PS1247

EZH2 Gain-of-Function Mutations Are Not Associated With More Favorable Prognosis in Relapsed/Refractory Follicular Lymphoma: A Preliminary Analysis on 908 Patients

Csaba Bödör[1], Connie Batlevi[2], Noémi Nagy[1], Jean-Marie Michot[3], Tamás Schneider[4], Hussain Alizadeh[5], Zsofia Simon[6], Julie Vose[7], Anas Younes[2], Vincent Ribrag[3], Jude Fitzgibbon[8], Jay Yang[9], Shefali Agarwal[9], Kate J. Newberry[9], Neil R. Michaud[9], Jessica Okosun[8]

[1]Semmelweis University, Budapest, Hungary; [2]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [3]Gustave Roussy, Villejuif, France; [4]National Institute of Oncology, Budapest, Hungary; [5]University of Pecs, Pecs, Hungary; [6]University of Debrecen, Debrecen, Hungary; [7]University of Nebraska Medical Center, Omaha, NE, USA; [8]Barts Cancer Institute, Queen Mary University of London, London, UK; [9]Epizyme, Cambridge, MA, USA

BACKGROUND

- EZH2 is an epigenetic regulator of gene expression and plays a critical role in multiple forms of cancer
 - Activating mutations of *EZH2* act as an oncogenic driver, especially in follicular lymphoma (FL), and are present in ~20–25% of patients[1,2]
- Small published studies suggest *EZH2* gain of function mutations may provide a prognostic benefit in the frontline setting (1L) in FL patients treated with immunochemotherapy regimens[3,4]
- However, one study suggested the impact of *EZH2* mutations over the long-term were lost when overall survival (OS) was evaluated[2]
- Tazemetostat, an investigational, selective, oral inhibitor of the histone methyltransferase EZH2, has shown antitumor activity in patients with relapsed and/or refractory (R/R) FL treated with greater than two prior lines of therapy (3L+). It is unknown whether *EZH2* mutations confer prognostic benefits in this setting
- This multi-center study is intended to evaluate the impact of *EZH2* activating mutations on outcomes in R/R FL patients from 5 academic sites. Results of an interim analysis from 4 (of 5) academic sites are presented
 - Best objective response rate (ORR) and progression-free survival (PFS) were assessed across multiple lines of therapy in R/R FL patients receiving systemic anticancer therapy. OS from time of diagnosis was also determined

METHODS

- Retrospective data on therapy types and clinical outcomes were collected from 4 academic sites
 - Barts Cancer Institute, London
 - Institute Gustave Roussy, Paris
 - Memorial Sloan Kettering Cancer Center, New York
 - Semmelweis University, Budapest
- Tumor tissues collected at the time of diagnosis, in most cases, were analyzed for activating mutations of *EZH2* (Y646X, A682G, A692V) using the following approaches:
 - Next generation sequencing, including use of targeted exome sequencing or a targeted gene panel such as the MSK-IMPACT panel
 - Digital droplet PCR (BioRad)
- Data were analyzed to compare clinical outcome parameters between R/R FL patients with mutant (MT) *EZH2* and wild-type (WT) *EZH2*
 - Best ORR, based on each center's response criteria, was compared by *EZH2* status and stratified by line of therapy using the Cochran-Mantel-Haenszel chi-square test
 - Best responses were assessed by the treating physician at each site based on the site's criteria of clinical response (rather than by International Working Group criteria)
 - PFS and OS were calculated using the Kaplan-Meier method and compared by the logrank test

