TPS11573

A Phase 1b/3 Global Randomized, Double-blind, Placebo-controlled Study of Tazemetostat in Combination with Doxorubicin as Frontline Therapy for Advanced Epithelioid Sarcoma

Shiraj Sen,[1] Meredith McKean,[2] Laura Sierra,[3] Jessica Ainscough,[3] Jay Yang,[3] Anthony Hamlett,[3] Tom Zimmerman,[3] Melinda Merchant,[3] Sant Chawla[4]

[1]Sarah Cannon Research Institute, Denver, CO, USA; [2]Tennessee Oncology, Nashville, TN, USA; [3]Epizyme Inc., Cambridge, MA, USA; [4]Sarcoma Oncology Center, Santa Monica, CA, USA

BACKGROUND

- Epithelioid sarcoma (ES) is a rare and aggressive subtype of soft tissue sarcoma (STS) that can originate in any anatomic location. It predominantly affects young adults (<40 years of age)[1]
 - The effectiveness of cytotoxic chemotherapy in ES is limited, with a median progression-free survival (PFS) of 4 months and 6 months for gemcitabine- and anthracycline-based therapies, respectively[2]
- Patients with advanced ES are treated with palliative chemotherapy but median overall survival (OS) is <12 months[3]
- In more than 90% of cases, ES is characterized by the loss of function of integrase interactor 1 (INI1)[4], a critical component of epigenetic regulation via the switch/sucrose nonfermentable (SWI/SNF) protein complex[5]
 - The loss of INI1 may allow enhancer of zeste homolog 2 (EZH2), a catalytic subunit of the polycomb repressive complex 2 (PRC2), to repress cell differentiation and promote tumorigenesis via increased trimethylation of histone H3 at lysine 27 (H3K27)[5]
- Tazemetostat is a selective, oral, small molecule inhibitor of EZH2[6] (**Figure 1**)

