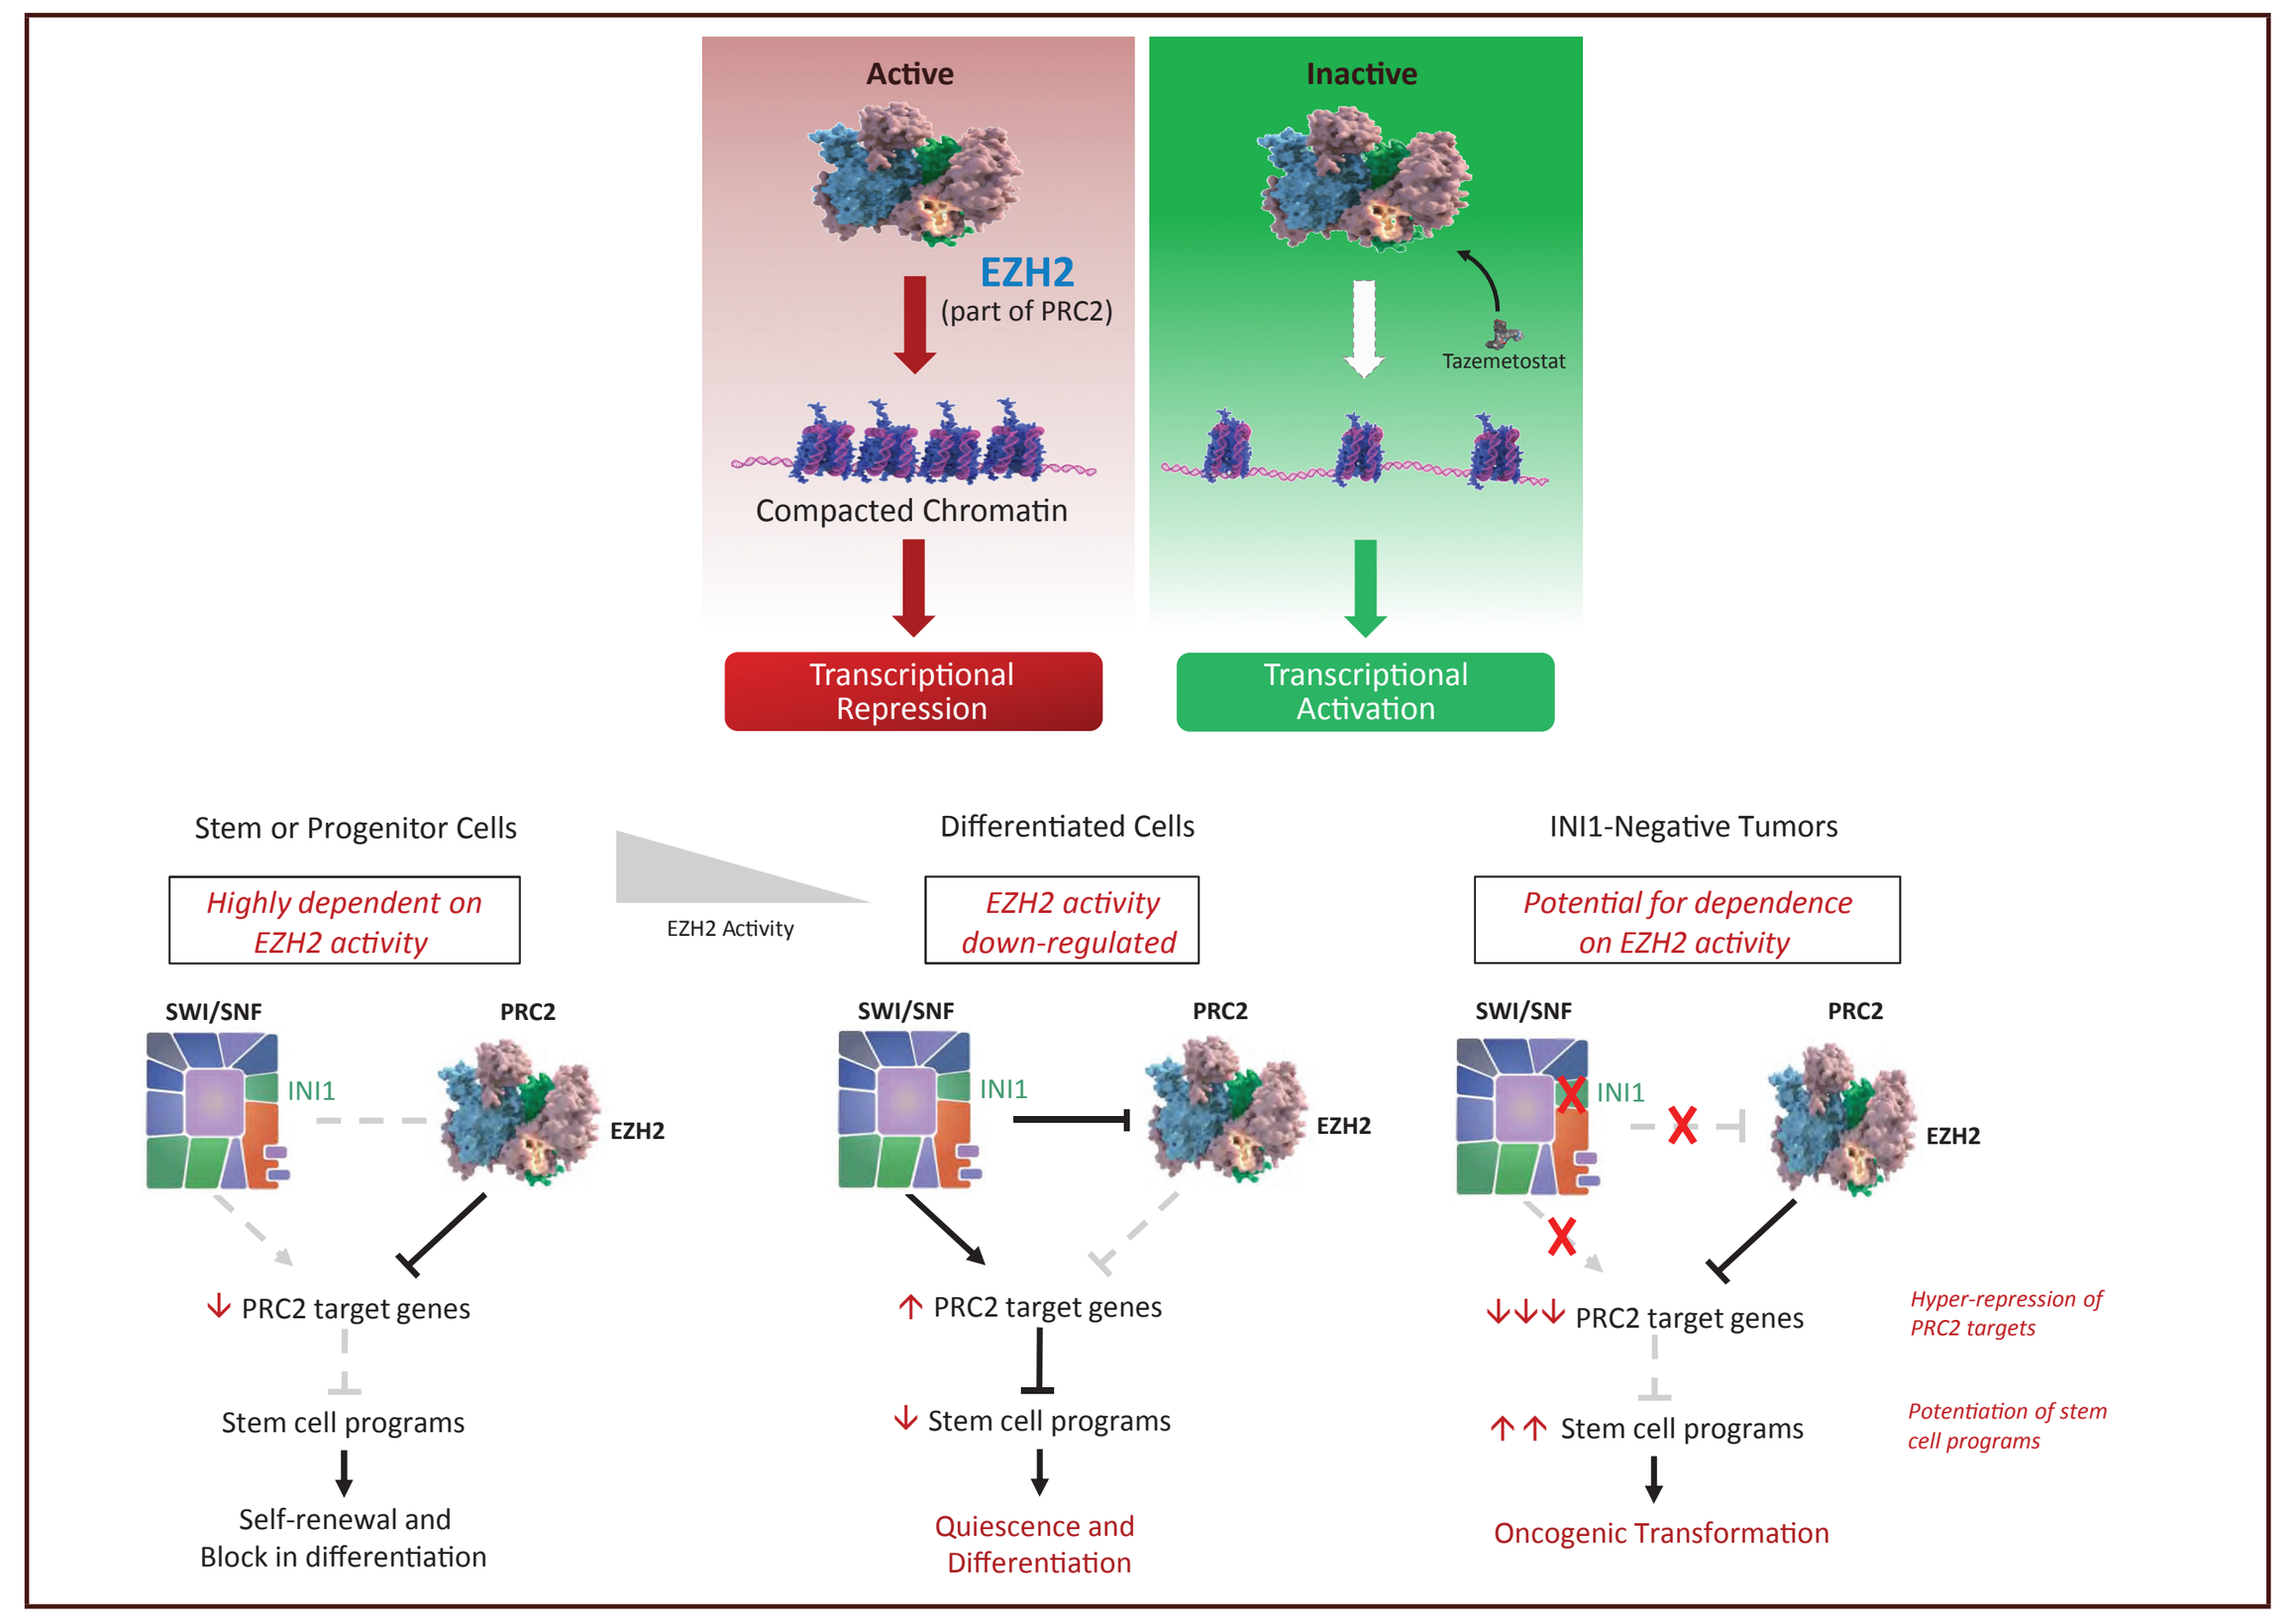
Active
Inactive
EZH2
(part of PRC2)
Tazemetostat
Compacted Chromatin
Transcriptional Repression
Transcriptional Activation
Stem or Progenitor Cells
Differentiated Cells
INI1-Negative Tumors
Highly dependent on EZH2 activity
EZH2 Activity
EZH2 activity down-regulated
Potential for dependence on EZH2 activity
SWI/SNF
PRC2
INI1
EZH2
PRC2 target genes
Stem cell programs
Self-renewal and Block in differentiation
Quiescence and Differentiation
Hyper-repression of PRC2 targets
Potentiation of stem cell programs
Oncogenic Transformation

