10525

Phase 1 Study of Tazemetostat, an Enhancer of Zeste Homolog-2 Inhibitor, in Pediatric Patients With Relapsed/Refractory Integrase Interactor 1-Negative Tumors

Susan N. Chi[1], Franck Bourdeaut[2], Theodore W. Laetsch[3], Maryam Fouladi[4], Margaret Macy[5], Guy Makin[6], Neerav Shukla[7], Cynthia Wetmore[8], Ashley Margol[9], Michela Casanova[10], Lindsay Kilburn[11], Joanna Yi[12], Darren Hargrave[13], Geoff McCowage[14], Navin Pinto[15], Dave Ebb[16], Giles Robinson[17], Laura Sierra[18], Melinda Merchant[18], Karsten Nysom[19]

[1]Dana-Farber Cancer Institute, Boston, MA, USA; [2]Curie Institute, Paris, France; [3]University of Texas Southwestern Medical Center and Children's Health, Dallas, TX, USA; [4]Cincinnati Children's Hospital, Cincinnati, OH, USA; [5]Children's Hospital Colorado, Aurora, CO, USA; [6]University of Manchester, Manchester, England, UK; [7]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [8]Phoenix Children's Hospital, Phoenix, AZ, USA; [9]Children's Hospital Los Angeles, Los Angeles, CA, USA; [10]Fondazione IRCCS Istituto Nazionale Tumori, Milano, Italy; [11]Children's National Hospital, Washington, DC; [12]Texas Children's Hospital, Houston, TX, USA; [13]Great Ormond Street Hospital for Children, London, England, UK; [14]Cancer Centre for Children at The Children's Hospital at Westmead, Westmead, New South Wales, Australia; [15]Seattle Children's Hospital, Seattle, WA, USA; [16]Massachusetts General Hospital, Boston, MA, USA; [17]St. Jude Children's Research Hospital, Memphis, TN, USA; [18]Epizyme, Cambridge, MA, USA; [19]Rigshospitalet, Copenhagen, Denmark

BACKGROUND

- Absence of integrase interactor 1 (INI1) expression is a defining molecular feature of rhabdoid tumors (RT; including atypical teratoid RTs [ATRT]), epithelioid sarcoma (ES), and chordomas. INI1 loss induces dependence on enhancer of zeste homolog-2 (EZH2).[1] These tumor types are often resistant to available therapies and associated with a poor prognosis. Specific tumor types include:
 - Malignant rhabdoid tumors (MRT) are aggressive forms of cancer associated with INI1 loss which were identified initially in the kidney (rhabdoid tumor of the kidney; RTK) or in the central nervous system (ATRT), but have also been identified in the liver and soft tissue[2,3]
 - ES are aggressive soft tissue tumors that can exhibit multifocal disease at presentation, local recurrence, and regional metastasis, and have a high rate of relapse[4]
 - Chordomas with INI1 loss occur in the skull base and spine, are poorly differentiated, and are associated with frequent metastasis and short survival time[5]
 - INI1 loss or mutations in SMARCB1 on the long arm of chromosome 22 have also been identified in molecular analysis of other tumor types, such as synovial sarcoma, renal medullary carcinoma, and myoepithelial carcinoma which were also included in this pediatric study
- Tazemetostat is a selective EZH2 inhibitor (**Figure 1**) recently approved by the FDA for treatment of patients ≥16 years with metastatic or locally advanced ES not eligible for complete resection[6,7]
- Data from a phase 1 (Ph1) pediatric dose-escalation study (Ph1a) of tazemetostat were previously reported;[8] herein we report interim safety and efficacy data from the Ph1 pediatric dose-expansion study (Ph1b)