Figure 1. Mechanism of Action of Tazemetostat

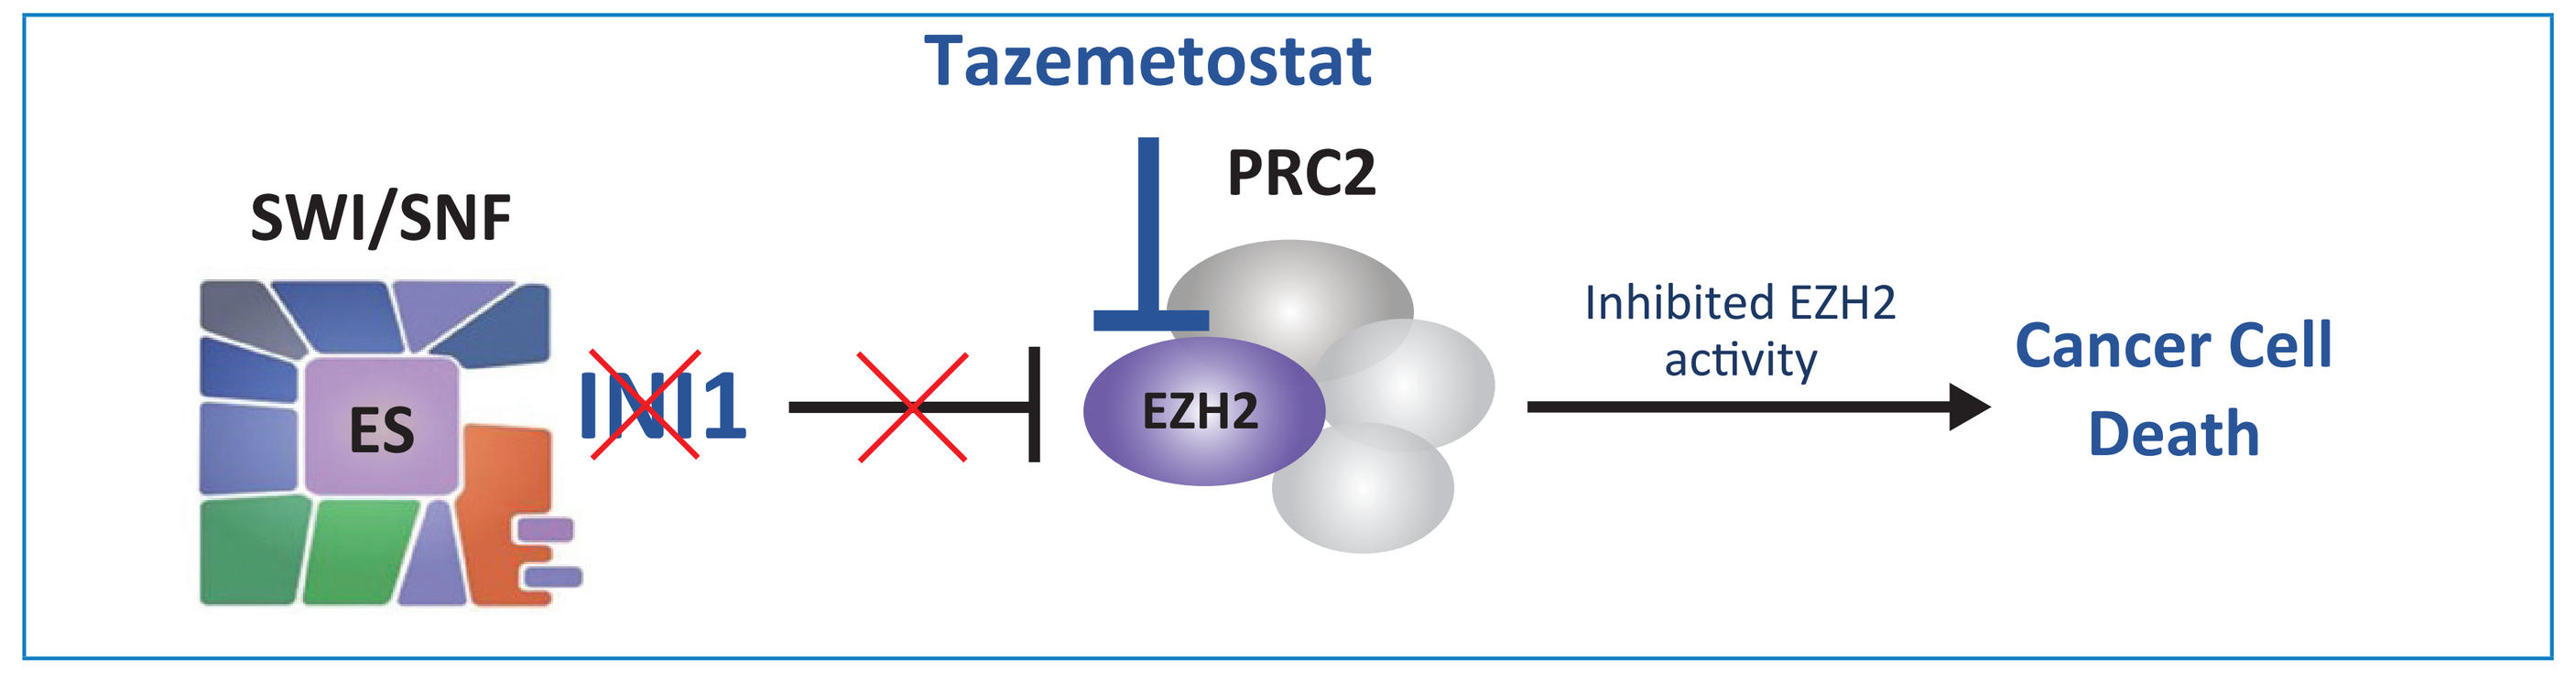

Tazemetostat
SWI/SNF
PRC2
ES
INI1
EZH2
Inhibited EZH2 activity
Cancer Cell Death

ES, epithelioid sarcoma; EZH2, zeste homolog 2; INI1, integrase interactor 1; PRC2, polycomb repressive complex 2; SWI/SNF, switch/sucrose nonfermentable.

- In preclinical studies using an ES cell line and a mouse xenograft model of synovial sarcoma, combination administration of tazemetostat + doxorubicin demonstrated synergistic antitumor activity superior to that observed with either therapy alone[7,8] (**Figure 2**)

Figure 2. Preclinical Data Demonstrate Tazemetostat and Doxorubicin Have Synergistic Antitumor Activity