EZH2, enhancer of zeste homolog 2; INI1, integrase interactor 1; PRC2, polycomb repressive complex 2; SWI/SNF, SWItch/Sucrose Non-Fermentable.

METHODS

- Study EZH-202 (NCT02601937) is a phase 2, open-label, multi-center study of tazemetostat that enrolled patients aged ≥16 years into 7 cohorts. The current study reflects patients with ES (Cohort 5)
- Tazemetostat 800 mg was administered orally twice daily in continuous 28-day cycles until disease progression, unacceptable toxicity, or withdrawal of consent
- All patients were required to have tumor INI1 loss confirmed by histology
 - Key inclusion and exclusion criteria are presented in Table 1
- In cohort 5, tazemetostat was studied in adults (≥16 years of age) with ES in a 2-stage Green-Dahlberg design (Figure 2)
- Primary endpoint is RECIST 1.1 overall response rate (ORR)
- Secondary endpoints included disease control rate (DCR; confirmed RECIST 1.1 complete response [CR] or partial response [PR] of any duration, or stable disease [SD] lasting ≥32 weeks), safety/tolerability, and duration of response
- Response assessment was evaluated after 8 weeks of treatment and then every 8 weeks thereafter while on study

Statistical considerations

- Stage 1 futility was performed after the first 15 patients enrolled completed at least the 24-week assessment, the final study visit, or terminated early and required ≥1 patient achieving an objective response (CR or PR)
- Stage 2 success required disease control in ≥5/30 treated patients
- Stage 2 futility criterion was passed in October, 2016 (final N=32); the Expansion Stage opened in December, 2016 (final N=32)

Table 1. Key inclusion and exclusion criteria for cohort 5

Inclusion
Advanced measurable disease
ECOG performance status of 0 or 1
Life expectancy >3 months
Morphology and immunophenotypic panel consistent with INI1-negative tumors and loss of INI1 confirmed by IHC
Molecular confirmation of tumor bi-allelic INI1 loss or mutation when INI1 IHC is equivocal or unavailable
Expansion Stage inclusion criteria required that the patient's disease had progressed within 6 months prior to study enrollment. Stages 1 and 2 did not include this as a criteria[a]
Exclusion
Prior exposure to tazemetostat or other inhibitor(s) of EZH2
Prior malignancy other than the malignancies under study **EXCEPTION:** disease-free for 5 years, or with a history of a completely resected non-melanoma skin cancer or successfully treated in situ carcinoma is eligible
Major surgery within 3 weeks prior to enrollment
Cardiovascular impairment

[a]77% of patients in Stages 1 and 2 had documented progression within 6 months prior to study enrollment.
CLIA/CAP, Clinical Laboratory Improvement Amendments/College of American Pathologists; ECOG, Eastern Cooperative Oncology Group; EZH2, enhancer of zeste homolog 2; IHC, immunohistochemistry; INI1, integrase interactor 1; NYHA, New York Heart Association.

