7572

Updated Interim Analysis of the Randomized Phase 1b/3 Study of Tazemetostat in Combination With Lenalidomide and Rituximab in Patients With Relapsed/Refractory Follicular Lymphoma

Connie Lee Batlevi, MD, PhD[1]; Steven I. Park, MD[2]; Tycel Phillips, MD[3]; Jennifer E. Amengual, MD[4]; David Andorsky, MD[5]; Philip Campbell, MB, ChB[6]; Pamela McKay, MD[7]; John P. Leonard, MD[8]; Manu Sondhi, MD, MPH[9]; Yingxue Chen, PhD[9]; Heather O'Connor, MD[9]; Pamela L. Slatcher, MHS, MT, PA[9]; Franck Morschhauser, MD, PhD[10]

[1]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [2]Levine Cancer Institute, Atrium Health, Charlotte, NC, USA; [3]University of Michigan, Ann Arbor, MI, USA; [4]Columbia University Irving Medical Center, New York, NY, USA; [5]Rocky Mountain Cancer Centers, Boulder, CO, USA; [6]Barwon Health, University Hospital Geelong, Geelong, VIC, Australia; [7]Beatson West of Scotland Cancer Centre, Glasgow, Scotland, UK; [8]Weill Cornell Medicine, New York, NY, USA; [9]Epizyme, Inc., Cambridge, MA, USA; [10]CHU Lille, Service des Maladies du Sang, Lille, France

INTRODUCTION

- Tazemetostat (TAZ), a first-in-class, oral, selective methyltransferase inhibitor of enhancer of zeste homolog 2 (EZH2) activity, is approved by the US Food and Drug Administration (FDA) for the treatment of patients with mutant (MT) *EZH2* relapsed or refractory (R/R) follicular lymphoma (FL) or patients with R/R FL who have no satisfactory alternative treatment options[1,2]
- In a phase 2 study of patients with R/R FL who received ≥2 prior systemic therapies, TAZ demonstrated objective response rates (ORRs) of 69% in 45 patients with MT *EZH2* and 35% in 54 patients with wild-type (WT) *EZH2*[2]
- Rituximab, an anti-CD20 chimeric monoclonal antibody, and lenalidomide, an immunomodulatory agent, are approved by the FDA as a combination treatment (R^2) for R/R FL[3,4]
 - In clinical studies in patients with R/R FL, R^2 demonstrated ORRs of 73% to 78%, and median progression-free survival (PFS) of 36 to 39 months[5,6]
- Preclinical data demonstrated in vitro synergistic antiproliferative activity when TAZ was combined with lenalidomide or rituximab, or with R^2
- SYMPHONY-1 (EZH-302) is a phase 1b/3 study designed to determine the recommended phase 3 dose (RP3D), efficacy, and safety of TAZ + R^2 in patients with R/R FL after ≥1 prior therapy

OBJECTIVE

- To report an updated interim analysis of the phase 1b safety run-in period, and to assess the safety, pharmacokinetics (PK), and clinical activity of TAZ when concomitantly administered with R^2 in patients with R/R FL

METHODS

Study Design

- This international, multicenter, randomized, double-blind, active-controlled, 3-stage, biomarker-enriched, phase 1b/3 study (NCT04224493) is evaluating TAZ + R^2 in patients with R/R FL (**Figure 1**)

Figure 1. Study Design

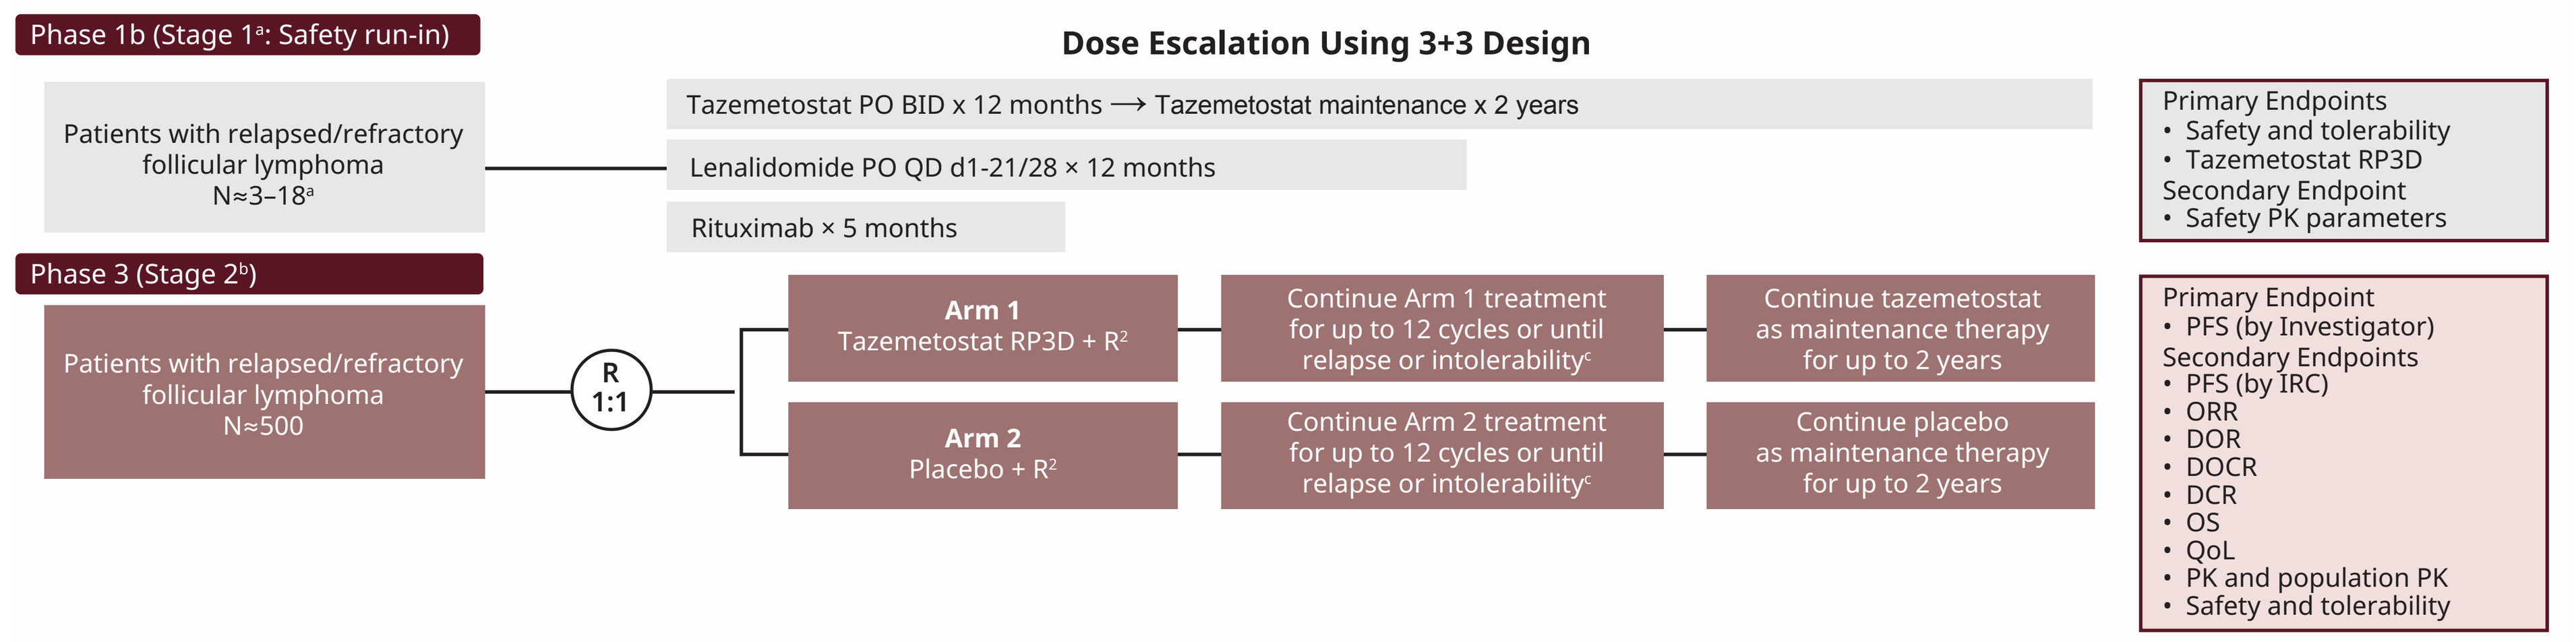
Phase 1b (Stage 1[a]: Safety run-in)
Dose Escalation Using 3+3 Design
Patients with relapsed/refractory follicular lymphoma N≈3–18[a]
Tazemetostat PO BID x 12 months ⟶ Tazemetostat maintenance x 2 years
Lenalidomide PO QD d1-21/28 × 12 months
Rituximab × 5 months
Primary Endpoints
• Safety and tolerability
• Tazemetostat RP3D
Secondary Endpoint
• Safety PK parameters
Phase 3 (Stage 2[b])
Patients with relapsed/refractory follicular lymphoma N≈500
R 1:1
Arm 1 Tazemetostat RP3D + R²
Continue Arm 1 treatment for up to 12 cycles or until relapse or intolerability[c]
Continue tazemetostat as maintenance therapy for up to 2 years
Arm 2 Placebo + R²
Continue Arm 2 treatment for up to 12 cycles or until relapse or intolerability[c]
Continue placebo as maintenance therapy for up to 2 years
Primary Endpoint
• PFS (by Investigator)
Secondary Endpoints
• PFS (by IRC)
• ORR
• DOR
• DOCR
• DCR
• OS
• QoL
• PK and population PK
• Safety and tolerability