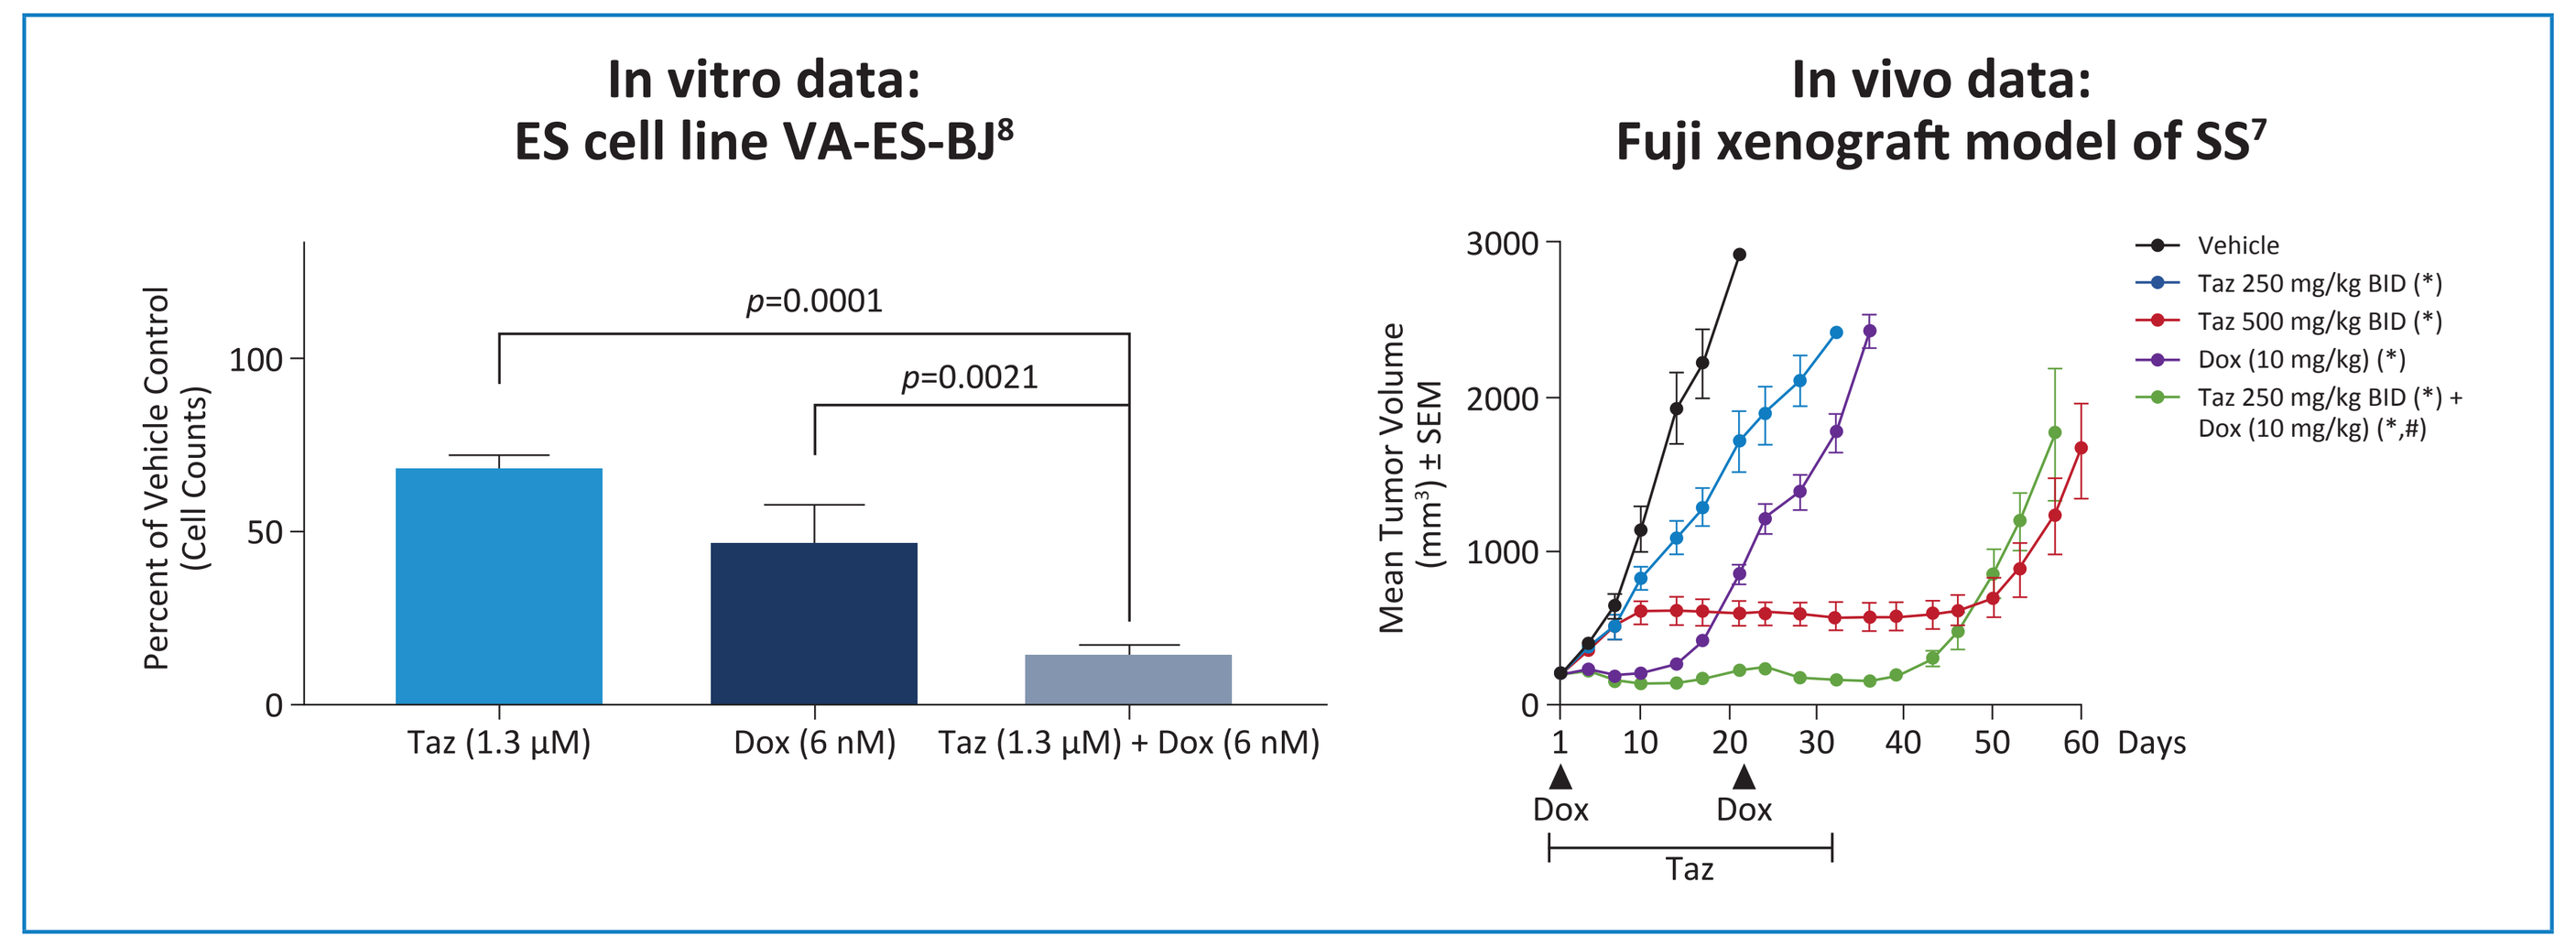

In vitro data:
ES cell line VA-ES-BJ[8]
Percent of Vehicle Control (Cell Counts)
p=0.0001
p=0.0021
Taz (1.3 μM)
Dox (6 nM)
Taz (1.3 μM) + Dox (6 nM)
In vivo data:
Fuji xenograft model of SS[7]
Mean Tumor Volume (mm³) ± SEM
Vehicle
Taz 250 mg/kg BID (*)
Taz 500 mg/kg BID (*)
Dox (10 mg/kg) (*)
Taz 250 mg/kg BID (*) + Dox (10 mg/kg) (*,#)
Days
Dox
Dox
Taz

*$p<0.05$ vs vehicle.
#$p<0.05$ vs both 250 mg/kg tazemetostat and doxorubicin; one-way analysis of variance followed by Tukey's multiple comparison test after logarithmic transformation.
Data shown as mean values ± SEM.
BID, twice daily; Dox, doxorubicin; ES, epithelioid sarcoma; SEM, standard error of mean; SS, synovial sarcoma; Taz, tazemetostat.