Figure 2. Study design

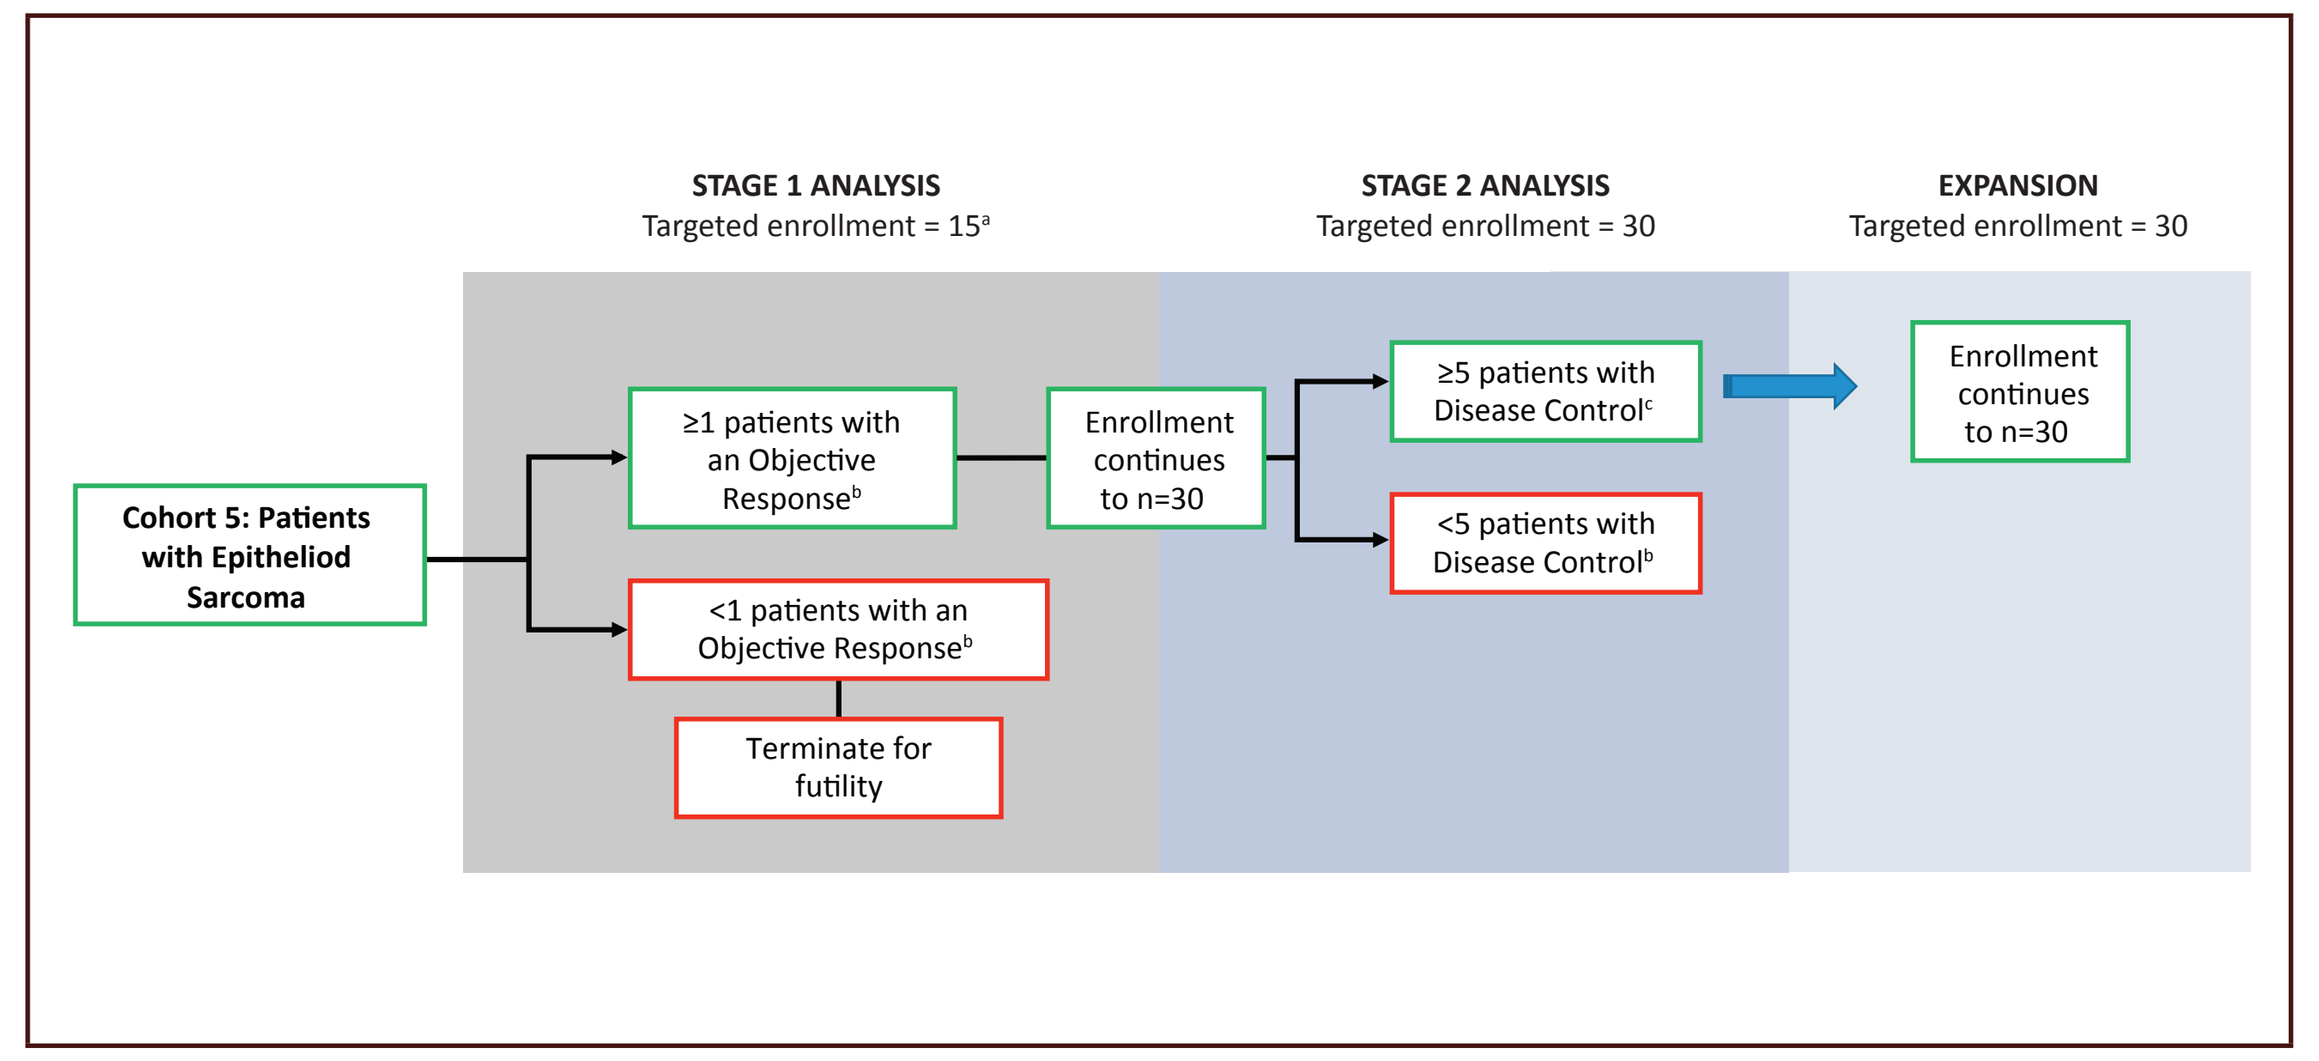
STAGE 1 ANALYSIS
Targeted enrollment = 15[a]
STAGE 2 ANALYSIS
Targeted enrollment = 30
EXPANSION
Targeted enrollment = 30
Cohort 5: Patients with Epitheliod Sarcoma
≥1 patients with an Objective Response[b]
<1 patients with an Objective Response[b]
Terminate for futility
Enrollment continues to n=30
≥5 patients with Disease Control[c]
<5 patients with Disease Control[b]
Enrollment continues to n=30