Figure 1. Structure and Mechanism of Action of Tazemetostat

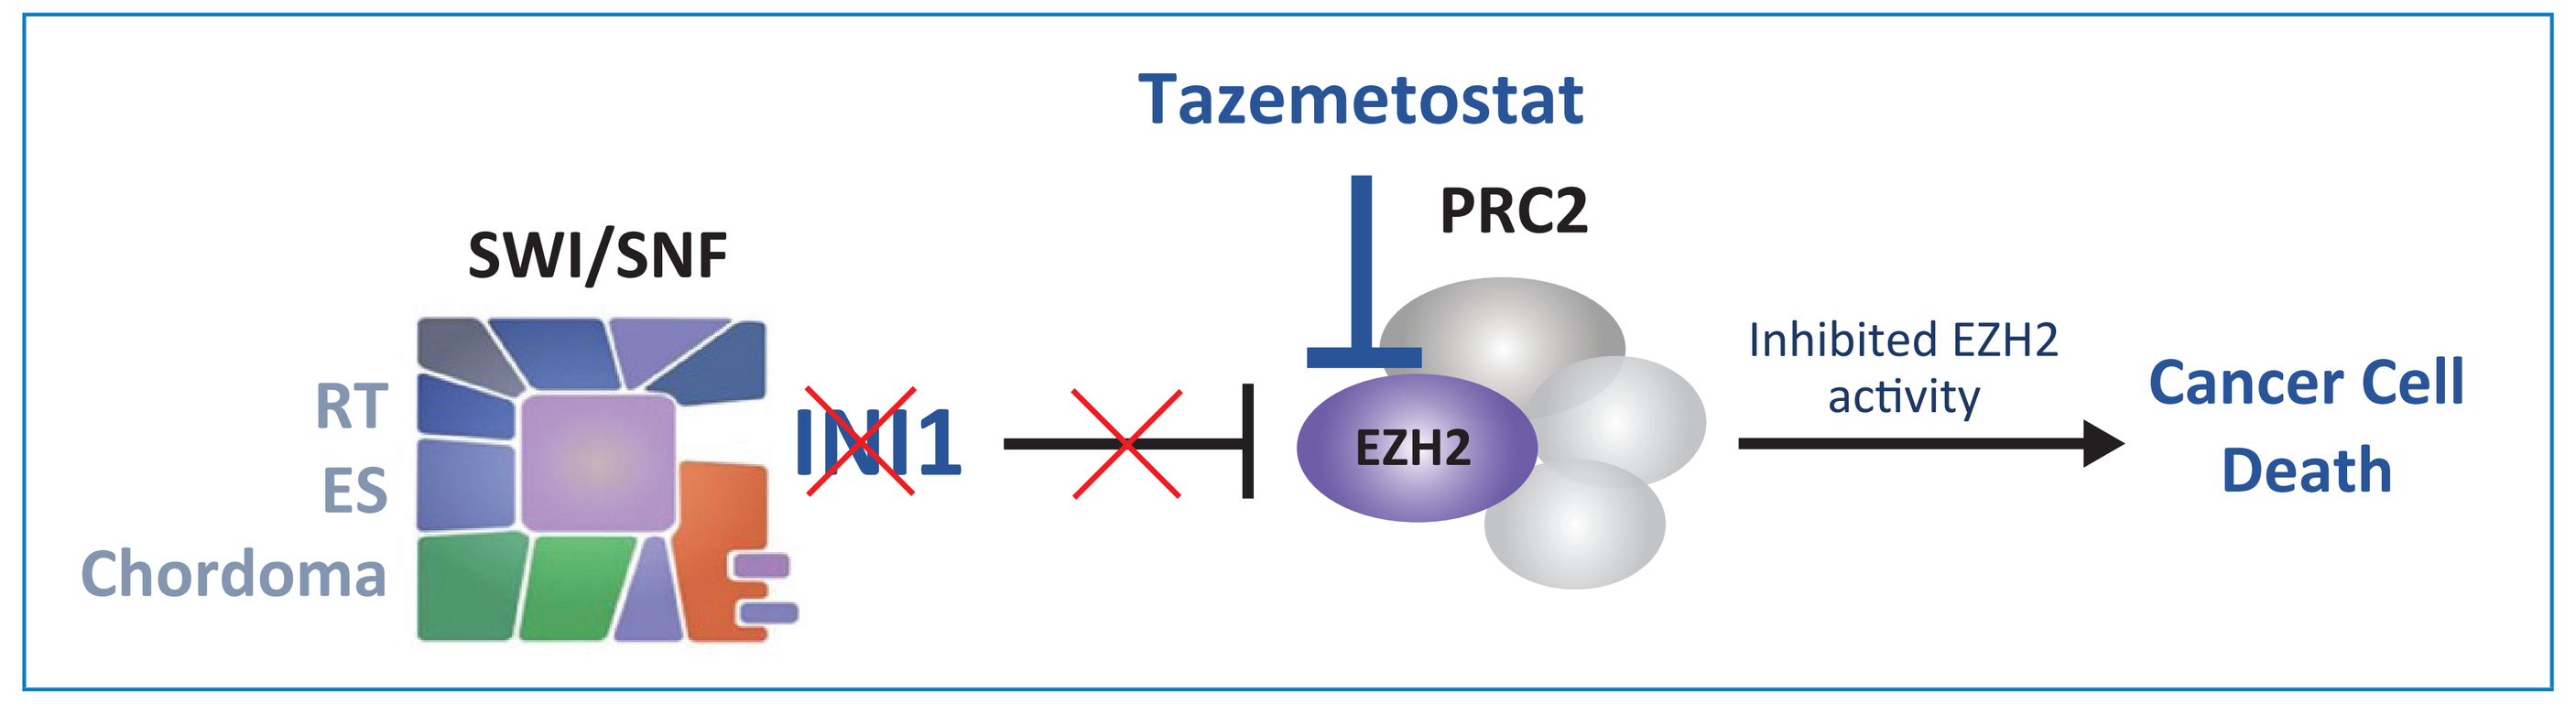

Tazemetostat
PRC2
SWI/SNF
RT
ES
Chordoma
INI1
EZH2
Inhibited EZH2 activity
Cancer Cell Death

ES, epithelioid sarcoma; EZH2, enhancer of zeste homolog-2; INI1, integrase interactor 1; PRC2, polycomb repressive complex 2; RT, rhabdoid tumor; SWI/SNF, SWItch/Sucrose Non-Fermentable.