- In a multicenter, phase 2 study in patients with INI1-negative ES (n=62), tazemetostat was shown to have meaningful clinical activity in some patients with advanced ES[9,10]
 - Tazemetostat was well tolerated and associated with a 15% objective response rate and a median OS of 82.4 weeks (approximately 19 months)[9]
- Tazemetostat has been approved by the US Food and Drug Administration for the treatment of adults and pediatric patients aged 16 years and older with metastatic or locally advanced ES not eligible for complete resection[10]

STUDY DESIGN

- EZH-301 (NCT04204941) is a phase 1b/3, global, randomized, double-blind, placebo-controlled study to evaluate the safety and efficacy of tazemetostat + doxorubicin in frontline therapy for patients with ES not eligible for resection (**Figure 3**)
- A safety run in (phase 1b portion) is currently enrolling patients with STS in order to identify the maximum tolerable dose and the recommended phase 3 dose of tazemetostat in combination with standard-of-care doxorubicin (75 mg/m^2 every 21 days for up to 6 cycles)
- Total study enrollment will be up to 24 patients with STS in phase 1b and approximately 140 patients with ES, and the total study duration will be approximately 9.5 years
 - One interim analysis is planned when 60 PFS events have been observed in phase 3 to assess for futility

Figure 3. EZH-301 Study Design Schema

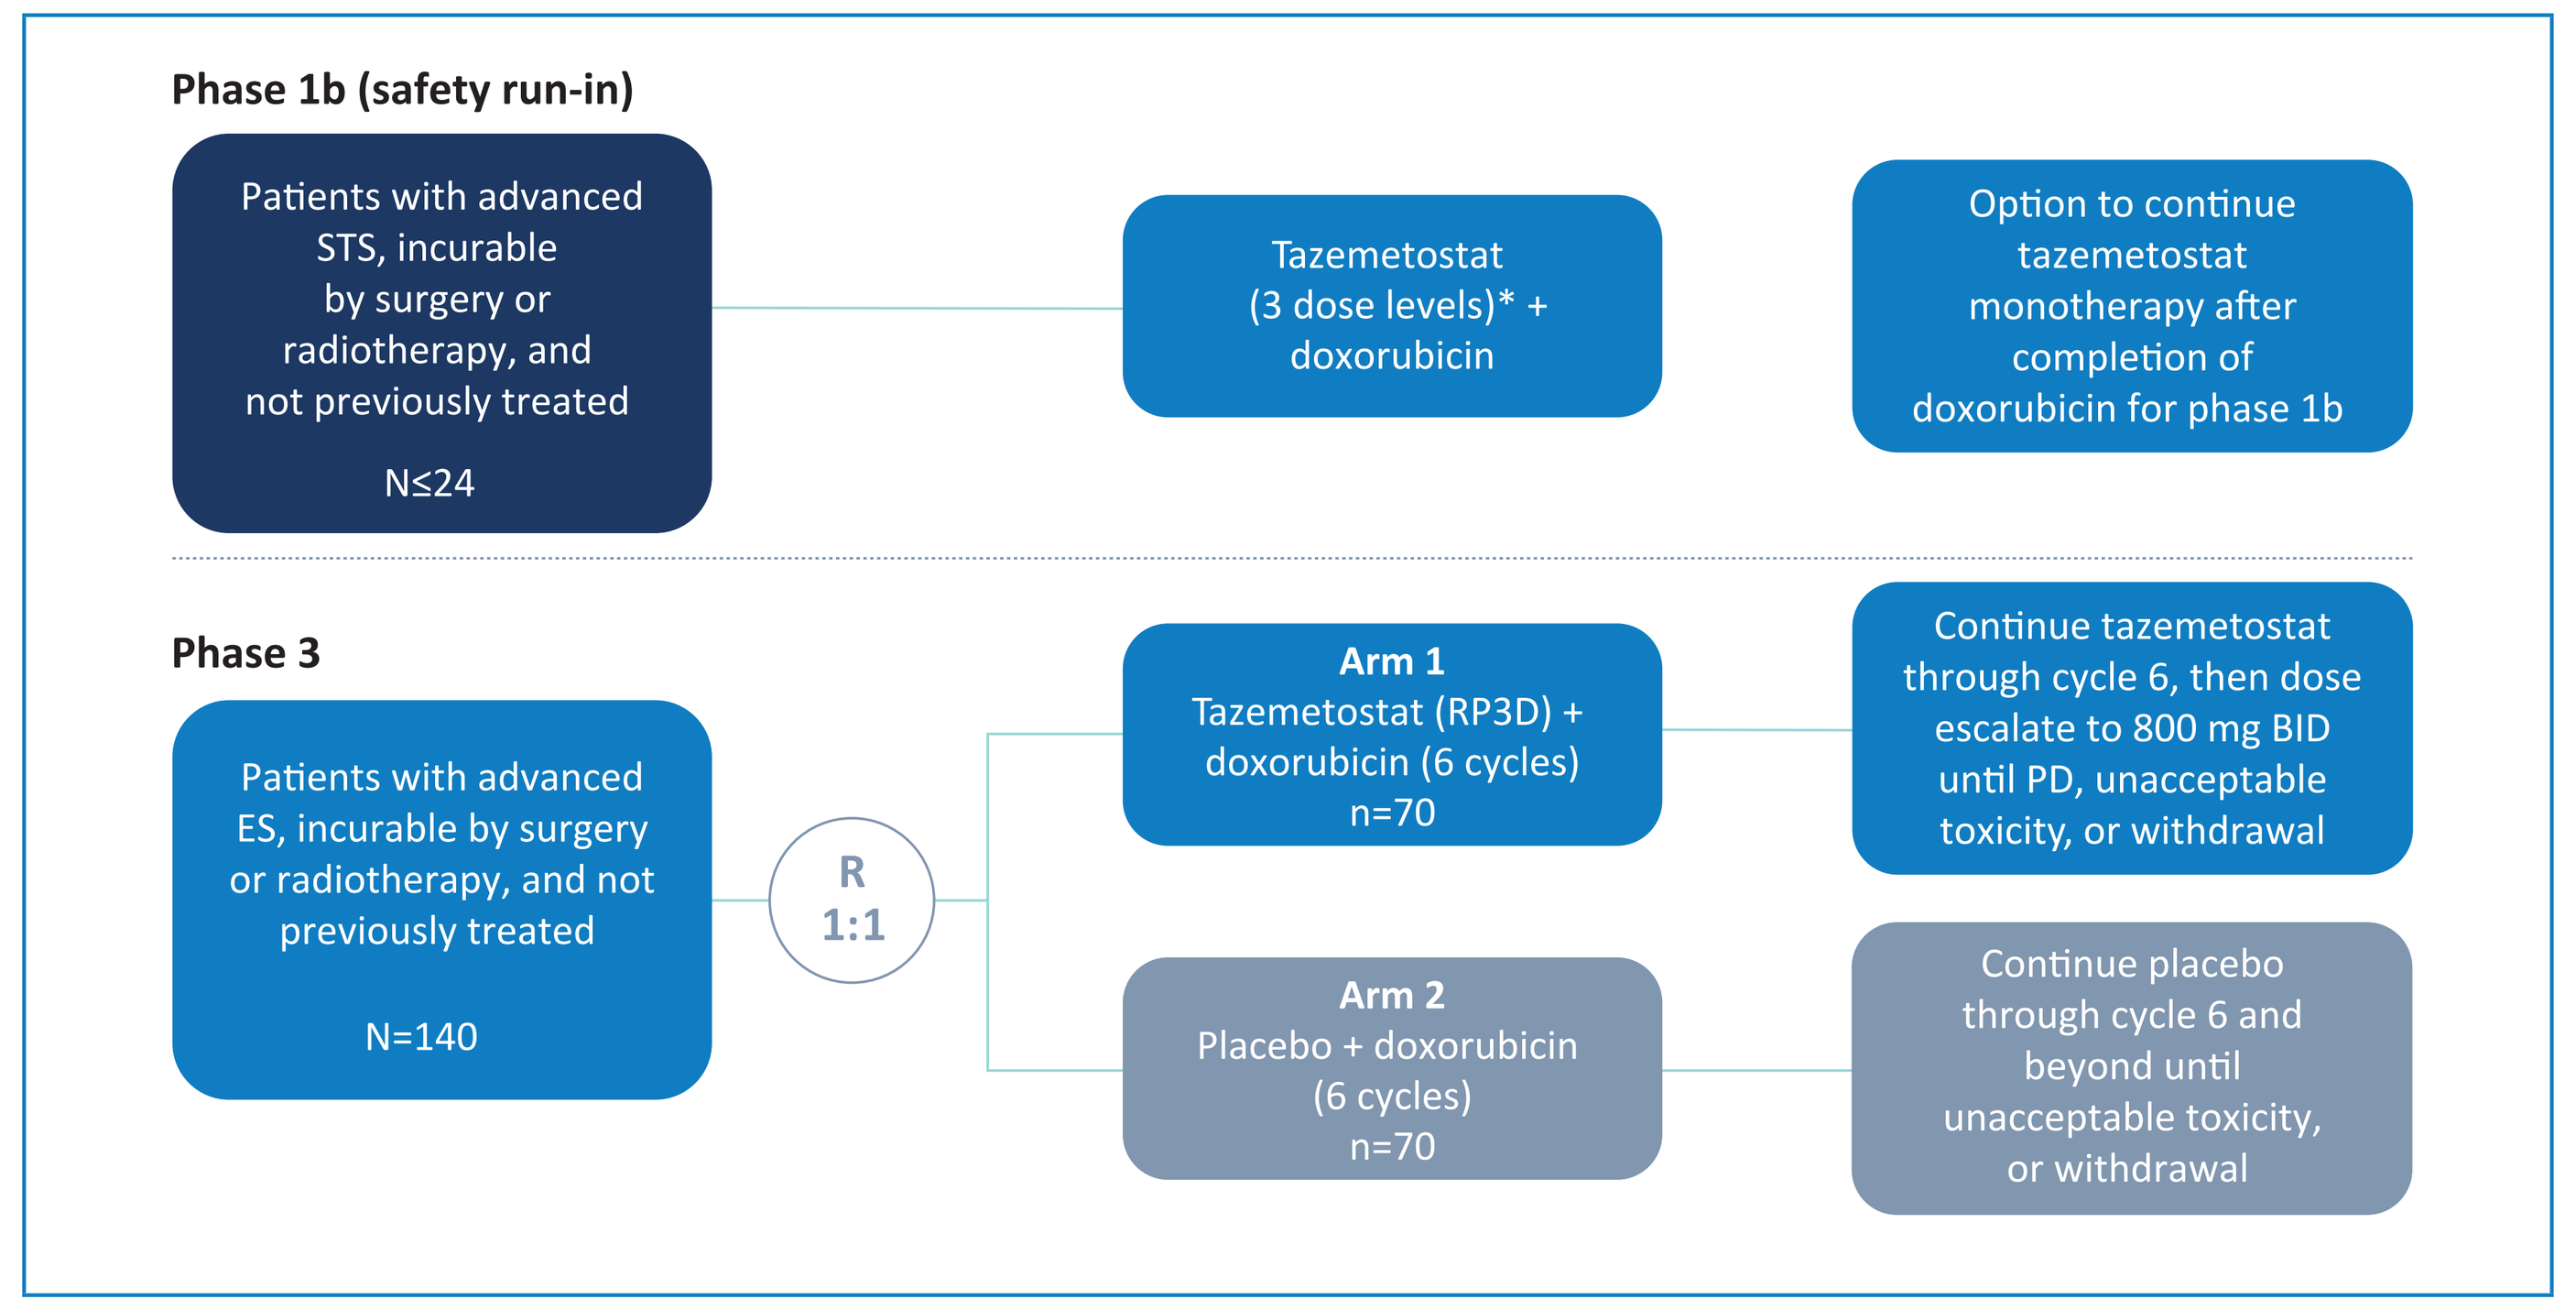

Phase 1b (safety run-in)
Patients with advanced STS, incurable by surgery or radiotherapy, and not previously treated
N≤24
Tazemetostat (3 dose levels)* + doxorubicin
Option to continue tazemetostat monotherapy after completion of doxorubicin for phase 1b
Phase 3
Patients with advanced ES, incurable by surgery or radiotherapy, and not previously treated
N=140
R 1:1
Arm 1
Tazemetostat (RP3D) + doxorubicin (6 cycles)
n=70