[a]Additional patients enrolled to further study safety in the 600-mg and 800-mg groups. [b]An optional stage 3, for patients with MT *EZH2* status only, will be executed if the efficacy in stage 2 fails for all patients but is sufficiently promising for MT *EZH2* patients (as assessed in a futility analysis during stage 2). [c]All patients receive treatment in 28-day cycles.
BID, twice daily; DCR, disease control rate; DOCR, duration of complete response; DOR, duration of response; IRC, independent radiology committee; MT, mutant; ORR, objective response rate; OS, overall survival; PFS, progression free survival; PK, pharmacokinetics; PO, orally; QD, once daily; QoL, quality of life; R^2, lenalidomide plus rituximab; RP3D, recommended phase 3 dose; WT, wild-type.

- The phase 1b safety run-in component evaluated TAZ at 3 dose levels (400 mg, 600 mg, and 800 mg orally twice daily [BID]) in 28-day cycles with standard-dose R^2
 - Rituximab was administered at 375 mg/m^2 intravenously on days 1, 8, 15, and 22 of cycle 1, then on day 1 of cycles 2 to 5
 - Lenalidomide was administered at 20 mg (creatinine clearance ≥60 mL/min) or 10 mg (if creatinine clearance <60 mL/min) orally once daily on days 1 to 21 every 28 days for 12 cycles
 - Protocol-mandated dose modifications included simultaneous dose interruption and/or reduction of TAZ and lenalidomide for neutropenia and thrombocytopenia
- Preliminary efficacy analysis was performed on the response-evaluable population
 - The response-evaluable population consists of patients from the intent-to-treat population who had adequate baseline and ≥1 post-baseline tumor assessment, per the International Working Group criteria for non-Hodgkin lymphoma
 - Efficacy was reported as best overall response per investigator assessment, according to Lugano 2014 response criteria
 - Median PFS and duration of response (DOR) were also analyzed
- The safety population is defined as all patients who receive ≥1 dose of study drug, and will be used for all safety analyses

Patient Population

- This study is being conducted in adults with histologically confirmed R/R FL who received ≥1 systemic cancer therapy, with no prior exposure to lenalidomide and/or an EZH2 inhibitor (**Table 1**)

Table 1. Key Inclusion and Exclusion Criteria

Key Inclusion Criteria	Key Exclusion Criteria
• Aged ≥18 years (≥20 years in Taiwan) • Histologically confirmed FL, grade 1–3A • Sufficient tumor tissue for *EZH2* mutation testing • Previously treated with ≥1 prior systemic chemotherapy, immunotherapy, or chemoimmunotherapy • Documented relapsed, refractory, or progressive disease following systemic treatment • Measurable disease, as defined per the Lugano classification • ECOG PS 0–2 • Adequate hematologic, hepatic, and renal function	• Grade 3b, mixed histology, or FL that has histologically transformed to DLBCL • Prior exposure to: – Lenalidomide – Tazemetostat or other EZH2 inhibitor(s) • Uncontrolled leptomeningeal or uncontrolled/previously treated brain metastasis • Active infection with HBV, HCV, HIV, or HTLV-1 • Prior history of T-LBL or T-ALL • Significant cardiovascular impairment

DLBCL, diffuse large B-cell lymphoma; ECOG PS, Eastern Cooperative Oncology Group performance status; EZH2, enhancer of zeste homolog 2; FL, follicular lymphoma; HBV, hepatitis B virus; HCV, hepatitis C virus; HIV, human immunodeficiency virus; HTLV-1, human T-cell lymphotropic virus 1; T-ALL, T-cell acute lymphoblastic leukemia; T-LBL, T-cell lymphoblastic lymphoma.