METHODS

- EZH-102 (NCT02601937) is a Ph1, multicenter study in patients 6 months to <18 years evaluating tazemetostat administered twice a day (BID) at 1200 mg/m^2 in Ph1b, per Ph1a recommendation (**Figure 2**)
- Ph1b enrolled patients based on tumor type; all received tazemetostat as an oral suspension (Cohorts 1–3) or tablet (Cohort 4)
 - Cohort 1: ATRT
 - Cohort 2: non–central nervous system (CNS) MRT
 - Cohort 3: other INI1-negative tumors (including ES and de-differentiated chordoma)
 - Cohort 4: synovial sarcoma with SS18-SSX rearrangement OR tumor types eligible for Cohorts 1–3
- Key inclusion and exclusion criteria are provided in **Table 1**
- The Ph1b primary endpoint was overall response rate (ORR)
- Secondary endpoints included safety/tolerability, duration of response (DOR), and survival

Figure 2. Design of Study EZH-102

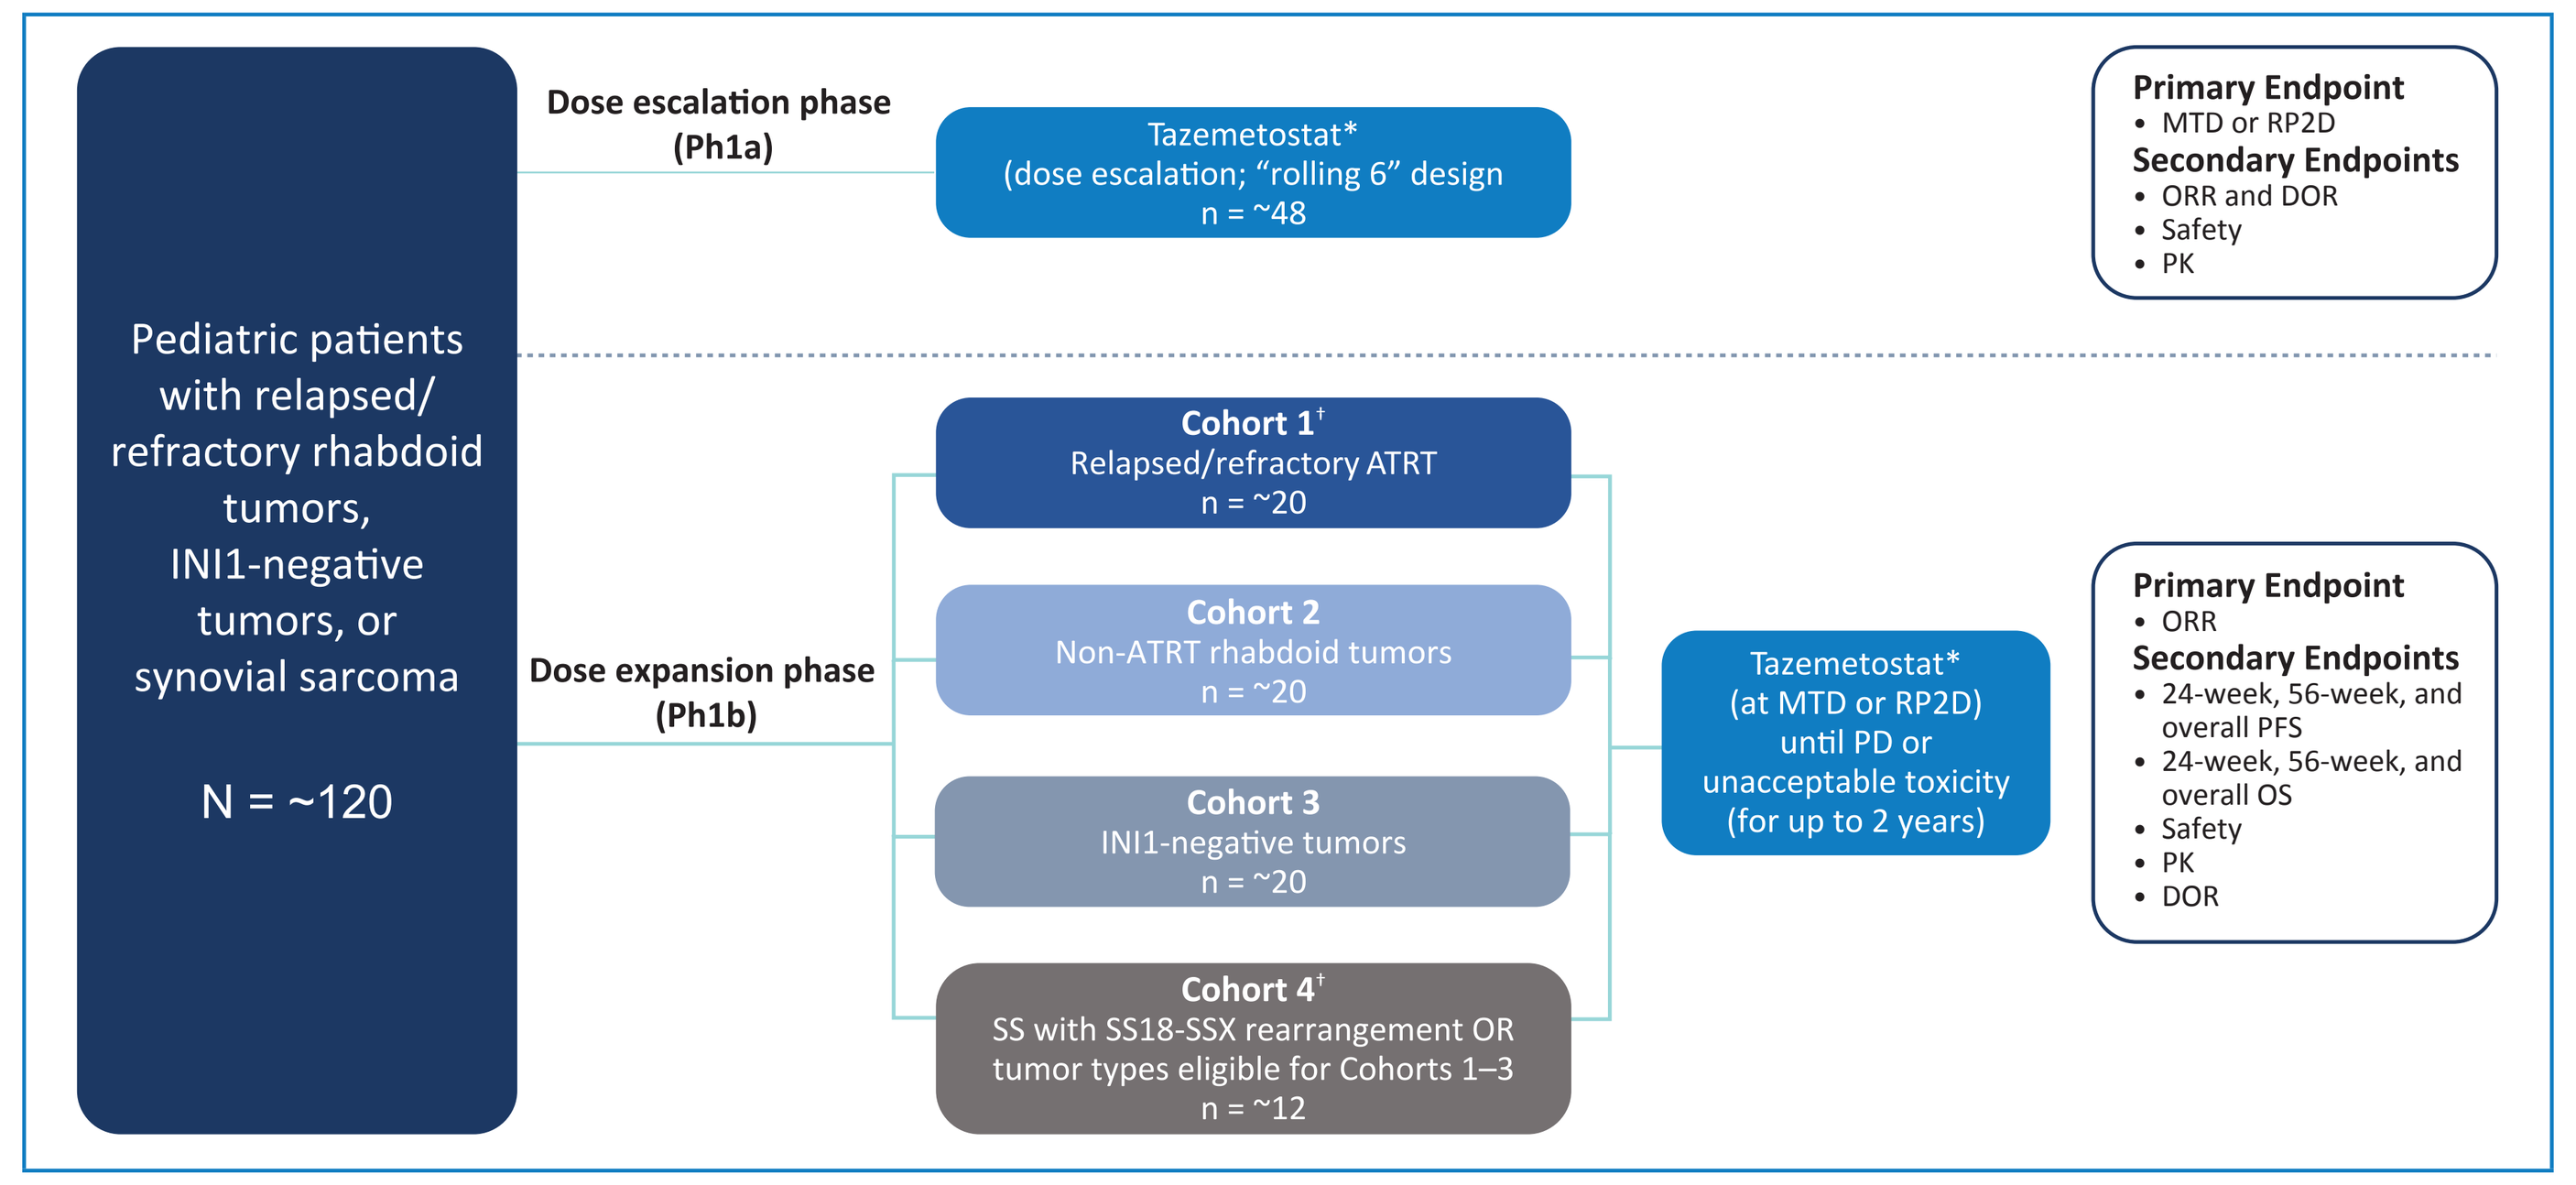

Pediatric patients with relapsed/ refractory rhabdoid tumors, INI1-negative tumors, or synovial sarcoma
N = ~120
Dose escalation phase (Ph1a)
Tazemetostat* (dose escalation; "rolling 6" design n = ~48
Primary Endpoint
• MTD or RP2D
Secondary Endpoints
• ORR and DOR
• Safety
• PK
Dose expansion phase (Ph1b)
Cohort 1† Relapsed/refractory ATRT n = ~20
Cohort 2 Non-ATRT rhabdoid tumors n = ~20
Cohort 3 INI1-negative tumors n = ~20
Cohort 4† SS with SS18-SSX rearrangement OR tumor types eligible for Cohorts 1–3 n = ~12
Tazemetostat* (at MTD or RP2D) until PD or unacceptable toxicity (for up to 2 years)
Primary Endpoint
• ORR
Secondary Endpoints
• 24-week, 56-week, and overall PFS
• 24-week, 56-week, and overall OS
• Safety
• PK
• DOR