Continue tazemetostat through cycle 6, then dose escalate to 800 mg BID until PD, unacceptable toxicity, or withdrawal
Arm 2
Placebo + doxorubicin (6 cycles)
n=70
Continue placebo through cycle 6 and beyond until unacceptable toxicity, or withdrawal

*Tazemetostat to be escalated from a starting dose of 400 mg BID PO to 600 mg BID PO to 800 mg twice daily PO as tolerated in a standard 3+3 design.
A futility analysis will be undertaken during the interim analysis and sample size re-estimation could be performed.
BID, twice daily; ES, epithelioid sarcoma; EZH, enhancer of zeste homolog; PD, progressive disease; PO, orally; RP3D, recommended phase 3 dose; STS, soft tissue sarcoma.

Phase 1b

- Using a standard 3+3 design, tazemetostat will be dose escalated in approximately 24 patients with STS
- Tazemetostat will be tested in up to 3 dose levels as tolerated in combination with doxorubicin: dose level 1 (400 mg), dose level 2 (600 mg), and dose level 3 (800 mg) oral administration twice daily (BID). Patients will receive doxorubicin 75 mg/m^2 intravenously on day 1 of each cycle for up to 6 cycles
- The maximum tolerated dose (MTD) will be identified as the highest dose level at which less than one-third of patients (0 of 3 patients or 1 of 6 patients) experience a dose-limiting toxicity. Up to 12 patients will be enrolled at the MTD to collect additional safety and pharmacokinetics (PK) data
- After completion of doxorubicin treatment, patients may continue to receive tazemetostat monotherapy at 800 mg BID, until disease progression or intolerable toxicity
- A safety review committee will be responsible for ongoing data review and ensuring patient safety during dose escalation

Phase 3

- Phase 3 will compare the recommended phase 3 dose of tazemetostat + doxorubicin with placebo + doxorubicin given as first-line treatment to patients with locally advanced, unresectable, or metastatic ES