[a]All patients will have completed at least the week 24 assessment, completed the final study visit, or terminated early from the study, whichever is sooner. [b]Objective response: RECIST 1.1 – confirmed complete response or partial response at week 24. [c]Disease control: RECIST 1.1 - confirmed response [CR + PR or (SD for at least 32 weeks)]. RECIST, Response Evaluation Criteria in Solid Tumors.

RESULTS

- Datacut of 21 August 2018

Patient characteristics

- Of 62 patients enrolled, mean age was 37 years, 63% were male, and the median number of prior systemic therapies was one (Table 2)

Patient disposition

- Overall, 51 (82%) patients discontinued from the study
 - The majority of patients discontinued due to disease progression (n=43; 69%)
 - Four patients discontinued due to death not related to study drug, 2 patients discontinued after refusing further treatment of study drug, and one patient each for investigator discretion and other
 - No patients discontinued due to a treatment-emergent adverse event (TEAE)

Clinical Activity

- PRs were confirmed in 8 patients with an ORR of 13% and a DCR [CR+PR+(SD ≥32 weeks)] of 24%
- Patients who were naïve to treatment demonstrated a greater ORR, DCR, and PFS

Table 2. Patient demographics and disease characteristics

Clinical Characteristics	Total Number of Patients N=62
Age (years), mean (SD)	37 (15)
Male/female, n (%)	39 (63) / 23 (37)
Subtype (%)	
Proximal	20 (32)
Distal	29 (47)
Unknown[a]	13 (21)
Stage at diagnosis	
I	2 (3)
II	7 (11)
III	7 (11)
IV	37 (60)
Not applicable/unknown	9 (15)
Median time (months) from last PD to study entry, median (range); n=59	1.4 (0.3 to 12.8)
Number of lines of prior anticancer therapy	
0	24 (39)
1	12 (19)
2	12 (19)
≥3	14 (23)
Median (range)	1 (0–9)

[a]Unknown reflects data not included by investigator; PD, progressive disease; SD, standard deviation.

Table 3. Best response to tazemetostat treatment

Endpoint Category, n (%)	No Prior Anticancer Therapy N=24	Prior Anticancer Therapy N=38	Total N=62
DCR [CR + PR + (SD≥32 weeks)] 90% CI	9 (38) (21.2, 56.3)	6 (16) (7.1, 28.8)	15 (24) (15.5, 34.8)
ORR (CR + PR) 90% CI	5 (21) (8.6, 38.9)	3 (8) (2.2, 19.2)	8 (13) (6.6, 22.1)
Best Response			
CR	0	0	0
PR	5 (21)	3 (8)	8 (13)
SD	16 (67)	20 (53)	36 (58)
PD	2 (8)	11 (29)	13 (21)
Non-evaluable, missing, or unknown	1 (4)	4 (11)	5 (8)
Median duration of response, weeks Range	41 (7.1+, 48.1+)	48+ (40.1, 95.0+)	48+ (7.1+, 95.0+)
Median progression-free survival, weeks 90% CI	25.7 (23.7, NE)	14.7 (8.3, 16.0)	16.1 (15.1, 25.3)
Median overall survival, weeks 90% CI	NE	47.4 (33.7, 64.1)	82.4 (47.7, NE)

CI, confidence interval; CR, complete response; DCR, disease control rate; NE, not evaluable; ORR, objective response rate, PD, progressive disease; PR, partial response; SD, stable disease; +, responses ongoing/medians not reached

Figure 3. Durability of disease control in patients without prior anticancer therapy