RESULTS

- As of January 22, 2022, 44 patients were receiving treatment with TAZ + R^2, and 44 patients were included in the analysis (400 mg [n=4], 600 mg [n=19], and 800 mg [n=21])
- At baseline, patients had a median age of 67 years (range, 39–83) and had received a median of 1 prior therapy (range, 1–4) (**Table 2**)
 - Median duration of treatment exposure was 32.0 weeks (range, 4.1–68.1)
- No dose-limiting toxicities were observed in phase 1b, and no new safety signals were identified as of the January 2022 data cutoff
- The RP3D of TAZ was determined to be 800 mg BID in combination with R^2

Table 2. Patient Characteristics and Demographics

Parameter	TAZ + R^2 (n=44)
Age (range), median, years	67 (39–83)
Male, n (%)	26 (59)
Age ≥65 years, n (%)	26 (59)
ECOG PS, n (%)	
0	32 (73)
1	12 (27)
Stage at diagnosis, n (%)	
I	11 (25)
II	20 (46)
IIIA	12 (27)
Unknown/not reported	1 (2)
LDH > ULN, n (%)	7 (16)
B symptoms, n (%)	6 (14)
Prior lines of systemic anticancer therapy,[a] n (%)	
1	30
2	7
3	2
4	5
Median (range)	1 (1–4)
Prior classes of treatment, n (%)	
Prior anti-CD20 antibody + chemotherapy (R/G-bendamustine, R/G-CHOP-based therapy), n (%)	33 (75)
Prior anti-CD20 antibody as only therapy, n (%)	11 (25)
MT *EZH2*, n (%)	6/42[b] (14)
Refractory to rituximab at baseline, n (%)	15/44 (34)
POD24, n (%)	10/40[c] (25)

[a]14 patients received rituximab maintenance, which was not counted as separate line of treatment. [b]Two patients had unknown *EZH2* status. [c]Excludes 4 patients with DLBCL who progressed and transformed to FL.
DLBCL, diffuse large B-cell lymphoma; EZH2, enhancer of zeste homolog 2; FL, follicular lymphoma; LDH, lactate dehydrogenase; MT, mutant; POD24, progression of disease within 24 months; R/G, rituximab/obinutuzumab; R/G-CHOP, rituximab/obinutuzumab, cyclophosphamide, doxorubicin, vincristine, and prednisone; R2, lenalidomide + rituximab; TAZ, tazemetostat; ULN, upper limit of normal.

Pharmacokinetics

- The mean maximum concentration (C_{max}) (**Figure 2**) and area under the concentration-curve from 0 to tau (AUC_{0-t}) (**Figure 3**) of TAZ 800 mg + R^2 at steady state were similar to those found for TAZ given as monotherapy
 - At cycle 1, day 15, TAZ monotherapy mean C_{max} (coefficient of variation % [CV%]) was 829 ng/mL (56%), and AUC_{0-12h} (CV%) was 3340 ng•h/mL (49%)
- The PK of TAZ at all dose levels was not altered by concomitant administration of daily oral lenalidomide 20 mg, and the PK of lenalidomide was not altered by concomitant administration of the TAZ dose

Figure 2. Pharmacokinetic Analysis of Tazemetostat (Maximum Concentration)

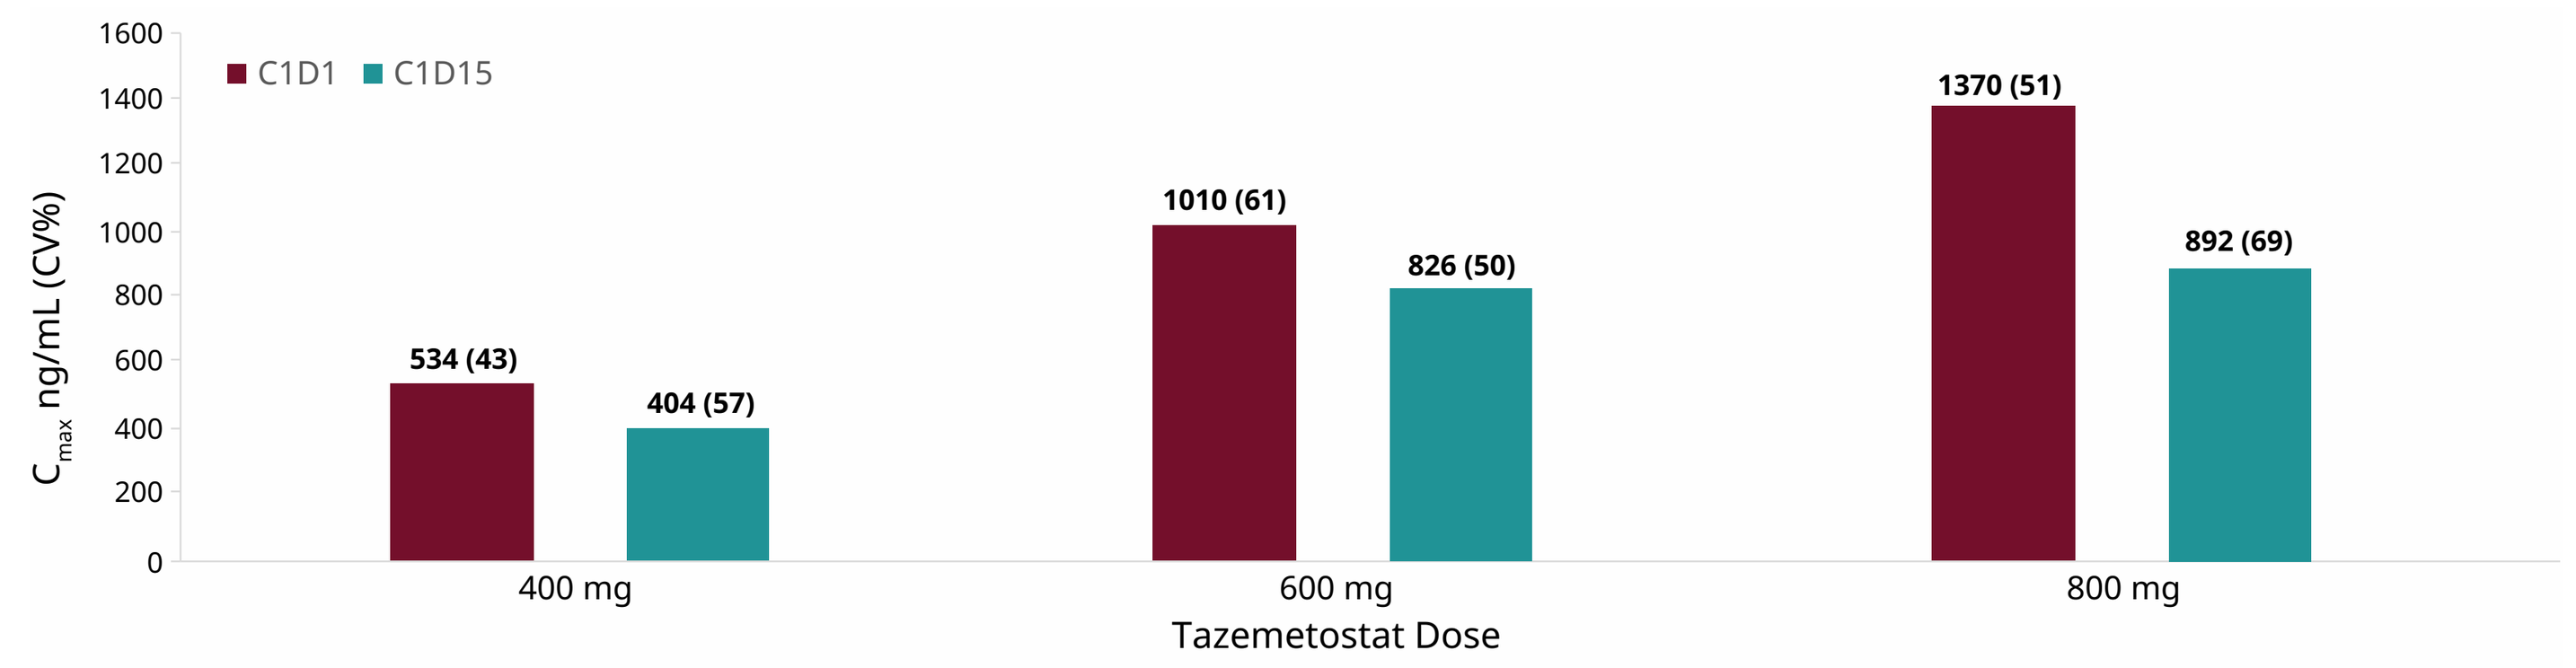
C1D1
C1D15
C_{max} ng/mL (CV%)
0
200
400
600
800
1000
1200
1400
1600
534 (43)
404 (57)
1010 (61)
826 (50)
1370 (51)
892 (69)
400 mg
600 mg
800 mg
Tazemetostat Dose