- Up to 140 patients will be randomized (1:1 ratio) to receive up to 6 cycles of tazemetostat + doxorubicin or placebo + doxorubicin in 21-day cycles. Randomization will be stratified by Eastern Cooperative Oncology Group status score (0 vs 1 to 2) and patient age (18–35 years vs >35 years)
- After completion of doxorubicin, patients will continue treatment with either blinded tazemetostat monotherapy or placebo until disease progression or intolerable toxicity
- An independent data-monitoring committee will be responsible for ongoing data review and ensuring patient safety during this phase of the study

PATIENT SELECTION

- Key patient eligibility criteria are presented in **Table 1**

Table 1. Key Eligibility Criteria

Key Inclusion Criteria	Key Exclusion Criteria
Male or female, ≥18 years of age, with a life expectancy of ≥3 months	Prior systemic anticancer therapy (for both safety run-in and phase 3), or prior exposure to tazemetostat or other EZH2 inhibitor
Histologically confirmed disease, incurable by surgery or radiotherapy: • Phase 1b: STS • Phase 3: Morphology and immunophenotypic panel consistent with ES (eg, CD34, epithelial membrane antigen, keratin, INI1)	Presence of any contraindications for, or hypersensitivity to, the study drugs
Measurable disease (RECIST v1.1)	Prior history of myeloid malignancies (including MDS or AML), or T-LBL/T-ALL
ECOG performance status score of 0–2	Major surgery within 4 weeks before first dose of study treatment
Adequate cardiac, hematologic, renal, and hepatic function	

AML, acute myeloid leukemia; ES, epithelioid sarcoma; ECOG, Eastern Cooperative Oncology Group; EZH2, enhancer of zeste homologue 2; INI1, integrase interactor 1; MDS, myelodysplastic syndrome; RECIST 1.1, Response Evaluation Criteria in Solid Tumors, version 1.1; STS, soft tissue sarcoma; T-ALL, T-cell acute lymphoblastic leukemia; T-LBL, T-cell lymphoblastic lymphoma.