RESULTS

Patient characteristics

- Data from 908 R/R FL patients (MT *EZH2*, n=193; WT *EZH2*, n=713; *EZH2* status unknown, n=2) treated with systemic anticancer therapy between December 1972 and December 2017 at 4 academic centers were included for analyses
 - Median follow-up from diagnosis to last follow up or death in all patients was 8.92 years (range, 0.03–38.52 years) (Table 1)
- The frequency of *EZH2* activating mutations was 21%. The mutation frequency at each hotspot residue was:
 - 81% for Y646(H,N,S,C,F)
 - 11% for A682G
 - 8% for A692V
- The vast majority of patients received systemic anticancer therapy (Table 1), including a range of chemotherapeutic regimens that often included rituximab (immunochemotherapy) (Table 2)
 - In some cases, patients may have also received radiotherapy and/or stem cell transplantation. These therapeutic regimens were not included as therapy lines in the analysis
- In 1L, most patients received combination immunochemotherapy (56%). A substantial number of patients received combination chemotherapy (37%) (Table 2)
 - Use of immunochemotherapy decreased in second line therapy (2L) and beyond (2L+) such that only 22% of 4L patients received rituximab-based chemotherapy
 - Use of combination chemotherapy alone was >40% in 2L+
 - Other therapy types, including anti-CD20 monotherapy and various targeted therapies, were employed in later lines of therapy

Table 1. Baseline Demographics

Characteristics	Total Number of Patients (N=908)
Age (years), median (range)	54 (21–90)
Males/Females, n (%)	453 (50) / 455 (50)
ECOG performance status 0–1, n (%)	414 (46)
ECOG performance status ≥ 2, n (%)	43 (5)
ECOG performance status NA or missing, n (%)	451 (50)
Patients receiving each therapy type at any time, n (%)	
Systemic anticancer therapy	805 (89)
Stem cell transplantation	190 (21)
Radiotherapy	270 (30)

ECOG, Eastern Cooperative Oncology Group; NA, not applicable.

Patient Outcomes

- No significant differences (P>0.05) were observed in ORR between MT and WT *EZH2* cohorts in 1L, 2L, or 3L+ (Table 4)
 - Differences in treatment regimens, methods of evaluating clinical response, regional differences in patient populations, and differences in the era in which patients were treated make it challenging to evaluate ORR in these patients
- There were no significant differences (P>0.05) in PFS between MT and WT *EZH2* FL patients for any line of therapy in the R/R setting (Figure 1, Table 3)
 - In the frontline setting, median PFS was improved in MT *EZH2* FL patients (3.27 years, 95% CI 2.38–6.77) compared with WT *EZH2* FL patients (2.52 years, 95% CI 2.18–2.91, P=0.0181; hazard ratio=1.31, 95% CI 1.05–1.64, P=0.0186)
- Further, there was no significant difference in median OS between FL patients with MT and WT *EZH2* (OS 13.31 vs 16.83 years, respectively; P=0.6580) (Figure 2)
- The frequency of *EZH2* mutations was consistent across all lines of therapy (range, 19–22%)

Table 2. Systemic anticancer therapy types by line of therapy

Line of Therapy	MT *EZH2* (n=193)[1]	WT *EZH2* (n=713)[1]
1L, n[2] (%)	n=178	n=625
Chemotherapy	69 (39)	228 (36)
Immunochemotherapy[3]	100 (56)	348 (56)
Targeted therapy	1 (<1)	3 (<1)
Anti-CD20 monotherapy	7 (4)	35 (6)
Other[4]	1 (<1)	11 (2)
2L, n[2] (%)	n=102	n=411
Chemotherapy	47 (46)	160 (39)
Immunochemotherapy[3]	37 (36)	167 (41)
Targeted therapy	0	7 (2)
Anti-CD20 monotherapy	6 (6)	24 (6)
Other[4]	12 (12)	53 (12)
3L, n[2] (%)	n=77	n=297
Chemotherapy	41 (53)	124 (42)
Immunochemotherapy[3]	17 (22)	91 (31)
Targeted therapy	2 (3)	20 (7)
Anti-CD20 monotherapy	6 (8)	22 (7)
Other[4]	11 (14)	40 (13)
4L, n[2] (%)	n=50	n=215
Chemotherapy	36 (72)	94 (44)
Immunochemotherapy[3]	5 (10)	52 (24)
Targeted therapy	2 (4)	14 (7)
Anti-CD20 monotherapy	1 (2)	19 (9)
Other[4]	6 (12)	36 (16)