C1D1, cycle 1 day 1; C1D15, cycle 1 day 15; C_{max}, maximum concentration; CV, coefficient of variation.

Figure 3. Pharmacokinetic Analysis of Tazemetostat (Area Under the Curve)

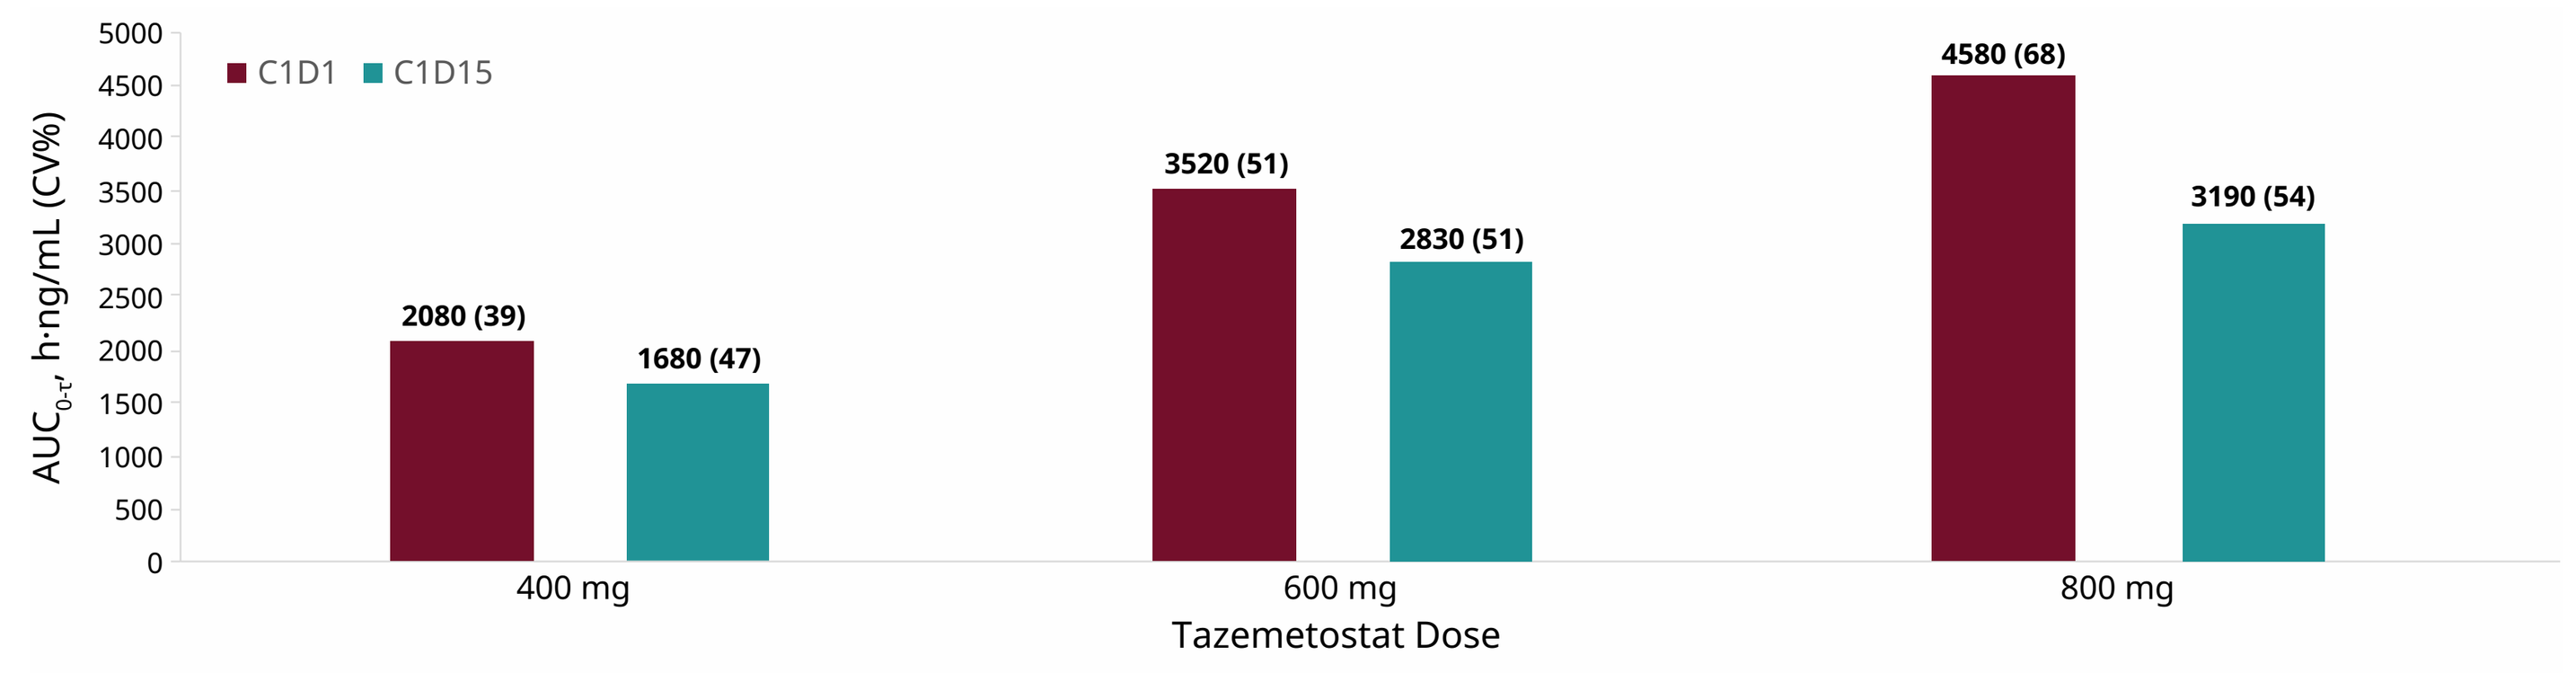
C1D1
C1D15
AUC_{0-t}, h·ng/mL (CV%)
0
500
1000
1500
2000
2500
3000
3500
4000
4500
5000
2080 (39)
1680 (47)
3520 (51)
2830 (51)
4580 (68)
3190 (54)
400 mg
600 mg
800 mg
Tazemetostat Dose