*Cohort closed to enrollment.
†Tazemetostat administered as an oral suspension in Cohorts 1–3 and as tablets in Cohort 4 in the dose expansion phase.
ATRT, atypical teratoid rhabdoid tumor; DOR, duration of response; INI1, integrase interactor 1; MTD, maximum tolerated dose; ORR, overall response rate; OS, overall survival; PD, progressive disease; PFS, progression-free survival; PK, pharmacokinetics; RP2D, recommended phase 2 dose; RT, rhabdoid tumors.

Table 1. Key Eligibility Criteria

Key Inclusion Criteria	Key Exclusion Criteria
Age ≥6 months to <18 years of age	Prior exposure to tazemetostat or other EZH2 inhibitor, or known tazemetostat hypersensitivity
Relapsed/refractory disease with no standard treatment options, and ineligible/inappropriate for other treatment options with effective potential	History of chronic HBV or HCV infection, HIV infection, T-LBL/T-ALL, or MDS
Lansky or Karnofsky performance status >50%	Major surgery within 2 weeks prior to enrollment
Adequate hematologic, renal, and hepatic function	Clinically significant active cardiovascular disease
For subjects with INI1-negative tumors only: Morphology and immunophenotypic panel consistent with INI1-negative tumors and loss of INI1 confirmed by IHC or molecular confirmation of tumor bi-allelic INI1 loss/mutation when INI1 IHC is equivocal or unavailable	Symptomatic venous thrombosis within 14 days prior to enrollment
	Patients with CNS involvement: active bleeding, or new intratumoral hemorrhage

CNS, central nervous system; EZH2, enhancer of zeste homologue 2; HBV, hepatitis B virus; HCV, hepatitis C virus; HIV, human immunodeficiency virus; IHC, immunohistochemistry; MDS, myelodysplastic syndrome; T-ALL, T-cell acute lymphoblastic leukemia; T-LBL, T-cell lymphoblastic lymphoma.

RESULTS

Safety

- Tazemetostat was generally well tolerated with no drug-related deaths
- During Ph1b, 38 of 48 (79.2%) patients had a treatment-related treatment-emergent adverse event (TEAE). The most common treatment-related TEAEs (≥20% of patients) were vomiting (n=21), nausea (n=15), and anemia (n=10) (**Table 2**)
- 6/48 (12.5%) patients had a TEAE leading to dose reduction and 3/48 (6.3%) had a TEAE leading to discontinuation
- During Ph1b enrollment, 1 patient with chordoma (dosed at tazemetostat 900 mg/m^2 BID for 15 months in Ph1a) developed a secondary malignancy (T-cell lymphoblastic lymphoma)
 - In response, the pediatric recommended Ph2 dose was revised to limit exposure in patients without CNS involvement to 520 mg/m^2 tazemetostat (maximum dosing of 1 year after response, patients to stop treatment until disease progression when they were eligible for retreatment for a total of 2 years of treatment). Patients with ATRT continued to receive 1200 mg/m^2

Table 2. Summary of Most Common Treatment-Related Treatment-Emergent Adverse Events (≥10% Incidence Overall)

Preferred Term	Any Grade N=48 n (%)	Grade 3-4 N=48 n (%)
Vomiting	21 (44)	0
Nausea	15 (31)	0
Anemia	10 (21)	4 (8)
Fatigue	8 (17)	0
Diarrhea	7 (15)	1 (2)
Decreased appetite	7 (15)	1 (2)
Asthenia	6 (13)	0

Efficacy

- Ph1b enrolled 48 patients who received tazemetostat (**Table 3**)
- Across all dose levels and tumor types, ORR was 17% (**Table 4**)
 - Responses were observed in 1 patient dosed at 520 mg/m^2 and 7 patients at 1200 mg/m^2
 - Responses were observed in ATRT (4/21), chordoma (2/5), and ES (2/7)
- Across all tumor types, mean duration of treatment was 20.4 weeks (range: 0–103; **Figure 3**)

Table 3. Baseline Demographics and Disease Characteristics – Dose Expansion (Intent-to-Treat Population)

Characteristics	ATRT n=21	Non-CNS RT n=12	ES n=7	Chordoma n=5	Other n=3	Total N=48
Mean age (range), years	5.3 (1–18)	4.5 (1–15)	16.1 (15–19)	8.0 (4–12)	16.7 (13–21)	7.7 (1–21)
Male/female, n	8/13	9/3	3/4	1/4	2/1	23/25
# of prior lines of therapy, n (%)						
0	0	0	2 (29)	0	0	2 (4)
1–2	18 (86)	7 (58)	5 (71)	5 (100)	2 (67)	37 (77)
≥3	3 (14)	5 (42)	0	0	1 (33)	9 (19)
Median, n (range)	2 (1–9)	2 (1–5)	1 (0–2)	1 (1–2)	2 (1–3)	2 (0–9)

ATRT, atypical teratoid rhabdoid tumor; CNS, central nervous system; ES, epithelioid sarcoma; RT, rhabdoid tumor.