[1]n=number of patients defined as either MT or WT *EZH2*, respectively, who received systemic anticancer therapy.
[2]n=total number of patients defined as MT or WT *EZH2* who received the specified line of systemic anticancer therapy.
[3]Anti-CD20 antibody + chemotherapy (eg, CHOP).
[4]Other=all other types of systemic anticancer therapy that could not be defined as immunochemotherapy, chemotherapy, targeted agent, or anti-CD20 monotherapy (eg, radioimmunotherapy, immunomodulatory agent, antibody-drug conjugate, etc).
L, line; MT, mutant; WT, wild-type.

Figure 1. PFS by line of therapy for FL patients with MT vs WT EZH2

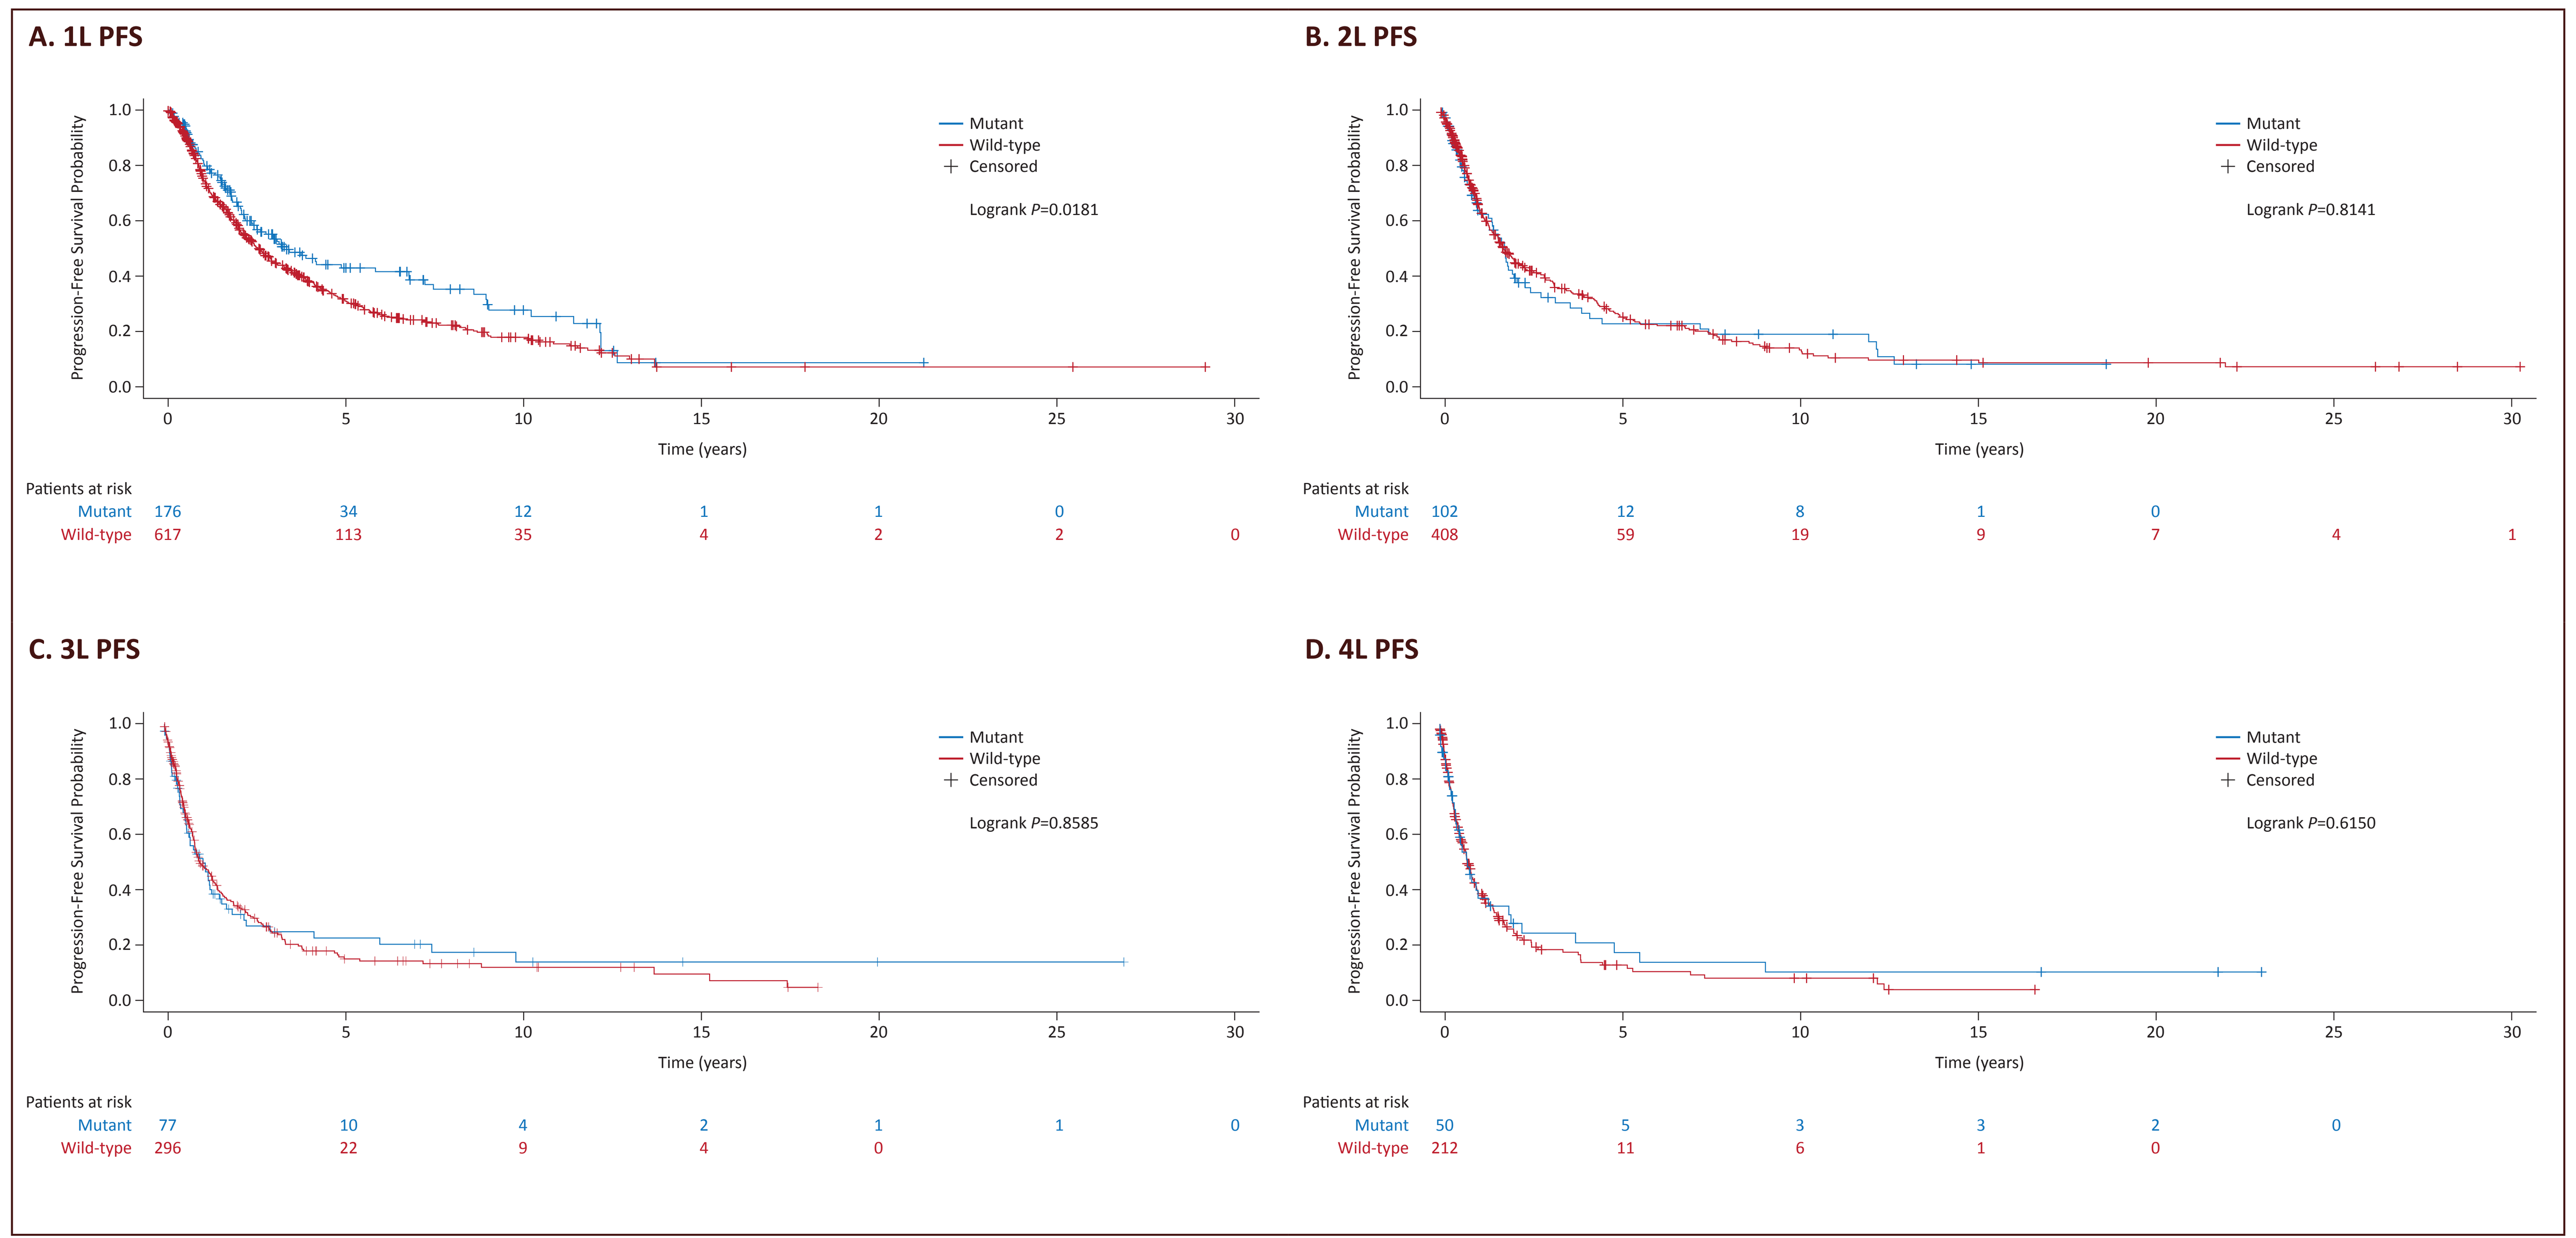

A. 1L PFS
Progression-Free Survival Probability
Mutant
Wild-type
Censored
Logrank P=0.0181
Time (years)
Patients at risk
Mutant 176 34 12 1 1 0
Wild-type 617 113 35 4 2 2 0
B. 2L PFS
Logrank P=0.8141
Patients at risk
Mutant 102 12 8 1 0
Wild-type 408 59 19 9 7 4 1
C. 3L PFS
Logrank P=0.8585
Patients at risk
Mutant 77 10 4 2 1 1 0
Wild-type 296 22 9 4 0
D. 4L PFS
Logrank P=0.6150
Patients at risk
Mutant 50 5 3 3 2 0
Wild-type 212 11 6 1 0

In each panel, the total number of MT and WT *EZH2* patients evaluable for PFS is shown at time=0 for that line of therapy.
L, line; MT, mutant; PFS, progression-free survival; WT, wild-type.