STUDY OBJECTIVES

- Phase 1b
 - Primary objective: Safety and tolerability of tazemetostat + doxorubicin and selection of a recommended phase 3 dose
 - Secondary objective: PK of tazemetostat when administered in combination with doxorubicin
- Phase 3
 - Primary objective: PFS by independent review committee for tazemetostat + doxorubicin vs placebo + doxorubicin
 - Secondary objectives:
 - PFS by investigator assessment
 - OS
 - Safety and tolerability
 - Disease control rate at 24 weeks
 - Objective response rate
 - Duration of response
 - Health-related quality of life using European Organization for Research and Treatment of Cancer Quality of Life Questionnaire-Core 30
 - PFS on next treatment (time from randomization to start of second line of treatment)
 - Time to subsequent anticancer therapy (from randomization to the earlier date of disease progression on the next anticancer therapy following study treatment or death)
 - Population PK parameters for tazemetostat when administered in combination with doxorubicin

ACKNOWLEDGMENTS

This study is sponsored by Epizyme, Inc. Third-party writing assistance was provided by Ashfield Healthcare Communications and funded by Epizyme, Inc.

STATISTICAL CONSIDERATIONS

- In phase 3, the efficacy analyses will be performed on the intent-to-treat population, defined as all randomized patients. Key efficacy analyses will also be performed on the per protocol population, defined as all subjects in the intent-to-treat population who did not have a major protocol deviation, as supportive evidence and to assess the robustness of the efficacy findings
- Phase 3 sample size: The sample size calculation is based on the primary endpoint of PFS. The null hypothesis for the primary efficacy endpoint will be that there is no difference for PFS between the 2 treatment groups: placebo + doxorubicin and tazemetostat + doxorubicin. The median PFS assumptions for the placebo + doxorubicin is 8 months and 15 months for tazemetostat + doxorubicin. This will require 81 PFS events with a total of 140 patients, and will provide 80% power for the analysis of PFS with a 2-sided total type I error of 0.05 to reject the null hypothesis at the final analysis
 - The sample size calculation assumes that the addition of tazemetostat to doxorubicin will prolong PFS by 88% (hazard ratio = 0.533) from 8 months (for placebo + doxorubicin) to 15 months, with 30% of patients lost to follow-up over a 6.5-year enrollment period, a 1-year treatment period, and a 2-year follow-up period

CURRENT STATUS

- EZH-301 was initiated in December 2019
- Enrollment of the 1st dose level was completed in March 2018 and the Safety Review Committee recommended escalation to 600 mg and is currently open for enrollment
- As of 11 May 2020, phase 1b is actively enrolling at the following open sites:
 - California: Sarcoma Oncology Research Center. Santa Monica, California, United States, 90403
 - Colorado: Sarah Cannon Research Institute at HealthONE. Denver, Colorado, United States, 80218
 - Colorado: University of Colorado Denver, Anschutz Medical Campus. Aurora, Colorado, United States, 80045
 - Michigan: University of Michigan. Ann Arbor, Michigan, United States, 48109
 - Missouri: Washington University. St. Louis, Missouri, United States, 63110
 - Tennessee: Sarah Cannon and HCA Research Institute. Nashville, Tennessee, United States, 37203
- The following global sites are projected to open for the phase 1b portion of this study toward the end of 2020 or the beginning of 2021: Canada, United Kingdom, and Taiwan (**Figure 4**)
 - Additional global sites will be added for the phase 3 portion of the study