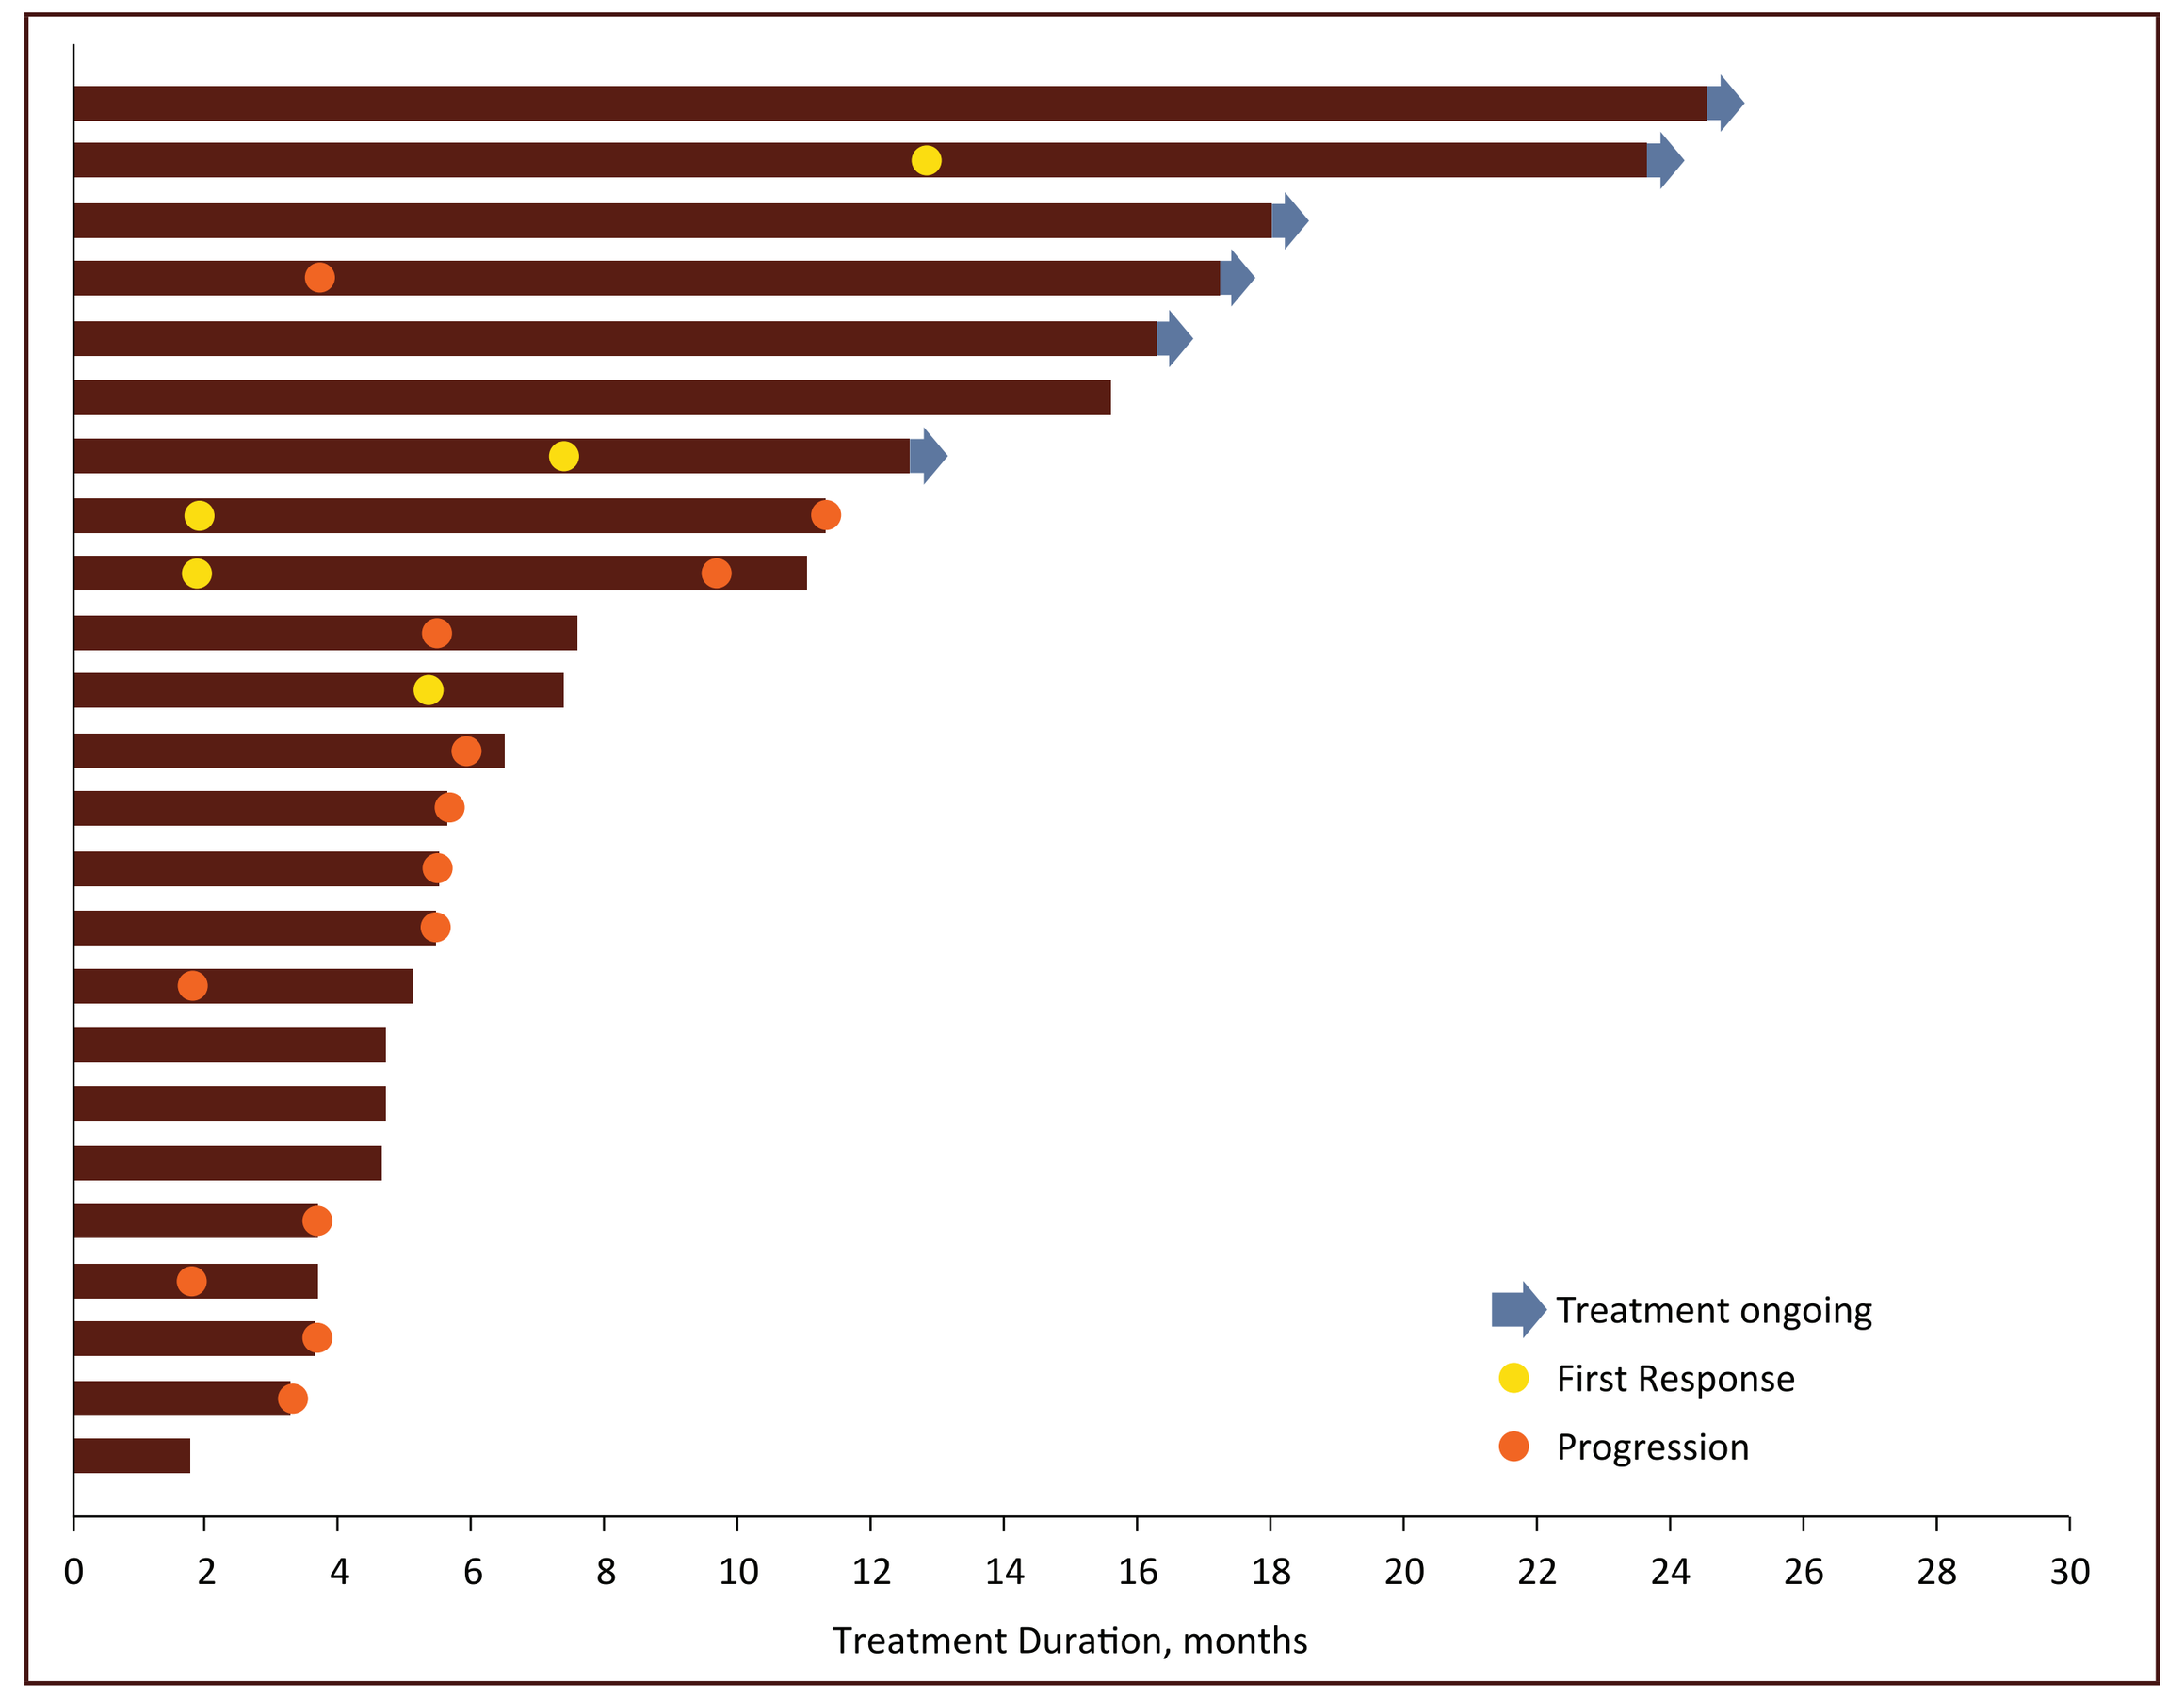
Treatment ongoing
First Response
Progression
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28 30
Treatment Duration, months