Table 3. PFS for FL patients with MT and WT EZH2 receiving systemic anticancer treatment by line of therapy

Line of Therapy	MT *EZH2*	WT *EZH2*	*P*-value
1L	n=178[1]	n=625[1]	0.0181
Median PFS, years	3.27	2.52	
95% CI, years	2.38, 6.77	2.18, 2.91	
2L	n=102[1]	n=411[1]	0.8141
Median PFS, years	1.77	1.73	
95% CI, years	1.32, 2.12	1.46, 2.08	
3L	n=77[1]	n=297[1]	0.8585
Median PFS, years	1.07	0.99	
95% CI, years	0.61, 1.53	0.84, 1.33	
4L	n=50[1]	n=215[1]	0.6150
Median PFS, years	0.77	0.76	
95% CI, years	0.44, 1.35	0.62, 0.95	

[1]n is defined as the total number of MT or WT *EZH2* patients who received that respective line of systemic anticancer therapy. In a small number of cases, it was not possible to calculate PFS, which is reflected in the smaller number of patients at time=0 for MT and/or WT *EZH2* for some lines of therapy in Figure 1 than depicted in Table 3.
CI, confidence interval; L, line; PFS, progression-free survival; MT, mutant; WT, wild-type.

Figure 2. OS for FL patients with MT vs WT EZH2

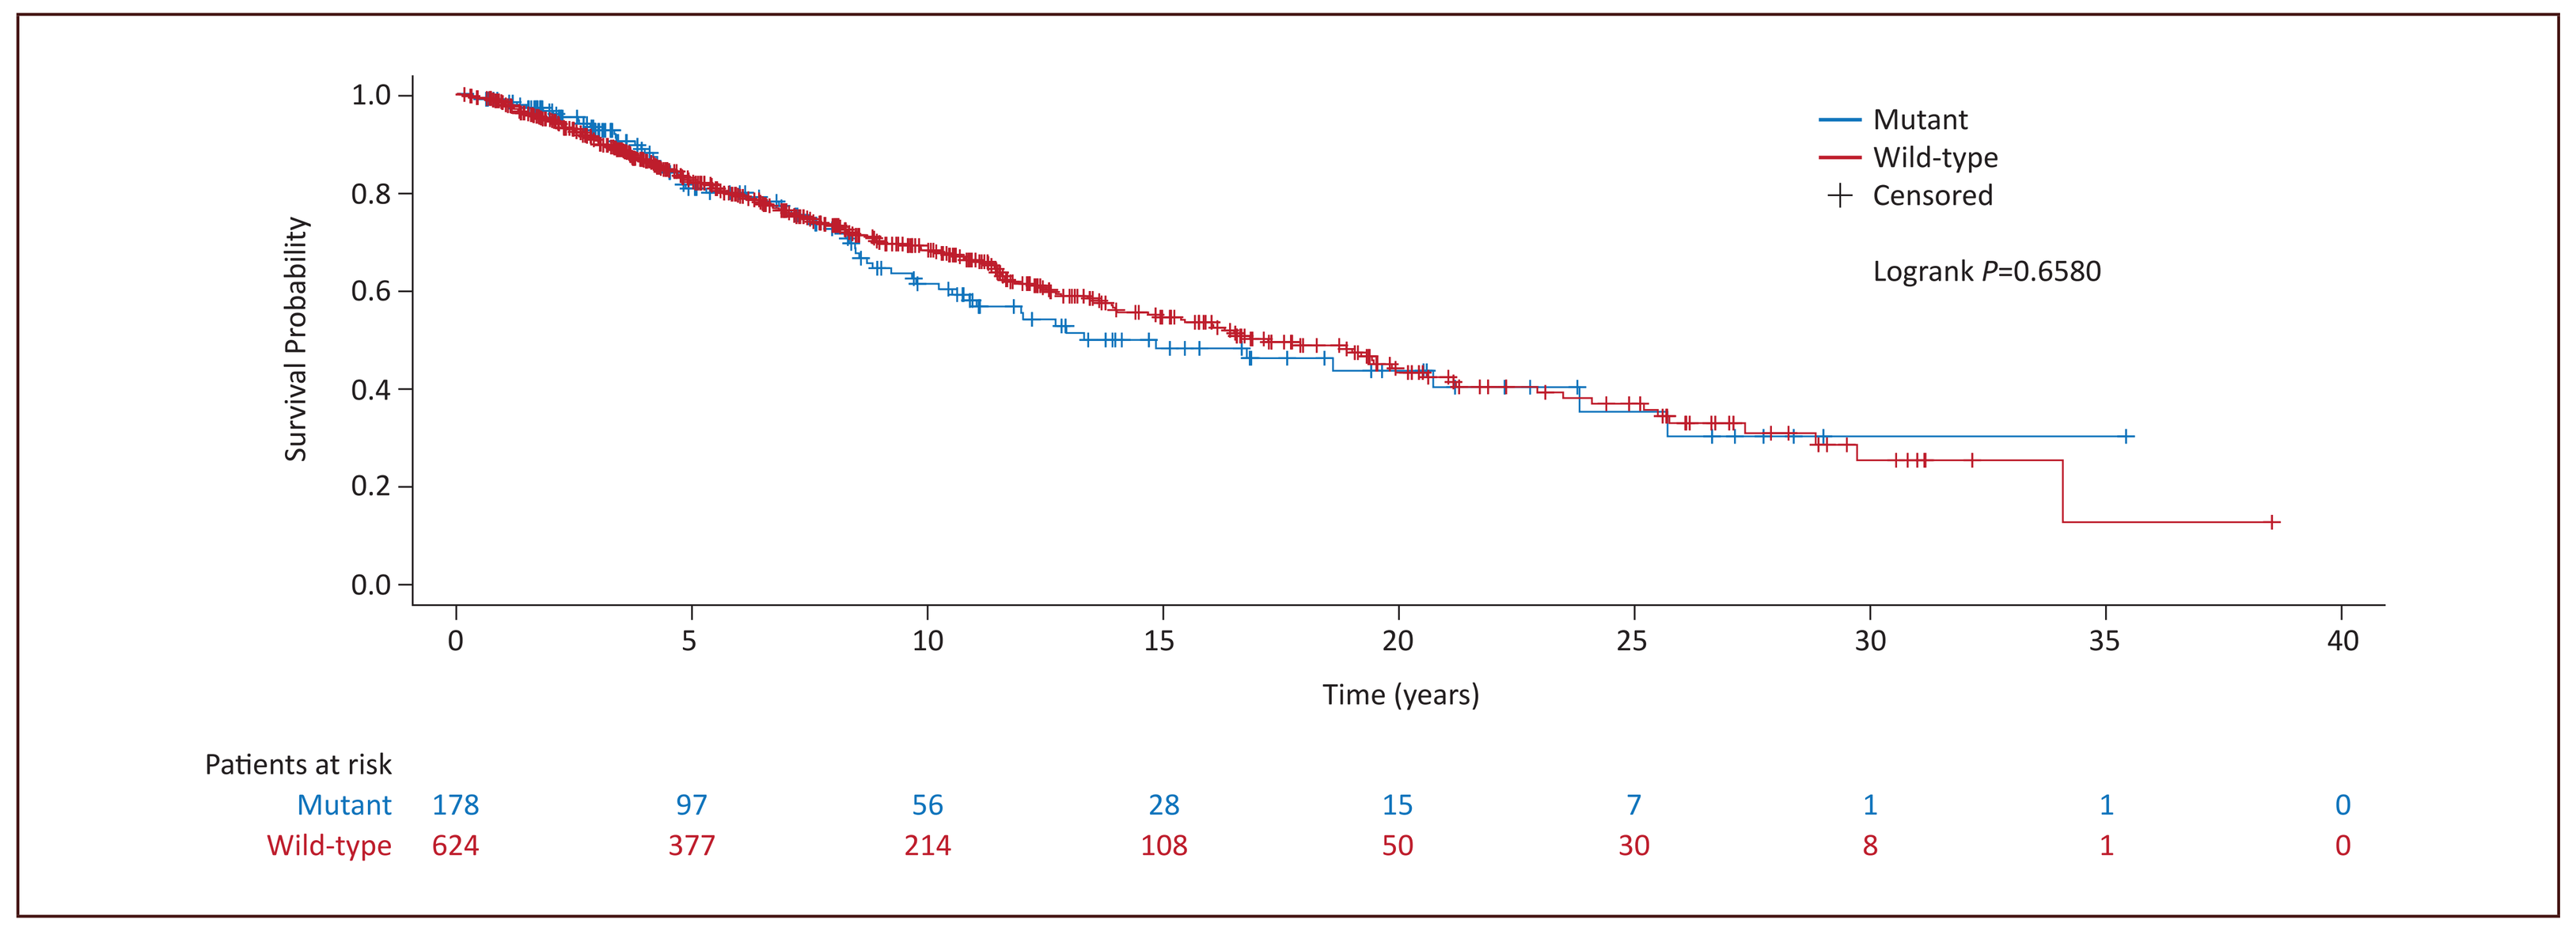