AUC_{0-t}, area under the concentration-time curve from time 0 to 10 hours; C1D1, cycle 1 day 1; C1D15, cycle 1 day 15; CV, coefficient of variation.

Study Drug Exposure

- The most frequent type of dose modification owing to a treatment-emergent adverse event (TEAE) was dose interruption (66%) (**Table 3**)
- As of the data cutoff, median duration of TAZ treatment was 32 weeks
 - Median relative dose intensity for TAZ in the 800-mg BID group was 98%
- For lenalidomide, median duration of exposure was 31 weeks
 - Median relative dose intensity for lenalidomide in the 800-mg BID group was 98%
- For rituximab, median duration of exposure overall was 16 weeks
 - Median relative dose intensity for rituximab in the 800-mg BID group was 100%

Table 3. Dose Modifications for Treatment-Emergent Adverse Events in the Safety Population

Parameter, n (%)	TAZ + R^2 (n=44)
Patients with ≥1 dose modification for a TEAE	30 (68)
Patients with ≥1 modification (for any study treatment)	
Dose reduction	6 (14)
Dose interruption	29 (66)
Treatment discontinuation	6 (14)
Patients with ≥1 modification (for tazemetostat)	
Dose reduction	2 (5)
Drug interruption	24 (55)
Treatment discontinuation	4 (9)
Patients with ≥1 modification (for lenalidomide)	
Dose reduction	5 (11)
Drug interruption	25 (57)
Treatment discontinuation	4 (9)
Patients with ≥1 modification (for rituximab)[a]	
Dose reduction	0
Dose interruption	14 (32)
Treatment discontinuation	2 (5)

[a]Dose interruption was identified only at the first infusion.
TAZ, tazemetostat.

Safety

- Fatigue is the most common any-grade TEAE (**Figure 4**)
- Grade 3/4 TEAEs were observed in 25/44 (57%) patients
- Serious TEAEs were observed in 14/44 (32%) patients

Figure 4. Treatment-Emergent Adverse Events Occurring in ≥20% of Patients in the Safety Population (n=44)