Figure 4. Durability of disease control in patients with prior anticancer therapy

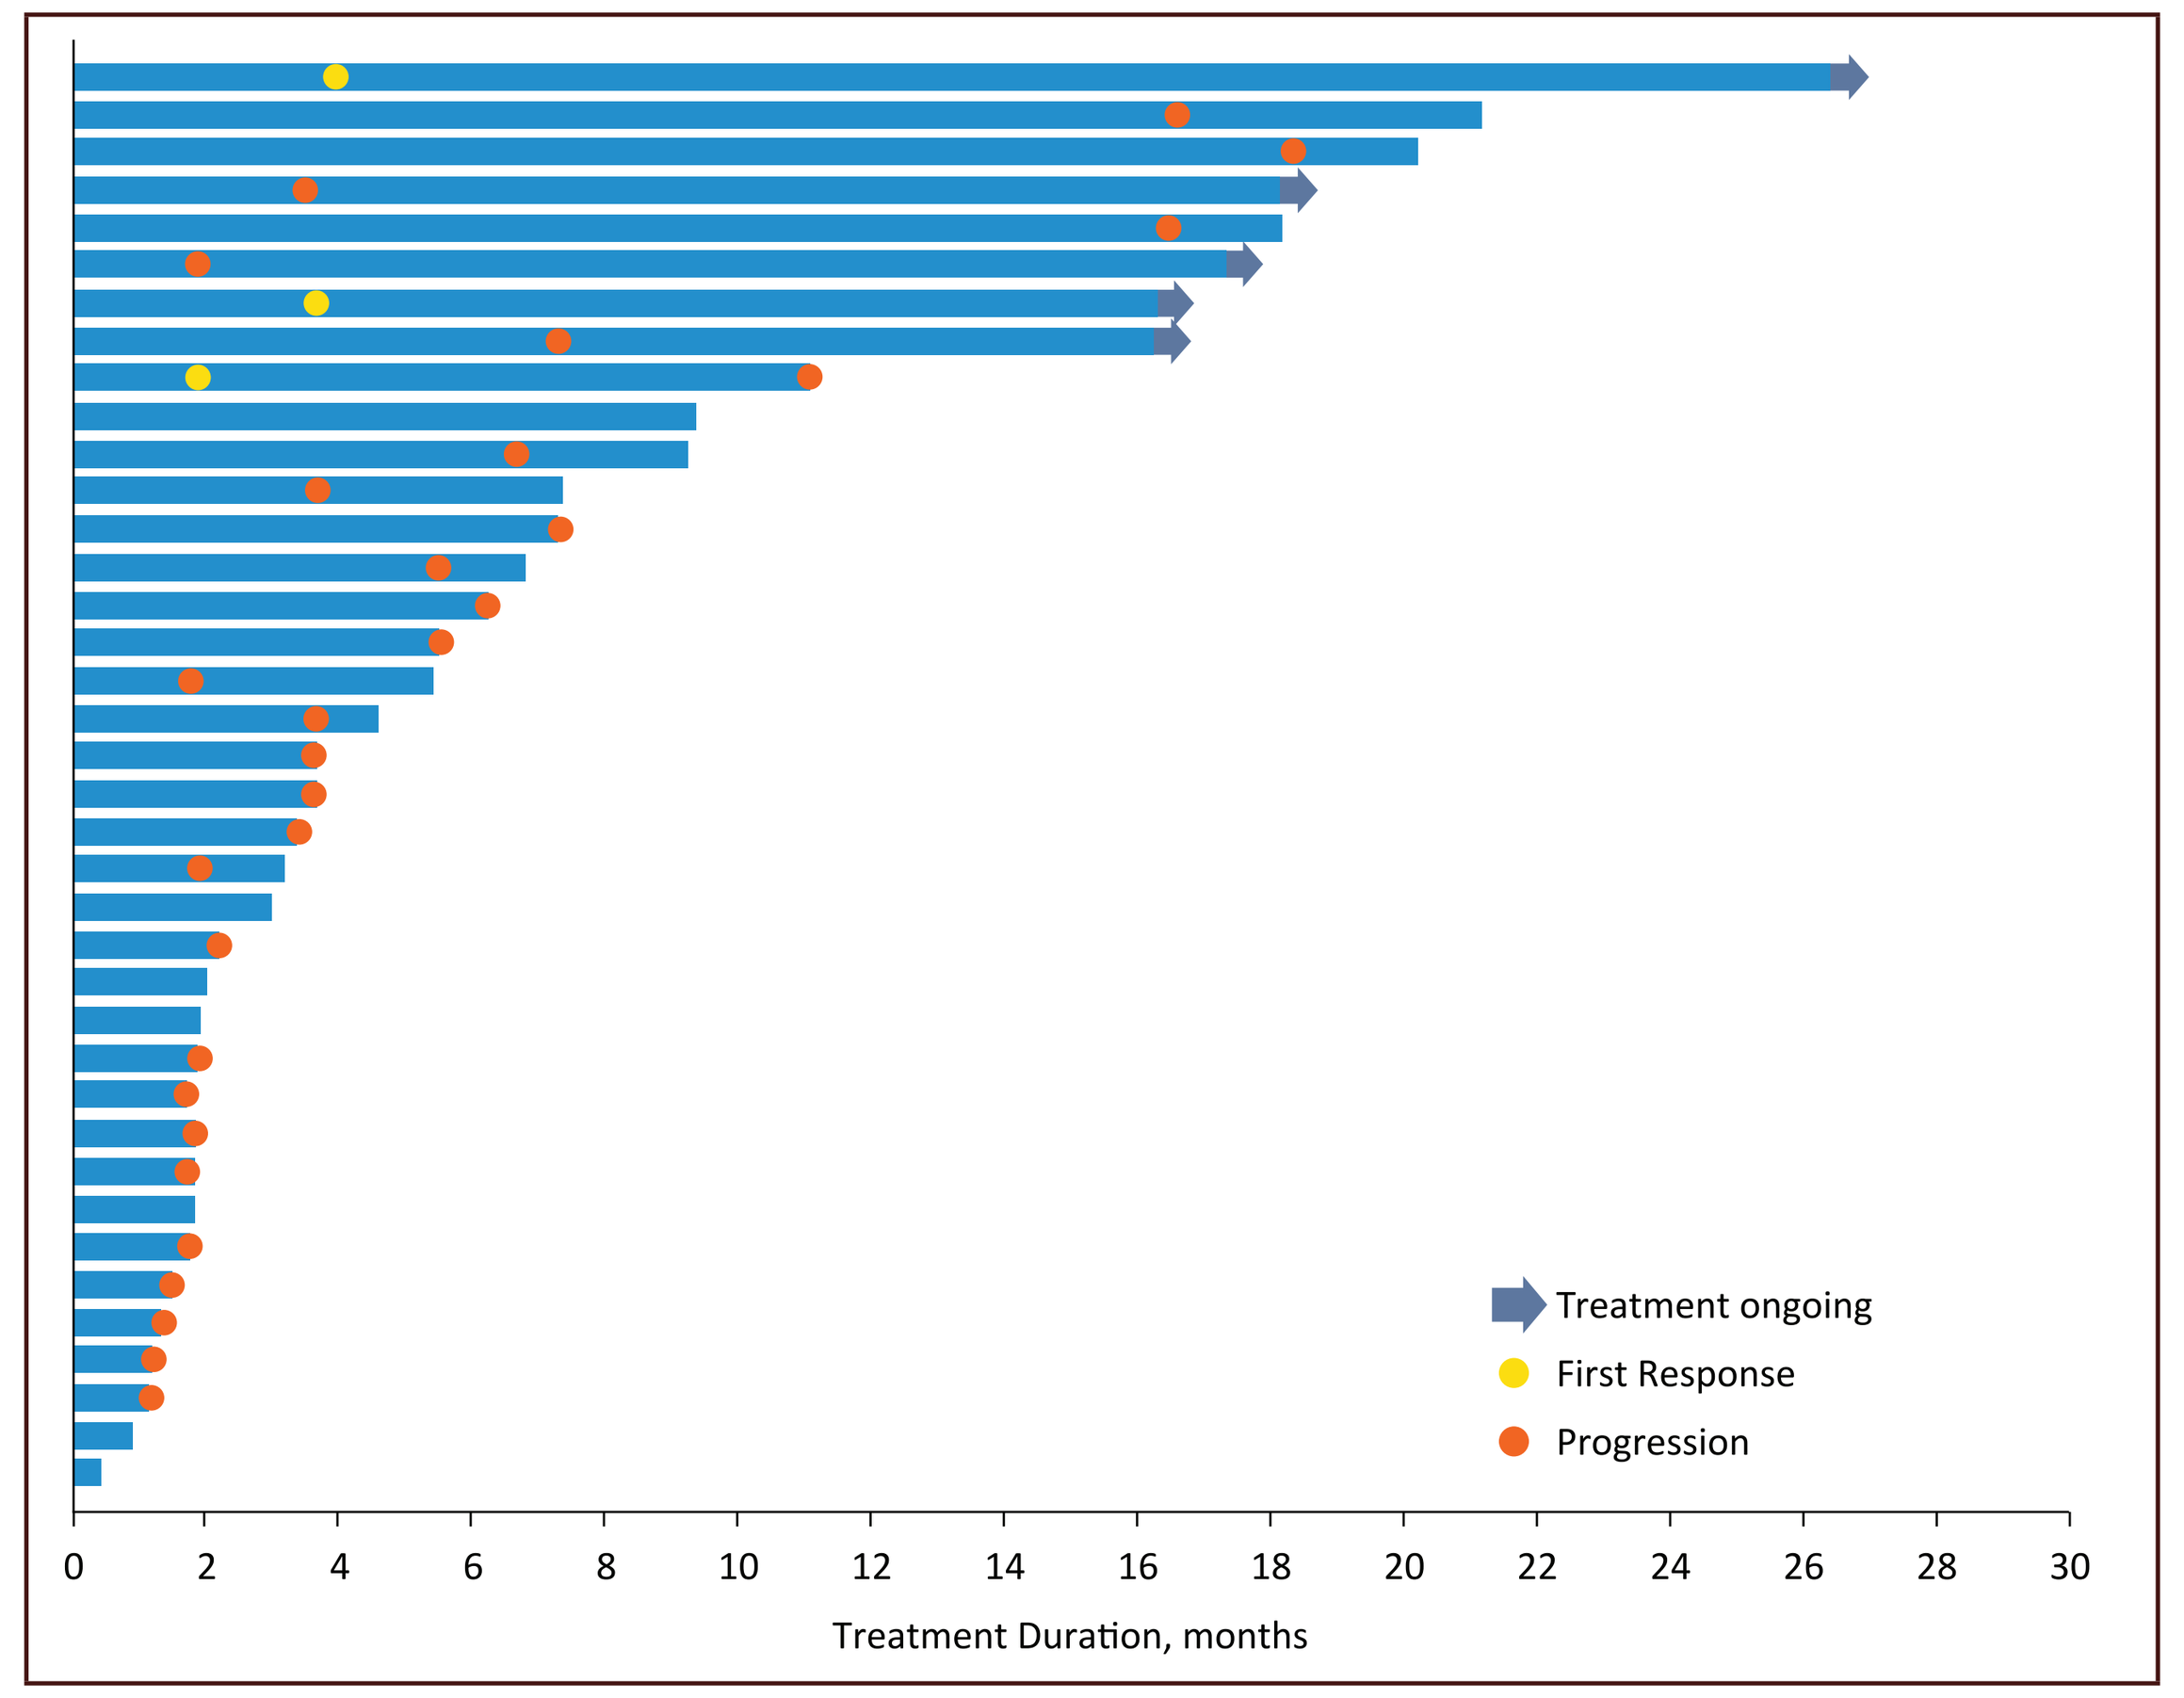
Treatment ongoing
First Response
Progression
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28 30
Treatment Duration, months

Figure 5. Progression-free survival and overall survival based on prior treatment status