Table 4. Summary of Best Overall Response and Duration of Response by Tumor Type (Treated Patients)

	ATRT n=21	Non-CNS RT n=12	ES n=7	Chordoma n=5	Other n=3	Total N=48
1200 mg/m^2 BID, n (%)	19 (90)	5 (42)	4 (57)	1 (20)	2 (67)	31 (65)
800 mg/m^2 TID tablet, n (%)	2 (10)	1 (8)	2 (29)	1 (20)	0	6 (13)
520 mg/m^2 BID, n (%)	0	6 (50)	1 (14)	3 (60)	1 (33)	11 (23)
ORR, (95% CI)*	19% (5–42)	0% (NE–NE)	29% (4–71)	40% (5–85)	0% (NE–NE)	17% (7–30)
CR, n (%)	1 (5)	0	1 (14)	0	0	2 (4)
PR, n (%)	3 (14)	0	1 (14)	2 (40)	0	6 (13)
SD, n (%)	3 (14)	2 (17)	3 (43)	1 (20)	1 (33)	10 (21)
PD, n (%)	12 (57)	8 (67)	2 (29)	0	2 (67)	24 (50)
Not known, n (%)	2 (10)	2 (17)	0	2 (40)	0	6 (13)
Median DOR (95% CI), weeks	28 (24–75)	NE (NE–NE)	NE (24–NE)†	NE (NE–NE)‡	NE (NE–NE)	28 (24–75)

*ORR calculated as the percentage of patients with a best response of either CR or PR.
†DOR of 23.9 and 55.3 (ongoing) weeks in patients with response.
‡DOR of 15.9 (ongoing) and 74.1 (ongoing) weeks in patients with response.
ATRT, atypical teratoid rhabdoid tumor; BID, twice a day; CI, confidence interval; CNS, central nervous system; CR, complete response; DOR, duration of response; ES, epithelioid sarcoma; NE, not evaluable; ORR, objective response rate; PD, progressive disease; PR, partial response; RT, rhabdoid tumor; SD, stable disease; TID, three times a day.