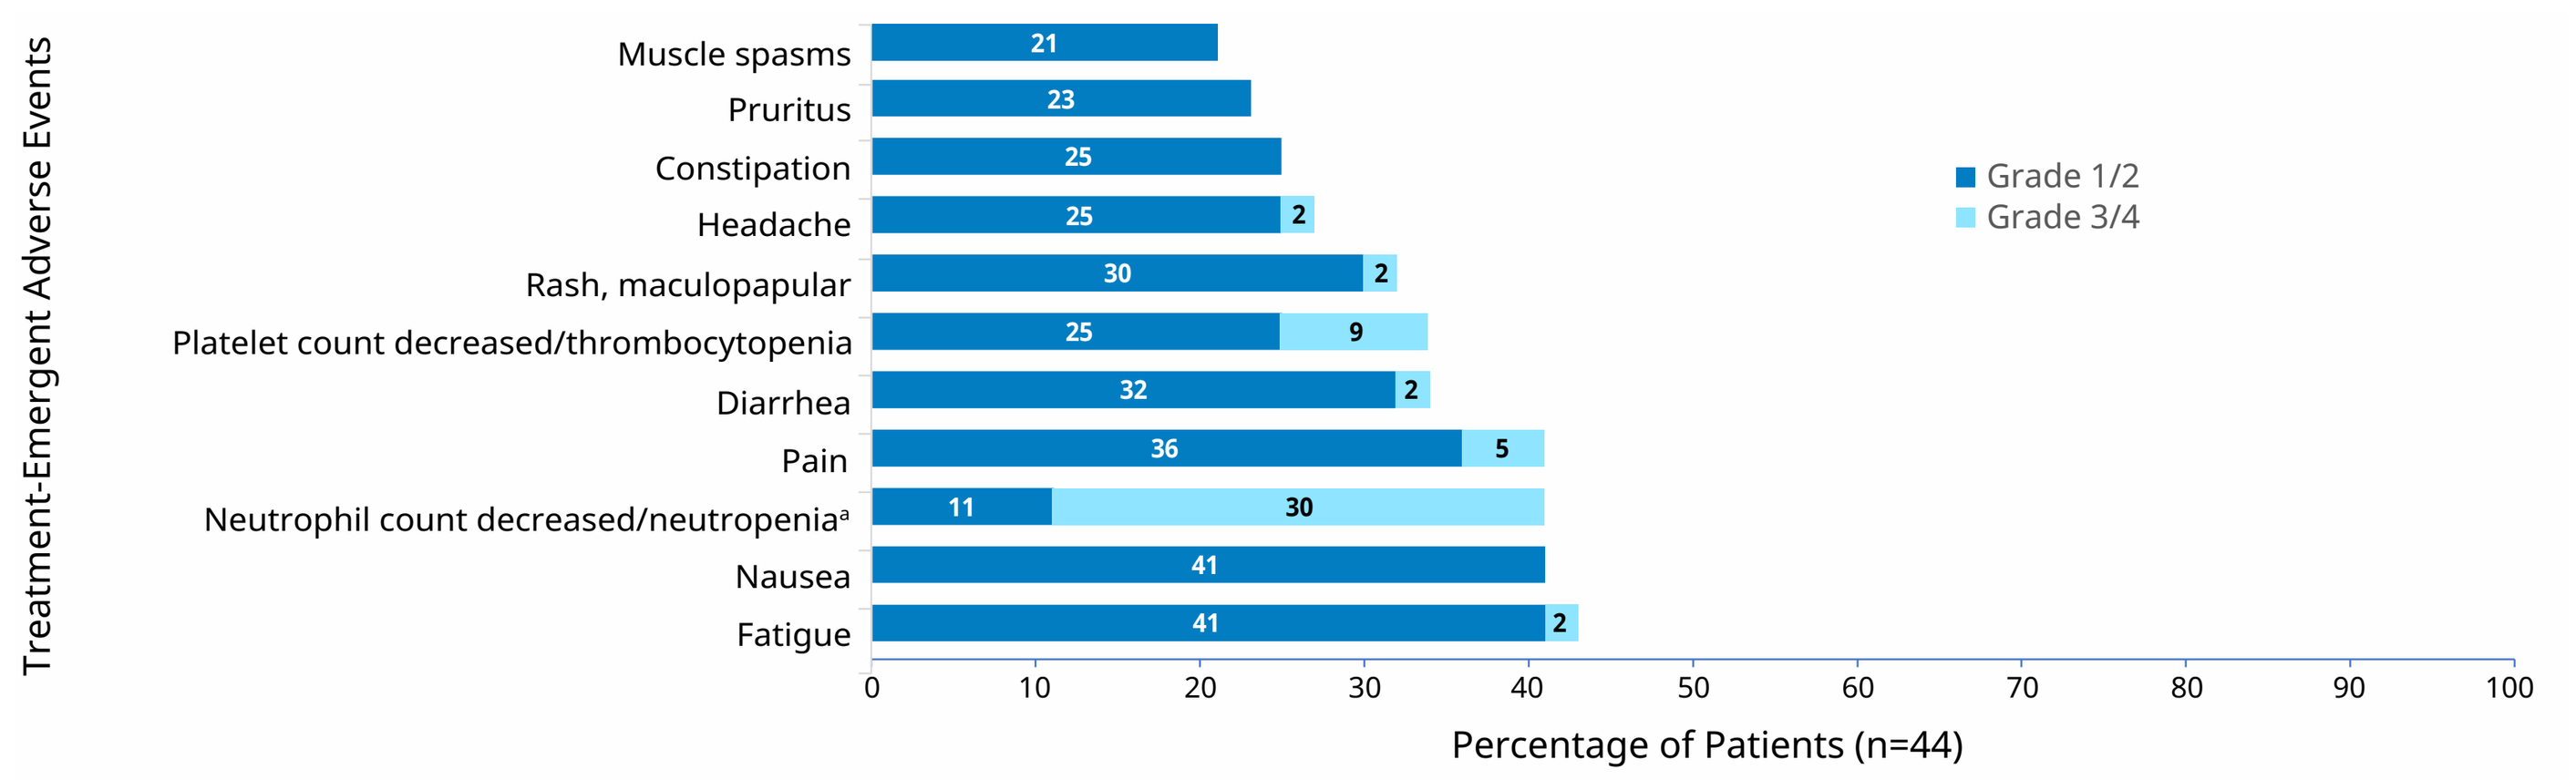
Treatment-Emergent Adverse Events
Muscle spasms 21
Pruritus 23
Constipation 25
Headache 25 2
Rash, maculopapular 30 2
Platelet count decreased/thrombocytopenia 25 9
Diarrhea 32 2
Pain 36 5
Neutrophil count decreased/neutropenia[a] 11 30
Nausea 41
Fatigue 41 2
Grade 1/2
Grade 3/4
0
10
20
30
40
50
60
70
80
90
100
Percentage of Patients (n=44)

[a]Growth factor support was only administered upon occurrence of neutropenia and was not given until after a patient had developed neutropenia. Neutropenia is a common adverse event seen with R^2 treatment.

Preliminary Efficacy

- Of the 38 patients evaluable for tumor assessment, the ORR was 95% (n=36; **Table 4**)
 - 19 patients (50%) had a complete response (CR)
 - 2 patients had progressive disease: 1 patient from the 400-mg cohort and 1 patient from the 600-mg cohort
- The ORR in patients with WT *EZH2* was 94% (n=30)
 - 15 patients (47%) had a CR
- The ORR in patients with MT *EZH2* was 100% (n=5)
 - 3 patients (60%) had a CR
- The ORR in patients with rituximab-refractory disease was 100% (n=13) and 92% (n=22) in patients with rituximab-sensitive disease
 - 6 patients (46%) and 12 patients (50%), respectively, had a CR
- The ORR in patients with POD24 was 100% (n=10) and 93% (n=26) in patients without POD24
 - 4 patients (40%) and 15 patients (54%), respectively, had a CR
- Median PFS (**Figure 5**) and DOR were not reached, with a median follow-up of 5.7 months (range, 2.6–13.9)

Table 4. Preliminary Efficacy Results in the Evaluable Population

Best Overall Response,[a] n (%)	All Dosing Cohorts TAZ + R^2 (n=38)[b]
Objective response rate	36 (95)
Complete response[c]	19 (50)
Partial response	17 (45)
Stable disease	2 (5)

[a]Overall, there were 31 PET-CT–based responses and 7 CT-based responses. [b]6 patients were not included in the initial efficacy assessments. [c]For complete response, 18 were PET-CT–based responses and 1 was a CT-based response.
CT, computed tomography; PET, positron emission tomography; R^2, lenalidomide + rituximab; TAZ, tazemetostat.