Figure 4. Global Sites Projected to Open for Phase 1b in Late 2020/Early 2021

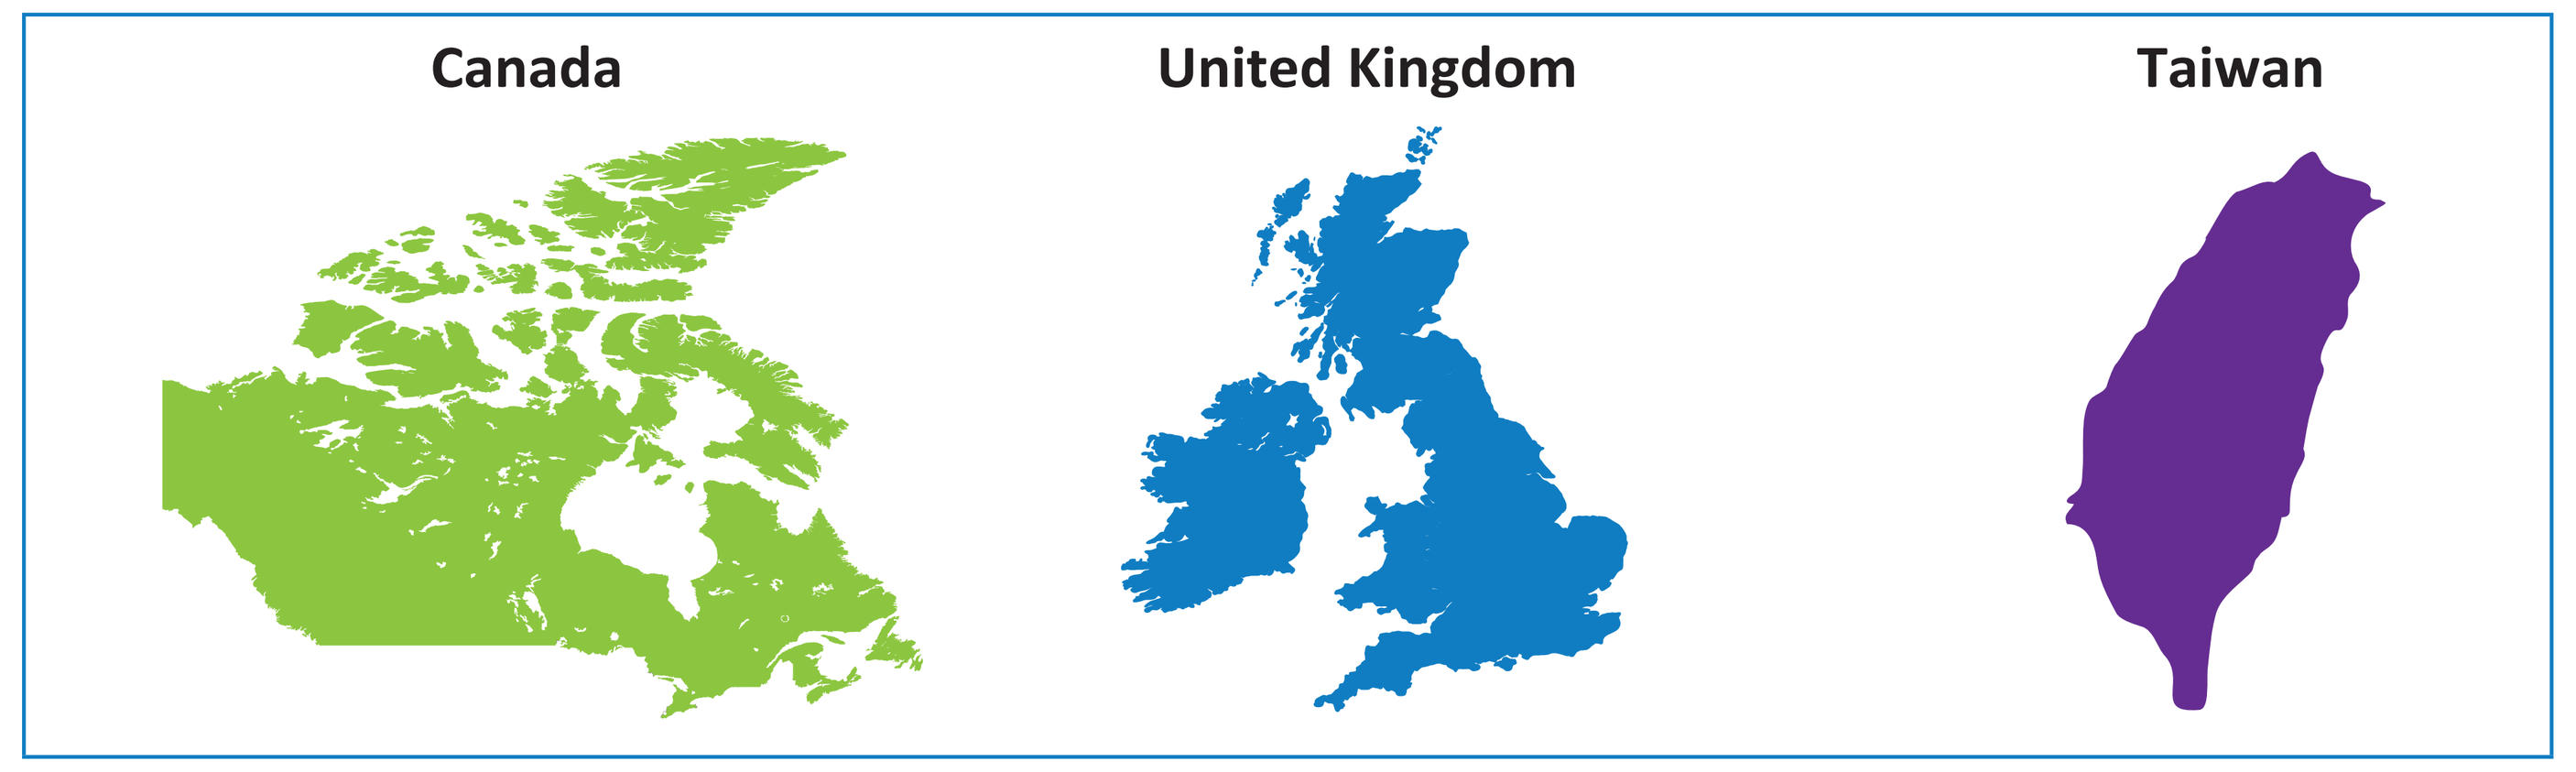

Canada
United Kingdom
Taiwan

For more information, please refer to ClinicalTrials.gov identifier: NCT04204941 (NDA 211723)

REFERENCES

1. Chbani L, et al. *Am J Clin Pathol.* 2009;131(2):222–227.
2. Frezza AM, et al. *JAMA Oncol*. 2018;4(9):e180219.
3. Jones RL, et al. *Am J Clin Oncol*. 2012;35(4):351–357.
4. Hornick JL, et al. *Am J Surg Pathol*. 2009;33(4):542–550.
5. Wilson BG, et al. *Cancer Cell*. 2010;18(4):316–328.
6. Knutson SK, et al. *Proc Natl Acad Sci USA*. 2013;110(19):7922–7927.
7. Kawano S, et al. *PLoS One*. 2016;11(7):e0158888.
8. Epizyme, Inc. Data on file.
9. Stacchiotti S, et al. Presented at the 2019 ASCO Annual Meeting Abstract # 11003.
10. TAZVERIK™ prescribing information 2020; https://www.accessdatafda.gov/drugsatfda_docs/label/2020/211723s000lbl.pdf

Please contact Dr. Shiraj Sen (shiraj.sen@sarahcannon.com) with questions or comments about this poster.

Poster presented at the American Society of Clinical Oncology Annual Meeting (Virtual Format), May 29–31, 2020