Figure 3. Time on Treatment

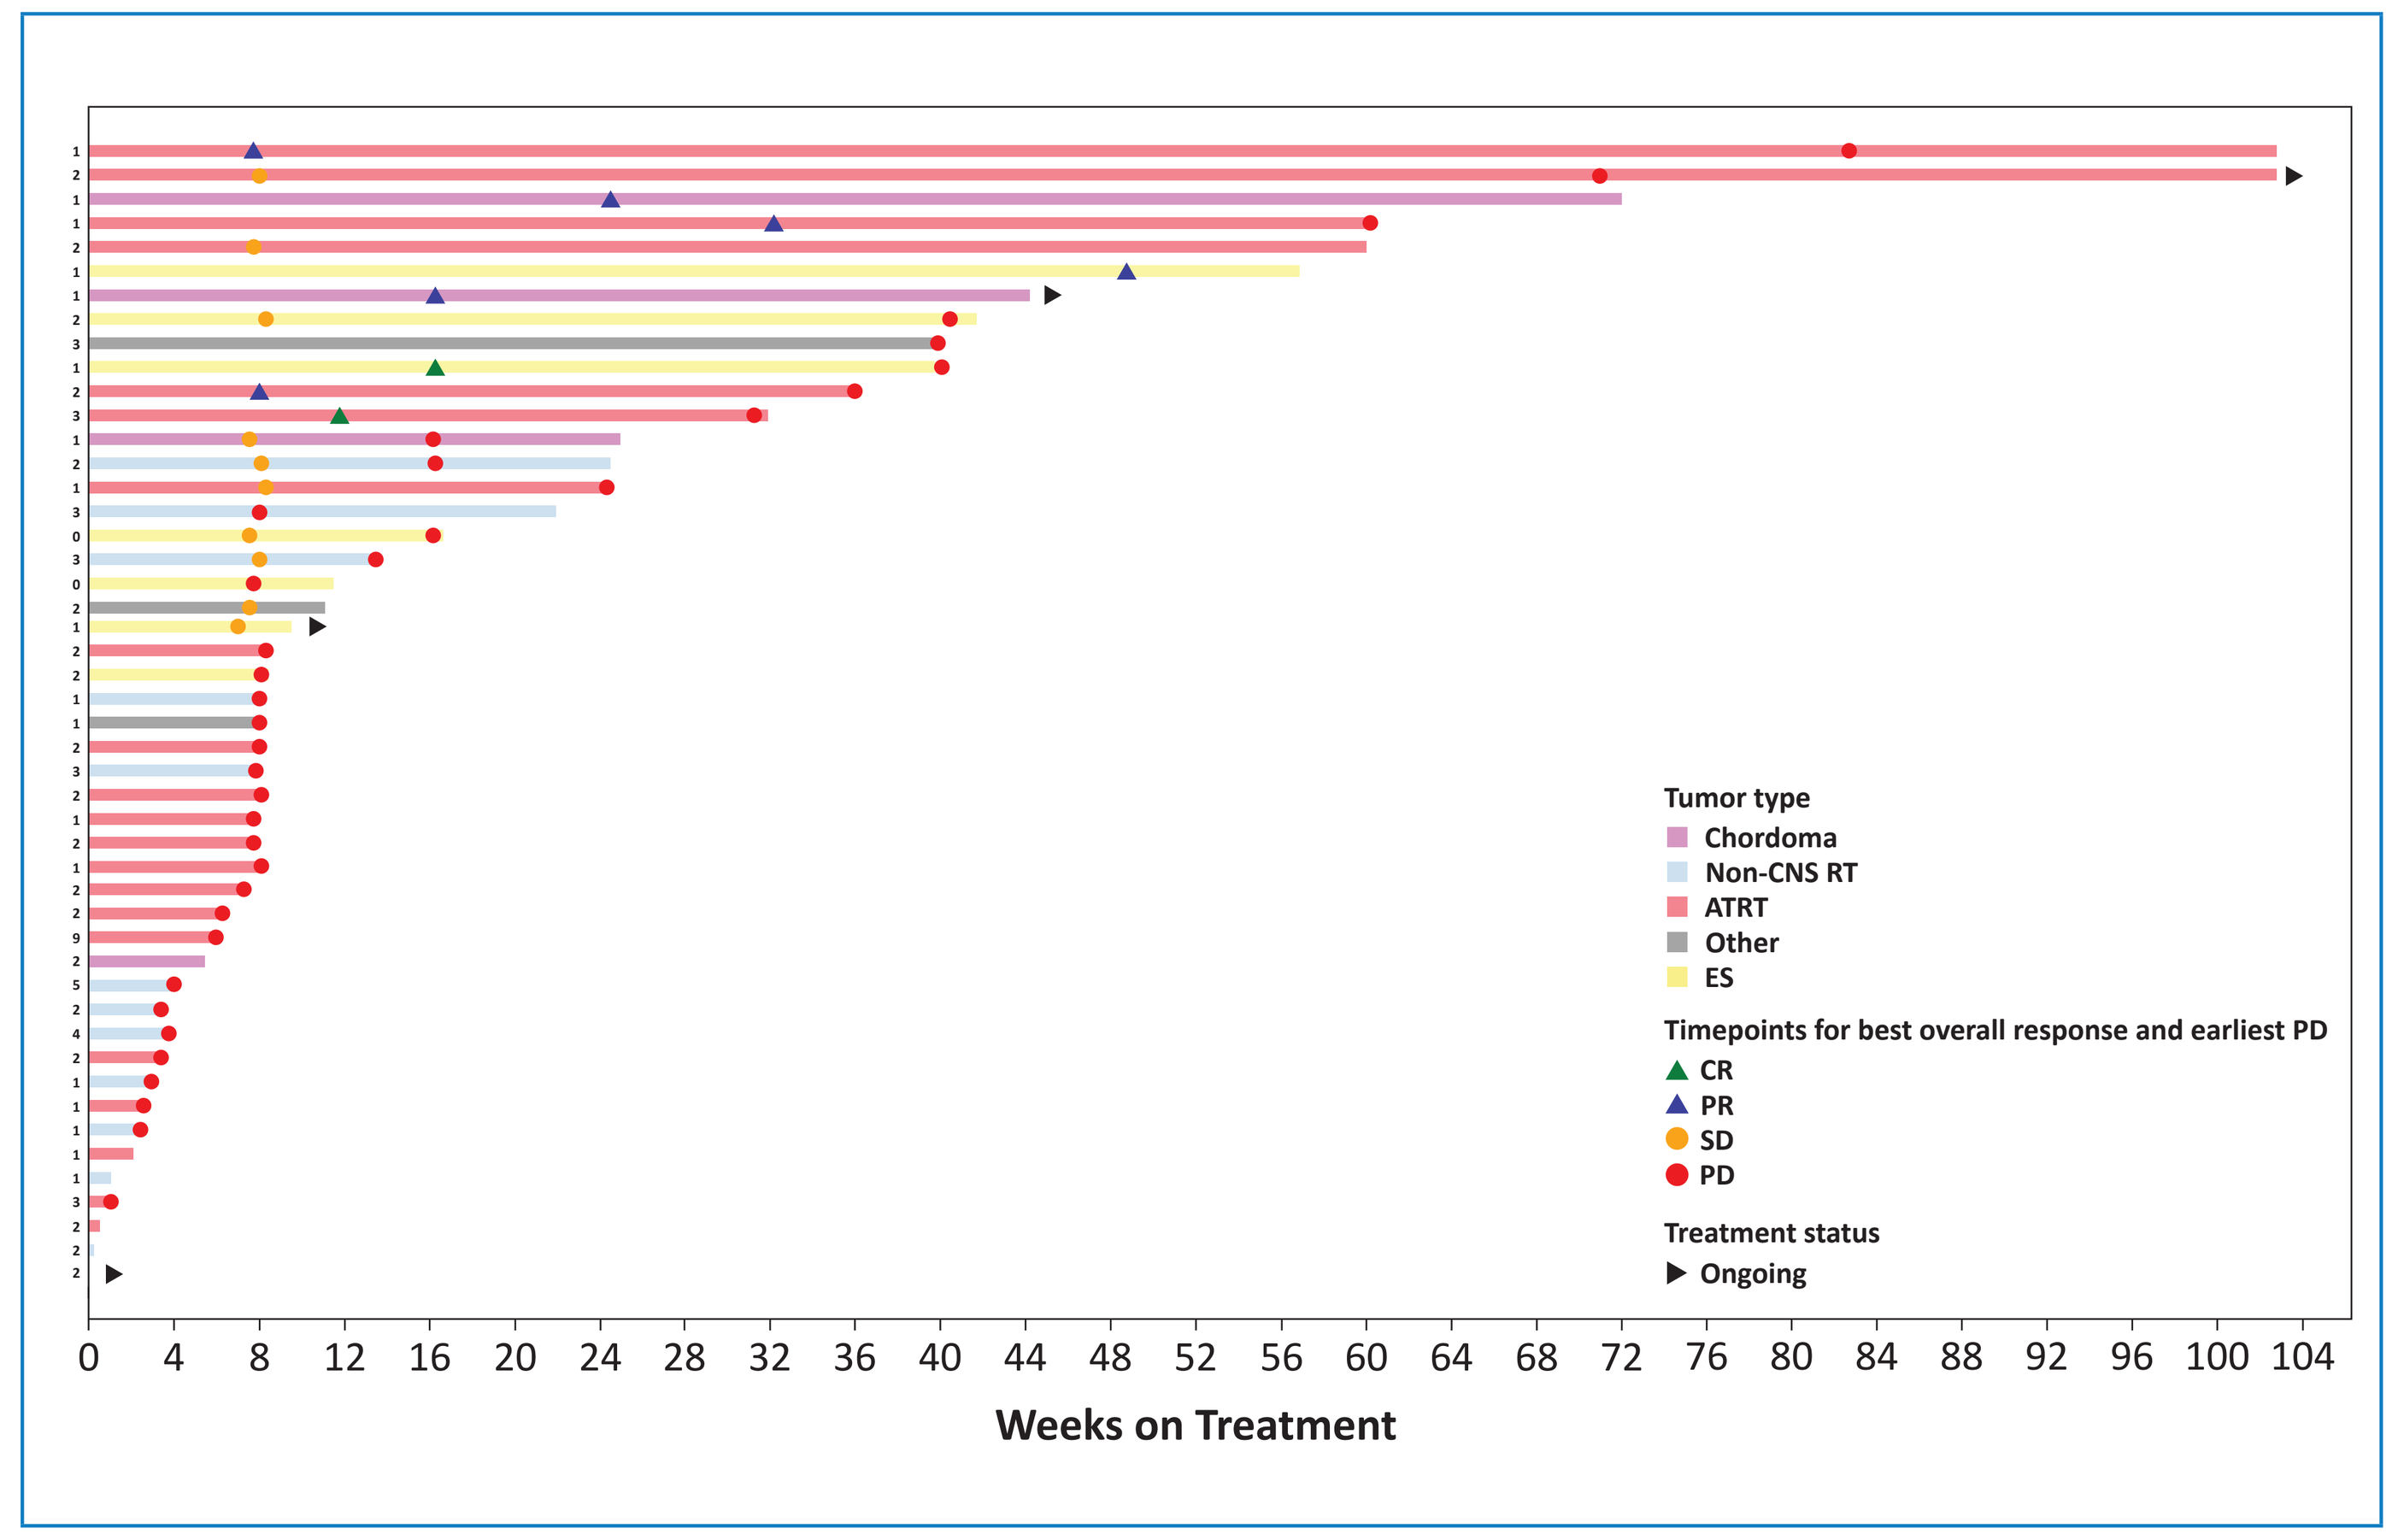

Tumor type
Chordoma
Non-CNS RT
ATRT
Other
ES
Timepoints for best overall response and earliest PD
CR
PR
SD
PD
Treatment status
Ongoing
0 4 8 12 16 20 24 28 32 36 40 44 48 52 56 60 64 68 72 76 80 84 88 92 96 100 104
Weeks on Treatment

Numbers along y-axis indicate number of prior lines of anticancer therapy.
ATRT, atypical teratoid rhabdoid tumor; CNS, central nervous system; CR, complete response; DOR, duration of response; ES, epithelioid sarcoma; PD, progressive disease; PR, partial response; RT, rhabdoid tumor; SD, stable disease.

CONCLUSIONS

- Interim results indicate tazemetostat is generally well tolerated in children with an AE profile similar to adults
- Patient enrollment in the non-ATRT, INI1-negative cohorts is ongoing
- Tazemetostat shows promising antitumor activity in a subset of pediatric tumors, including ATRT, chordoma, and ES
- The response rate observed in children with ES is comparable to that seen in adults