Figure 5. Progression-Free Survival[a]

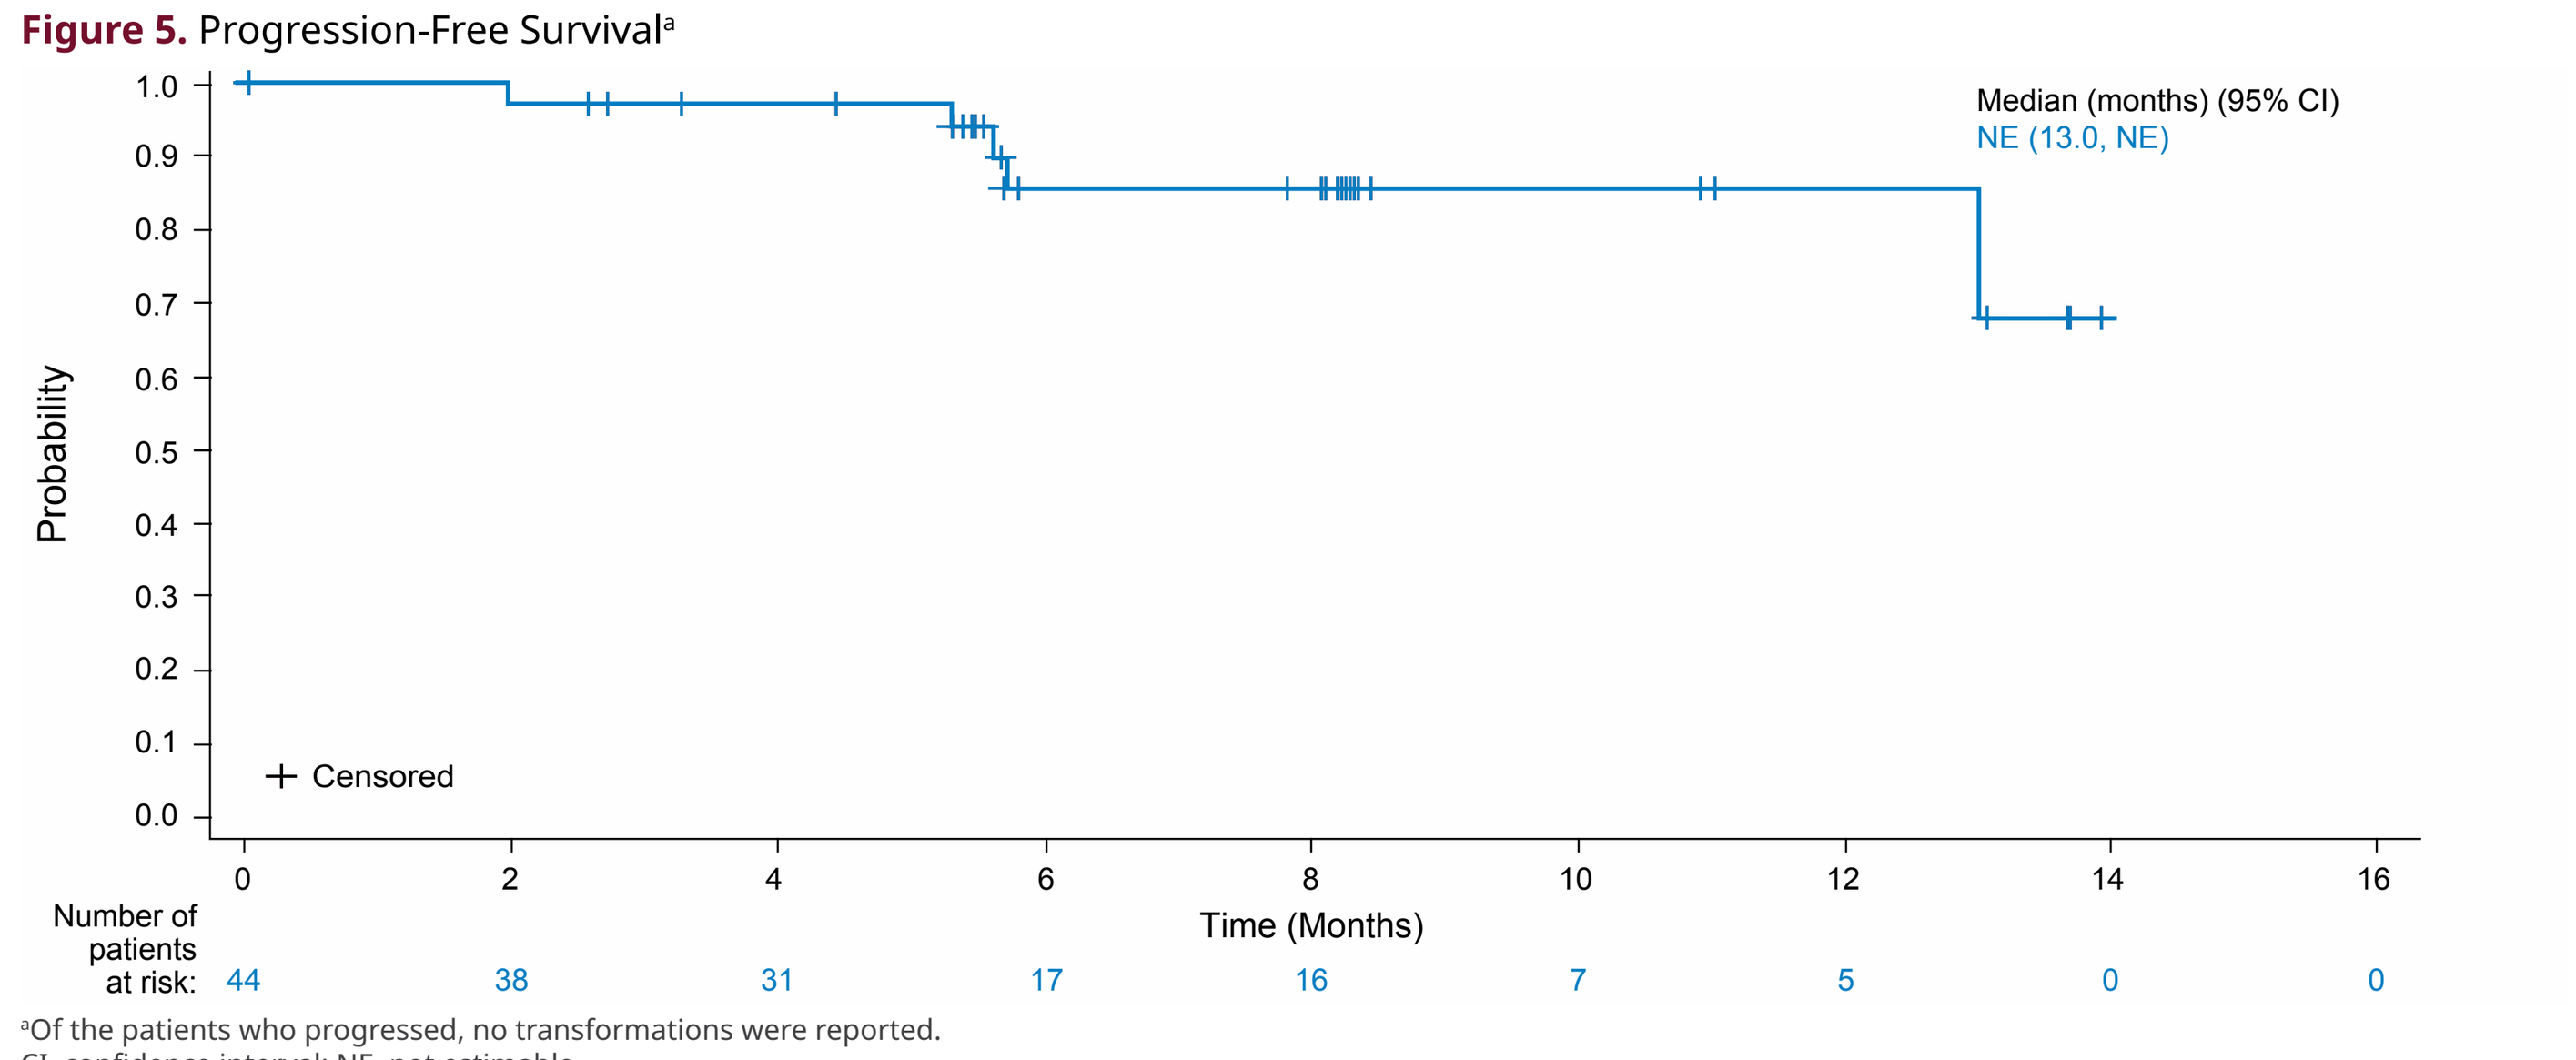
Probability
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
Median (months) (95% CI)
NE (13.0, NE)
+ Censored
0
2
4
6
8
10
12
14
16
Time (Months)
Number of patients at risk: 44 38 31 17 16 7 5 0 0