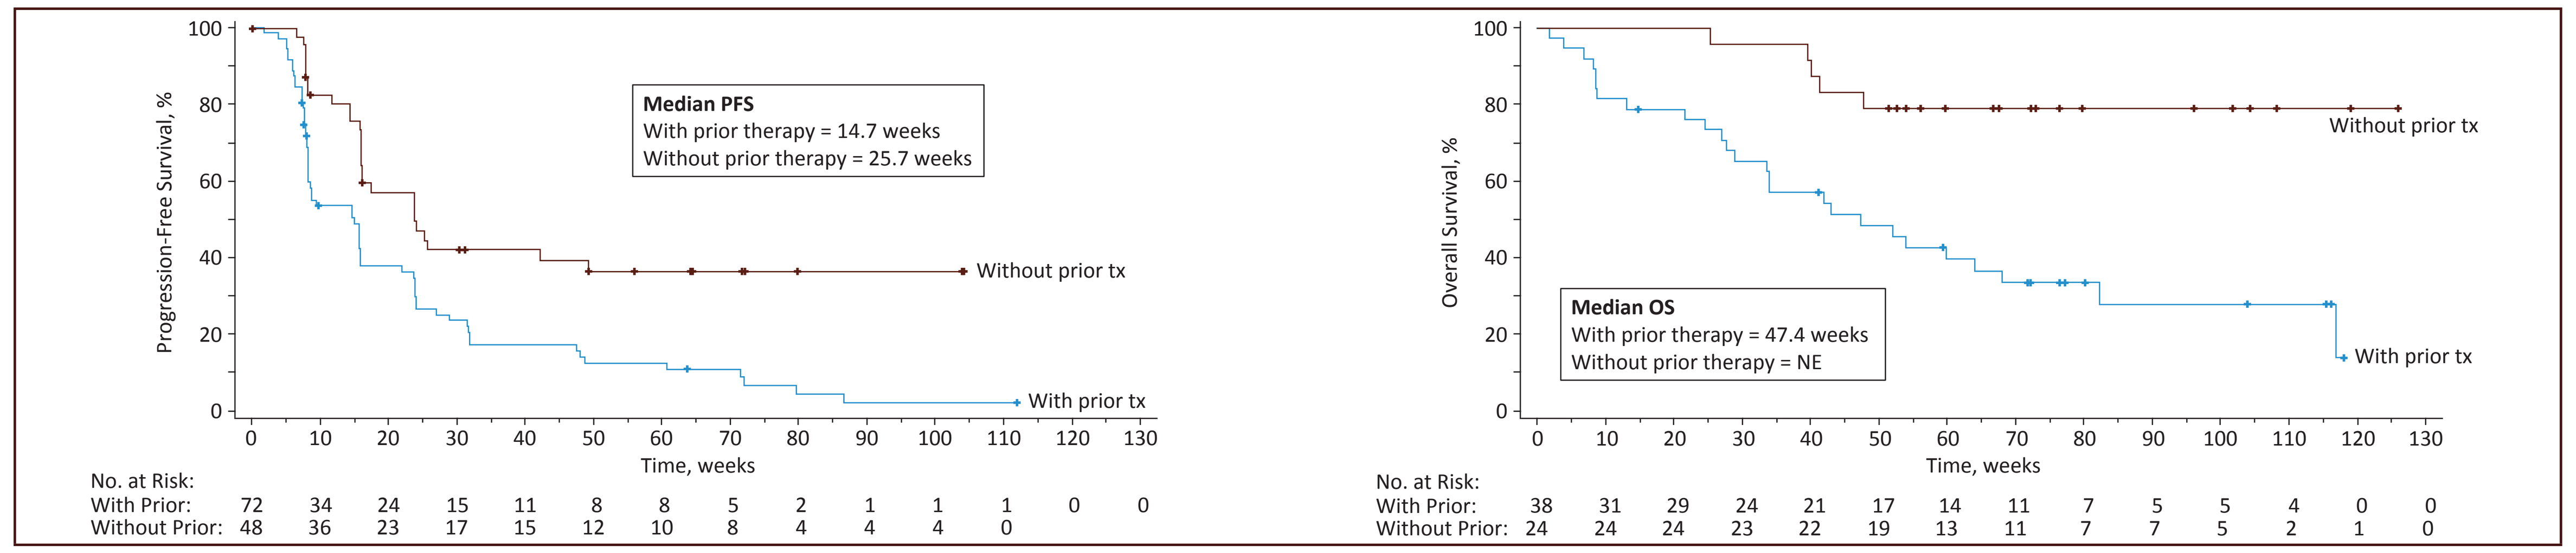
Progression-Free Survival, %
Median PFS
With prior therapy = 14.7 weeks
Without prior therapy = 25.7 weeks
Without prior tx
With prior tx
Time, weeks
No. at Risk:
With Prior: 72 34 24 15 11 8 8 5 2 1 1 1 0 0
Without Prior: 48 36 23 17 15 12 10 8 4 4 4 0
Overall Survival, %
Median OS
With prior therapy = 47.4 weeks
Without prior therapy = NE
Without prior tx
With prior tx
Time, weeks
No. at Risk:
With Prior: 38 31 29 24 21 17 14 11 7 5 5 4 0 0
Without Prior: 24 24 24 23 22 19 13 11 7 7 5 2 1 0

+, censored; NE, not estimable; OS, overall survival; PFS, progression-free survival; TX, therapy.

Safety

- The most common TEAEs included fatigue (37%), nausea (35%), and cancer pain (32%) (Table 4)
 - Treatment-related TEAEs grade ≥3 reported in ≥2 patients were anemia (6%) and decreased weight (3%)

Table 4. TEAEs occurring in ≥10% of patients

Adverse event, n (%)	Patients (N=62)			
	All TEAEs		Treatment-related TEAEs	
	All grades	Grade ≥3	All grades	Grade ≥3
Fatigue	23 (37)	1 (2)	16 (26)	1 (2)
Nausea	22 (35)	0	16 (26)	0
Cancer pain	20 (32)	3 (5)	3 (5)	0
Decreased appetite	16 (26)	3 (5)	10 (16)	1 (2)
Constipation	13 (21)	0	5 (8)	0
Vomiting	15 (24)	0	10 (16)	0
Diarrhea	10 (16)	0	8 (13)	0
Cough	11(18)	0	0	0
Anemia	10 (16)	8 (13)	6 (10)	4 (6)
Dyspnea	8 (13)	4 (6)	0	0
Headache	11 (18)	0	4 (6)	0
Weight decreased	10 (16)	4 (6)	8 (13)	2 (3)
Pleural effusion	7 (11)	4 (6)	0	0

[a]Ten patients experienced TEAEs that were uncoded at the time of the datacut.
TEAE, treatment-emergent adverse event.

CONCLUSION

- This was the largest prospective clinical trial of ES to date
- Tazemetostat showed promising single agent activity, resulting in durable confirmed responses (13%) and a DCR of 24%
- Tazemetostat demonstrated favorable safety with no patients discontinuing due to an adverse event and very few patients experiencing grade ≥3 TEAEs related to treatment
- As ES is an aggressive sarcoma subtype, and currently available treatments for metastatic STS have been shown to have limited activity, tazemetostat represents a promising potential therapeutic option

REFERENCES

1. Jacobs AJ, et al. *Sarcoma*. 2015:279601.
2. Armah HB, Parwani AV. *Arch Pathol Lab Med*. 2009;133:814–819.
3. Jawad MU, et al. *Clin Orthop Relat Res*. 2009;467:2939–2948.
4. Hollman TJ, Hornick JL. *Am J Surg Pathol*. 2011;35:e47–e63.
5. Wilson BG, et al. *Cancer Cell*. 2010;18:316–328.
6. Shain AH, et al. *Proc Natl Acad Sci USA*. 2012;109:E252–E259.
7. Wang X, et al. *Nat Genet*. 2017;49:289–295.
8. Knutson SK, et al. *Proc Natl Acad Sci USA*. 2013;110:7922–7927.
9. Italiano A et al. *The Lancet Oncology*. 2018;19:649–659.
10. Jones RL, et al. Presented at ESMO 2018.

ACKNOWLEDGMENTS

- We thank all of the physicians, study coordinators, site staff and most of all, the patients, caregivers, and families who have contributed to this study
- Third-party writing assistance was provided by Ashfield Healthcare and funded by Epizyme, Inc.

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission from the author of this poster.

Mrinal Gounder, gounderm@mskcc.org

Presented at the 2018 Annual Meeting of the European Society of Medical Oncology (ESMO); 19–23 October 2018; Munich, Germany