REFERENCES

1. Wang X, et al. *Nat Genet*. 2017;49:289–295.
2. Tomlinson GE, et al. *J Clin Oncol*. 2005;23:7641–7645.
3. Bourdeaut F, et al. *Pediatr Blood Cancer*. 2008;51:363–368.
4. Thway K, et al. *Adv Anat Pathol*. 2016;23:41–49.
5. Antonelli M, et al. *Am J Surg Pathol*. 2017;41:56–61.
6. Knutson SK, et al. *Proc Natl Acad Sci USA*. 2013;110:7922–7927.
7. TAZVERIK™ prescribing information 2020; https://www.accessdata.fda.gov/drugsatfda_docs/label/2020/211723s000lbl.pdf
8. Chi SN, et al. *J Clin Oncol*. 2016;34:15 suppl, TPS10587-TPS10587.

ACKNOWLEDGMENTS

This study is sponsored by Epizyme, Inc. Third-party writing assistance was provided by Ashfield Healthcare Communications, part of UDG Healthcare plc, and funded by Epizyme, Inc.

Please contact Dr. Susan N. Chi (Susan_Chi@dfci.harvard.edu) with questions or comments about this poster.

Poster presented at the American Society of Clinical Oncology Annual Meeting (Virtual Format), May 29–31, 2020