[a]Of the patients who progressed, no transformations were reported.
CI, confidence interval; NE, not estimable.

CONCLUSIONS

- The safety run-in period comprised 44 patients, including one third each with rituximab-refractory and POD24/transformed FL, who were followed for ≈6 months
- The safety profile of this combination is consistent with previously reported safety information for R^2 and TAZ[1,4]
- There was no clear dose-response relationship for TEAEs and dose modifications
- The mean C_{max} and AUC_{0-t} of TAZ 800 mg + R^2 at steady state was similar to that of TAZ monotherapy
- Preliminary efficacy results are encouraging, with 19 CRs and 17 PRs in the evaluable population of 38 patients
- The safety run-in analysis shows that this combination is safe and tolerable, and that patients are able to maintain a high dose intensity
- The randomized phase 3 portion of the study is actively enrolling patients, aiming to evaluate and compare PFS with TAZ 800 mg BID + R^2 vs placebo + R^2 in ≈500 patients with R/R FL who completed ≥1 prior systemic therapy

References

1. Tazverik [package insert]. Cambridge, MA: Epizyme; 2020.
2. Morschhauser F, et al. *Lancet Oncol*. 2020;21:1433-42.
3. Rituxan [package insert]. South San Francisco, CA: Genentech; 2021.
4. Revlimid [package insert]. Summit, NJ: Celgene; 2021.
5. Leonard JP, et al. *J Clin Oncol*. 2019;37:1188-99.
6. Andorsky DJ, et al. *J Clin Oncol*. 2019;37(15 suppl):7513.

Author Disclosures

C. L. Batlevi has received research funding from Autolus, Bayer, Epizyme, Janssen, Novartis, Roche/Genentech, and Xynomic, and has served as a consultant for GLG Pharma, Juno/Celgene, Kite Pharma, Life Sciences, and Seattle Genetics. **S. Park** has served as a consultant for G1 Therapeutics and Teva; has received research funding from BMS and Seattle Genetics; has served on a speaker's bureau for Gilead and Seattle Genetics; and has served on an advisory board for Rafael Pharma. **T. Phillips** has served as a consultant for AbbVie, AstraZeneca, Bayer, BeiGene, Bristol Myers Squibb, Cardinal Health, Incyte, Karyopharm, Pharmacyclics, and Seattle Genetics; received travel expenses from Incyte; and received research funding from AbbVie and Bayer. **J. Amengual** has received a research grant from Appia Pharmaceuticals and has served as a speaker for Epizyme, Inc. **D. Andorsky** has served on steering committees for AstraZeneca and has served as a consultant for AbbVie. **P. Campbell** has served as a consultant for Amgen, AstraZeneca, CSL Behring, Janssen, Novartis, and Roche, and received research funding from Amgen, Celgene (BMS), Janssen, Novartis, and Roche. **P. McKay** has received honoraria from Gilead, Janssen, Roche, and Takeda and has served on a board of directors or advisory board for BeiGene, Celgene, Gilead, Janssen, Roche, and Takeda. J. Leonard has served as a consultant for ADC Therapeutics, AstraZeneca, Bayer, BMS/Celgene, Epizyme, Inc., Genmab, Gilead/Kite, Karyopharm, BMS/Celgene, Regeneron, MEI Pharma, Miltenyi, Roche/Genentech, and **Sutro. M Sondhi, Y. Chen , H. O'Connor, P. Slatcher,** are or were employees of Epizyme, Inc., and may own stock/options in the company. **F. Morschhauser** has served as a consultant for F. Hoffmann-La Roche, Genentech, and Servier; has received honoraria from Janssen; and has served on an advisory board or board of directors for AbbVie, Celgene, Epizyme, F. Hoffmann-La Roche, and Gilead.

Acknowledgments

This study was sponsored by Epizyme, Inc. Medical writing and editorial support were provided by Peloton Advantage, LLC, an OPEN Health company, and funded by Epizyme, Inc.

Contact: Jennifer E. Amengual, jea2149@cumc.columbia.edu

Presented at the 2022 ASCO Annual Meeting (American Society of Clinical Oncology), June 3–7, 2022, Chicago, IL.