Survival Probability
Mutant
Wild-type
Censored
Logrank P=0.6580
Time (years)
Patients at risk
Mutant 178 97 56 28 15 7 1 1 0
Wild-type 624 377 214 108 50 30 8 1 0

The number of MT and WT *EZH2* patients evaluable to determine OS from time of diagnosis to death or time of last follow up is shown at time=0.
FL, follicular lymphoma; MT, mutant; OS, overall survival; WT, wild-type.

Table 4. ORR for FL patients with MT and WT EZH2 across lines of therapy

All Sites	MT *EZH2*, # Responders/# Treated (%)	WT *EZH2*, # Responders/# Treated (%)	All, # Responders/# Treated (%)	*P*-value
1L	162/178 (91)	540/625 (86)	704/805 (87)	0.1019
2L	75/102 (74)	290/411 (71)	365/515 (71)	0.5539
3L+[1]	66/77 (86)	241/297 (81)	308/376 (82)	0.3521

[1]The # Responders is the sum of all patients who responded during any line of therapy, including 3L or any subsequent line of therapy.
L, line; MT, mutant; ORR, objective response rate; WT, wild-type.

CONCLUSIONS

- ORR was similar between patients with MT *EZH2* compared with WT *EZH2* in all lines of therapy
- Analysis of OS indicates that there are no significant differences between MT and WT *EZH2* R/R FL patients followed for more than 30 years
- Improved PFS was observed in 1L patients with MT *EZH2* compared with WT *EZH2*, similar to results reported by Pastore et al.[3] and Huet et al.[4]
 - However, MT *EZH2* did not act as a positive prognostic factor in R/R FL patients treated in 2L+
- Based on this large retrospective analysis, these data suggest that the clinical activity observed in R/R FL patients with standard of care agents or tazemetostat is due to the drugs' mechanisms of action and not due to a prognostic effect of *EZH2* mutation

REFERENCES

1. Morin RD, et al. Nature Genetics. 2010;42(2):181–5.
2. Bödör C, et al. Blood. 2013;122(18):3165–8.
3. Pastore A, et al. The Lancet Oncology. 2015;16(9):1111–22.
4. Huet S, et al. Blood Cancer Journal. 2017;7(4):e555.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission.

Presented at the 24th European Hematology Association Congress; June 13–16, 2019; Amsterdam, The Netherlands